Systemic Small Molecule TREX1 Inhibitors to Selectively Activate STING in the TME of Metastatic Disease

Brian Francica, Dara Burdette, Ryan Clark, Jamie Cope, David Freund, Anja Holtz, David Hsieh, Andy Jennings, Saheli Samanta, Peppi Prasit, Chan Whiting, Thomas W. Dubensky

Tempest Therapeutics, 7000 Shoreline Ct, South San Francisco, CA, USA

ABSTRACT

Genetic evidence from human disease and mouse genetic knock-out studies identify the **St**imulator of **In**terferon **G**enes (STING) pathway as a critical innate immune sensor for the development of immunity. Tumor cells can evolve to avoid immune recognition through inactivating the STING pathway by diverse mechanisms, indicating that it is important to generating tumor-specific immunity. However, the clinical activity of STING agonists given by intratumoral (IT) administration has not compared well to preclinical studies. The scientific hypothesis supporting these first clinical studies is that localized CD8⁺ T cell priming would have activity against distal non-injected tumors, but findings that tumors in advanced malignancies have unique antigenic repertoires suggests that this approach may have limited activity against distal tumors. Global innate activation in metastases may therefore be necessary to prime a broadly active CD8⁺ T cell population targeting diverse antigens, in addition having the benefit of reversing the immune suppressive tumor microenvironment (TME). However, ubiquitous expression of STING presents a significant challenge to achieving a therapeutic index with systemic delivery of direct STING agonists. Selective activation of the STING pathway may be achieved through targeted inhibition of TREX1, a cytosolic DNA exonuclease that modulates cGAS/STING signaling. Expression of TREX1, in contrast to STING, is increased in tumor cells due to elevated levels of cytosolic DNA resulting from genetic instability, DNA repair mutation, inflammatory mediators or DNA-modifying anti-cancer therapies. These observations provide the principal scientific rationale to selectively activate the STING pathway in metastatic disease through targeted inhibition of TREX1. Utilizing published TREX1 X-ray crystal structures to guide medicinal chemistry, we discovered small molecule inhibitors of TREX1 and transformed these molecules from >100 μM leads into a series with drug-like physicochemical properties and picomolar potency against both human and mouse TREX1. We evaluated the activity of lead molecules in cell-based assays, in which TREX1 inhibition enhanced cGAS/STING signaling, and advanced molecules with desired pharmacokinetic profiles to mouse tumor models. We observed significant anti-tumor activity in mice with CT26 tumors given a combined therapy of low dose doxorubicin to induce dsDNA breaks and increase TME TREX1 expression along with lead series TREX1 inhibitors. Recognizing that TREX1 is a DNA repair enzyme, we also showed that TREX1 inhibitors were cytotoxic in DNA repair deficient human tumor cell lines, informing advancement of this new class of STING therapeutics as a clinical approach to both activate the cGAS/STING pathway to initiate immune recognition, as well as to inhibit DNA repair orthogonal to existing tumor-driver DNA repair mutations.

INTRODUCTION

Drugging the STING Pathway Through Targeting TREX1

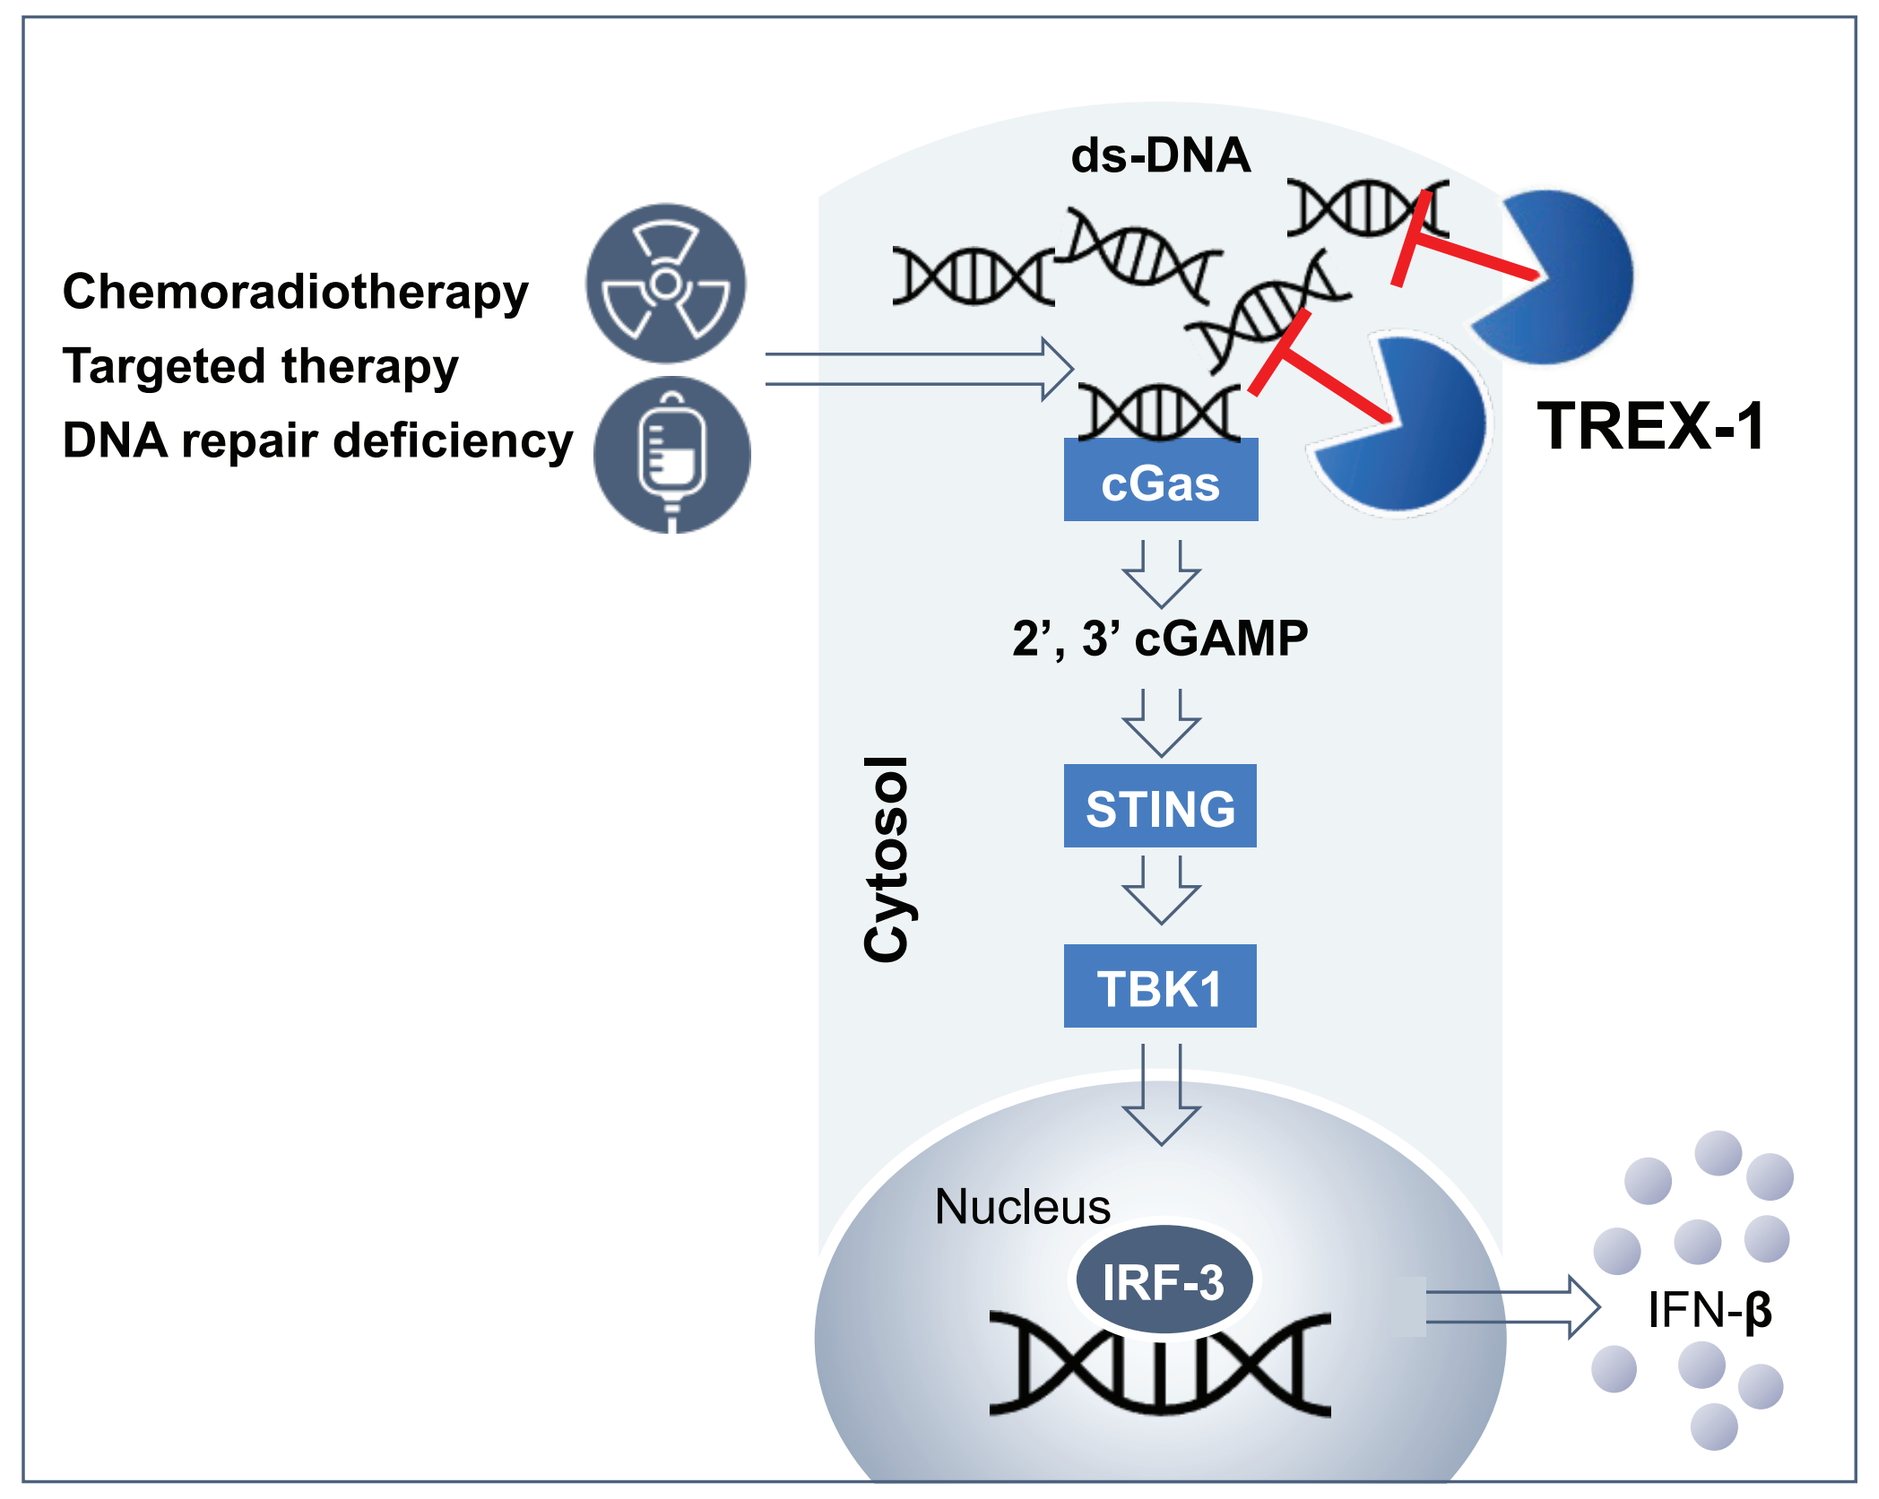
Chemoradiotherapy
Targeted therapy
DNA repair deficiency
ds-DNA
cGas
TREX-1
2', 3' cGAMP
Cytosol
STING
TBK1
Nucleus
IRF-3
IFN-β

- **STING** is a critical innate immune sensor for the development of immunity
 - STING activating mutations result in profound immune inflammatory diseases
 - Malignant cells evolve to inactivate STING by diverse mechanisms
- **TREX1** is a negative regulator of STING
 - Cytosolic dsDNA 3'- 5' non-processive exonuclease
 - DNA base and nucleotide excision repair enzyme
 - Reduces cGAS-STING activation by DNA
- **TREX1** expression can be upregulated in the TME
 - Tumor genetic instability and DNA damaging therapies increase cytosolic dsDNA and induce TREX1
 - TREX1 upregulation protects tumor from immune recognition
- TREX1 ***inhibition*** activates the STING pathway
 - Enhanced TREX1 expression in tumors vs healthy tissues should improve therapeutic index compared to direct STING agonists

APPROACH

Figure 1. Development of Selective TREX1 Inhibitors

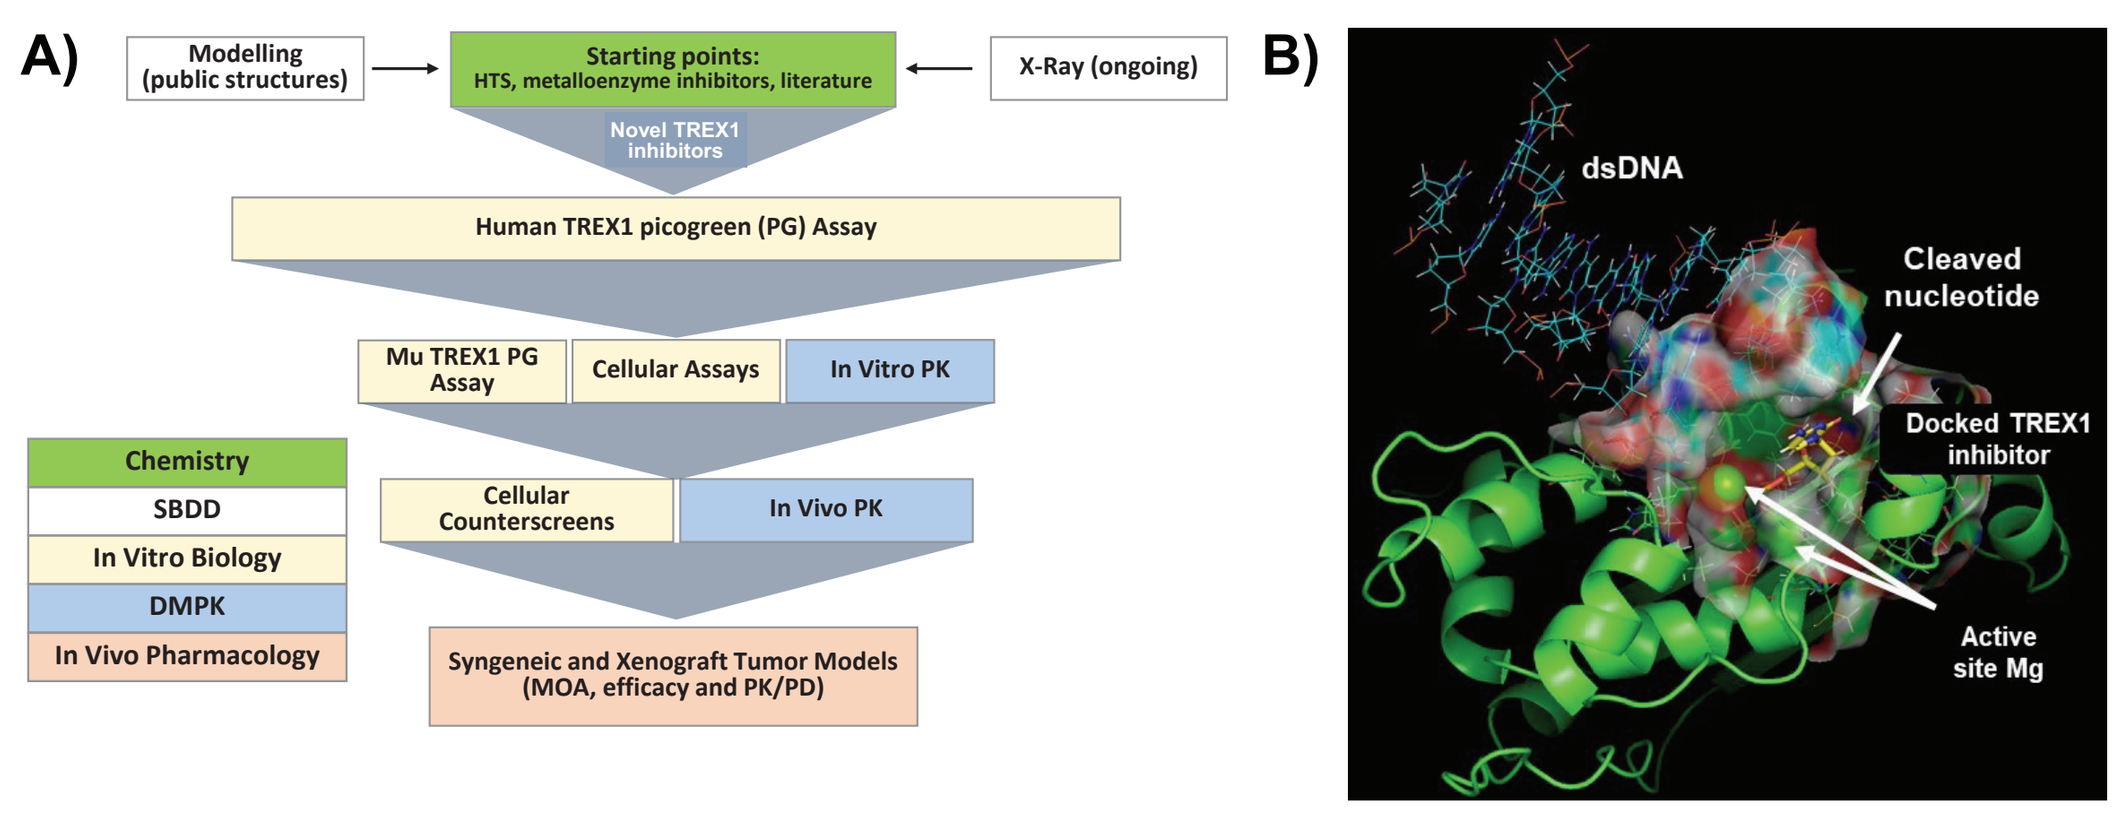
A)
Modelling (public structures)
Starting points: HTS, metalloenzyme inhibitors, literature
X-Ray (ongoing)
Novel TREX1 inhibitors
Human TREX1 picogreen (PG) Assay
Mu TREX1 PG Assay
Cellular Assays
In Vitro PK
Cellular Counterscreens
In Vivo PK
Syngeneic and Xenograft Tumor Models (MOA, efficacy and PK/PD)
Chemistry
SBDD
In Vitro Biology
DMPK
In Vivo Pharmacology
B)
dsDNA
Cleaved nucleotide
Docked TREX1 inhibitor
Active site Mg

Approach to develop potent and selective TREX1 inhibitors. A) Screening funnel schematic illustrating steps taken to select TREX1-specific inhibitors. Steps shown are iterative based on experimental results; **B)** Lead Series molecules are predicted to bind in the TREX1 active site. Our docking model (based on a published mouse TREX1/DNA co-crystal structure) aligns with ongoing SAR (not shown).

RESULTS

Figure 2. Activity of a Lead Series TREX1 inhibitors in biochemical assay

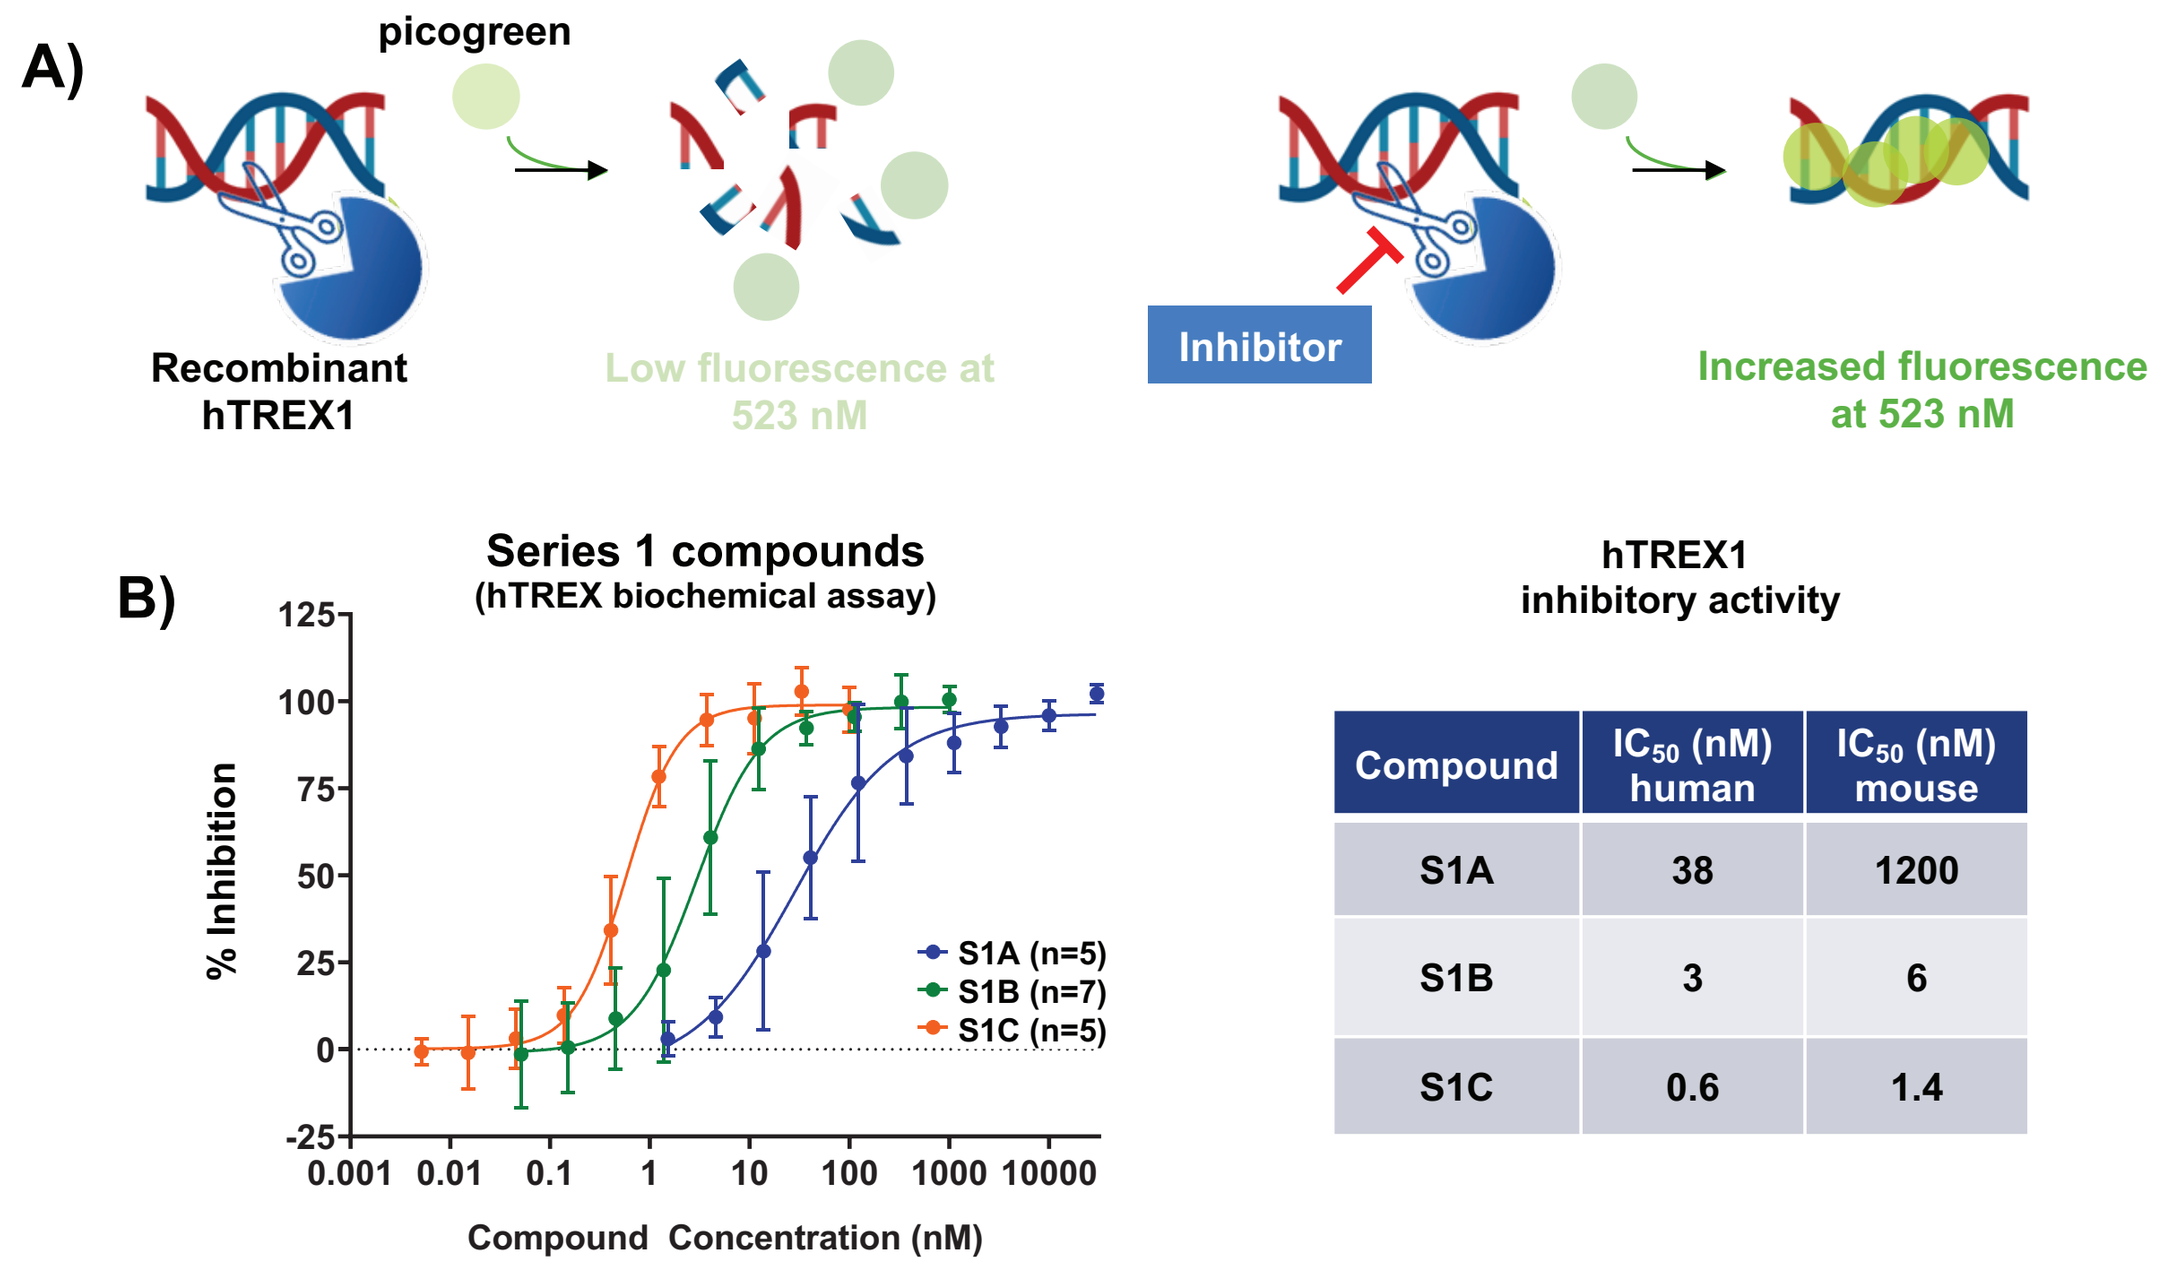
A)
picogreen
Recombinant hTREX1
Low fluorescence at 523 nM
Inhibitor
Increased fluorescence at 523 nM
B)
Series 1 compounds
(hTREX biochemical assay)
% Inhibition
Compound Concentration (nM)
S1A (n=5)
S1B (n=7)
S1C (n=5)
hTREX1
inhibitory activity

Compound	IC_{50} (nM) human	IC_{50} (nM) mouse
S1A	38	1200
S1B	3	6
S1C	0.6	1.4

Potent inhibition of TREX1 inhibitor compounds in biochemical fluorescent assay A) Assay schematic illustrating combination of compound with dsDNA template labeled with picogreen to assess compound TREX1 inhibitory activity. Enzymatic assays were conducted with 8 ng/mL TREX1 protein in buffer with DDT incubated at 25 °C for 30 min with compound at desired concentrations followed by addition of dsDNA template at 50 nM final concentration, mixing, and incubation at 25 °C for 5 min. Inhibition of DNA degradation was measured by fluorescence at 520 nm upon 480 nm excitation immediately following the addition of EDTA stop solution containing picogreen; **B)** Human TREX1 inhibitory activity of lead molecules determined over 10-point dose range. Table shows IC_{50} values of lead molecules against both human and mouse TREX1.

Figure 3. Activity of a Lead Series TREX1 inhibitors in THP-1 reporter cell lines

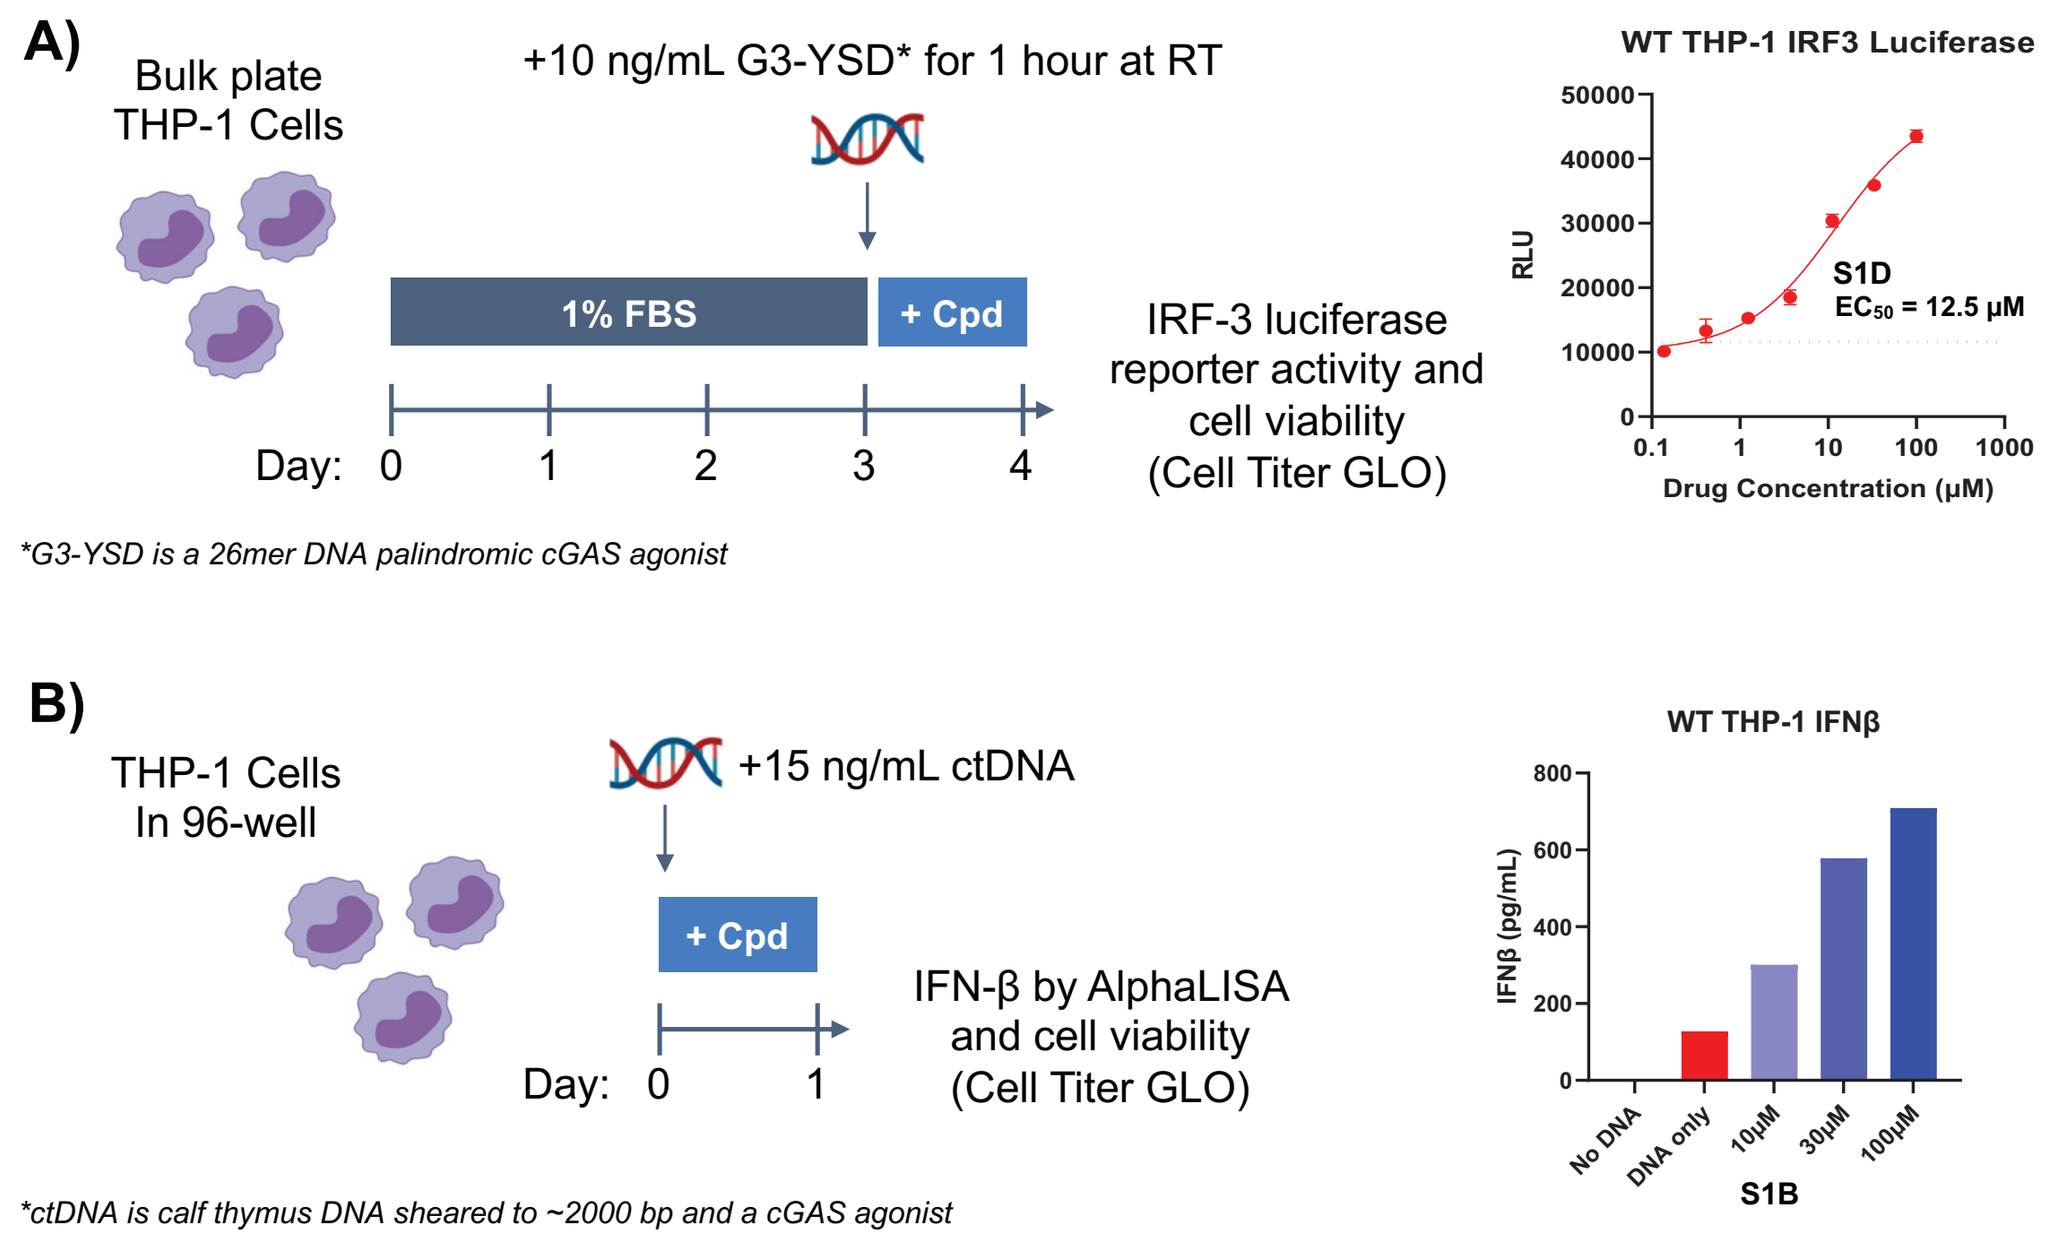
A)
Bulk plate THP-1 Cells
+10 ng/mL G3-YSD* for 1 hour at RT
1% FBS
+ Cpd
Day: 0 1 2 3 4
IRF-3 luciferase reporter activity and cell viability (Cell Titer GLO)
WT THP-1 IRF3 Luciferase
RLU
S1D
EC50 = 12.5 μM
Drug Concentration (μM)
*G3-YSD is a 26mer DNA palindromic cGAS agonist
B)
THP-1 Cells In 96-well
+15 ng/mL ctDNA
+ Cpd
Day: 0 1
IFN-β by AlphaLISA and cell viability (Cell Titer GLO)
WT THP-1 IFNβ
IFNβ (pg/mL)
No DNA
DNA only
10μM
30μM
100μM
S1B
*ctDNA is calf thymus DNA sheared to ~2000 bp and a cGAS agonist

A) TREX1 inhibitors increase STING activation in THP-1 reporter cell lines. Schematic showing process for assessment of inhibitor activity through measurement of IRF-3 activation of luciferase reporter. THP-1 cells were batch transfected with 10 ng/mL G3-YSD in Lyovec for 1 h at RT. Cells were added to 96-well plates and incubated for 24 h at 37 °C following addition of drug. Reporter expression w s measured in cell supernatants with Quanti-Luc. Example of EC_{50} results with TREX1 inhibitor determined over 6 different dose levels. **B) TREX1 inhibitors increase production shown of IFN-β in THP-1 reporter cell lines.** 96-well plates were prepared as in (A) except DNA stimulation was with 15 ng/mL sheared ctDNA in Lyovec. IFN-β expression was measured in cell supernatants by AlphaLISA. Example shown of IFN-β production in the presence of TREX1 inhibitor at different concentrations.

Figure 4. Activity of a Lead Series TREX1 inhibitors in CT26 tumor cells

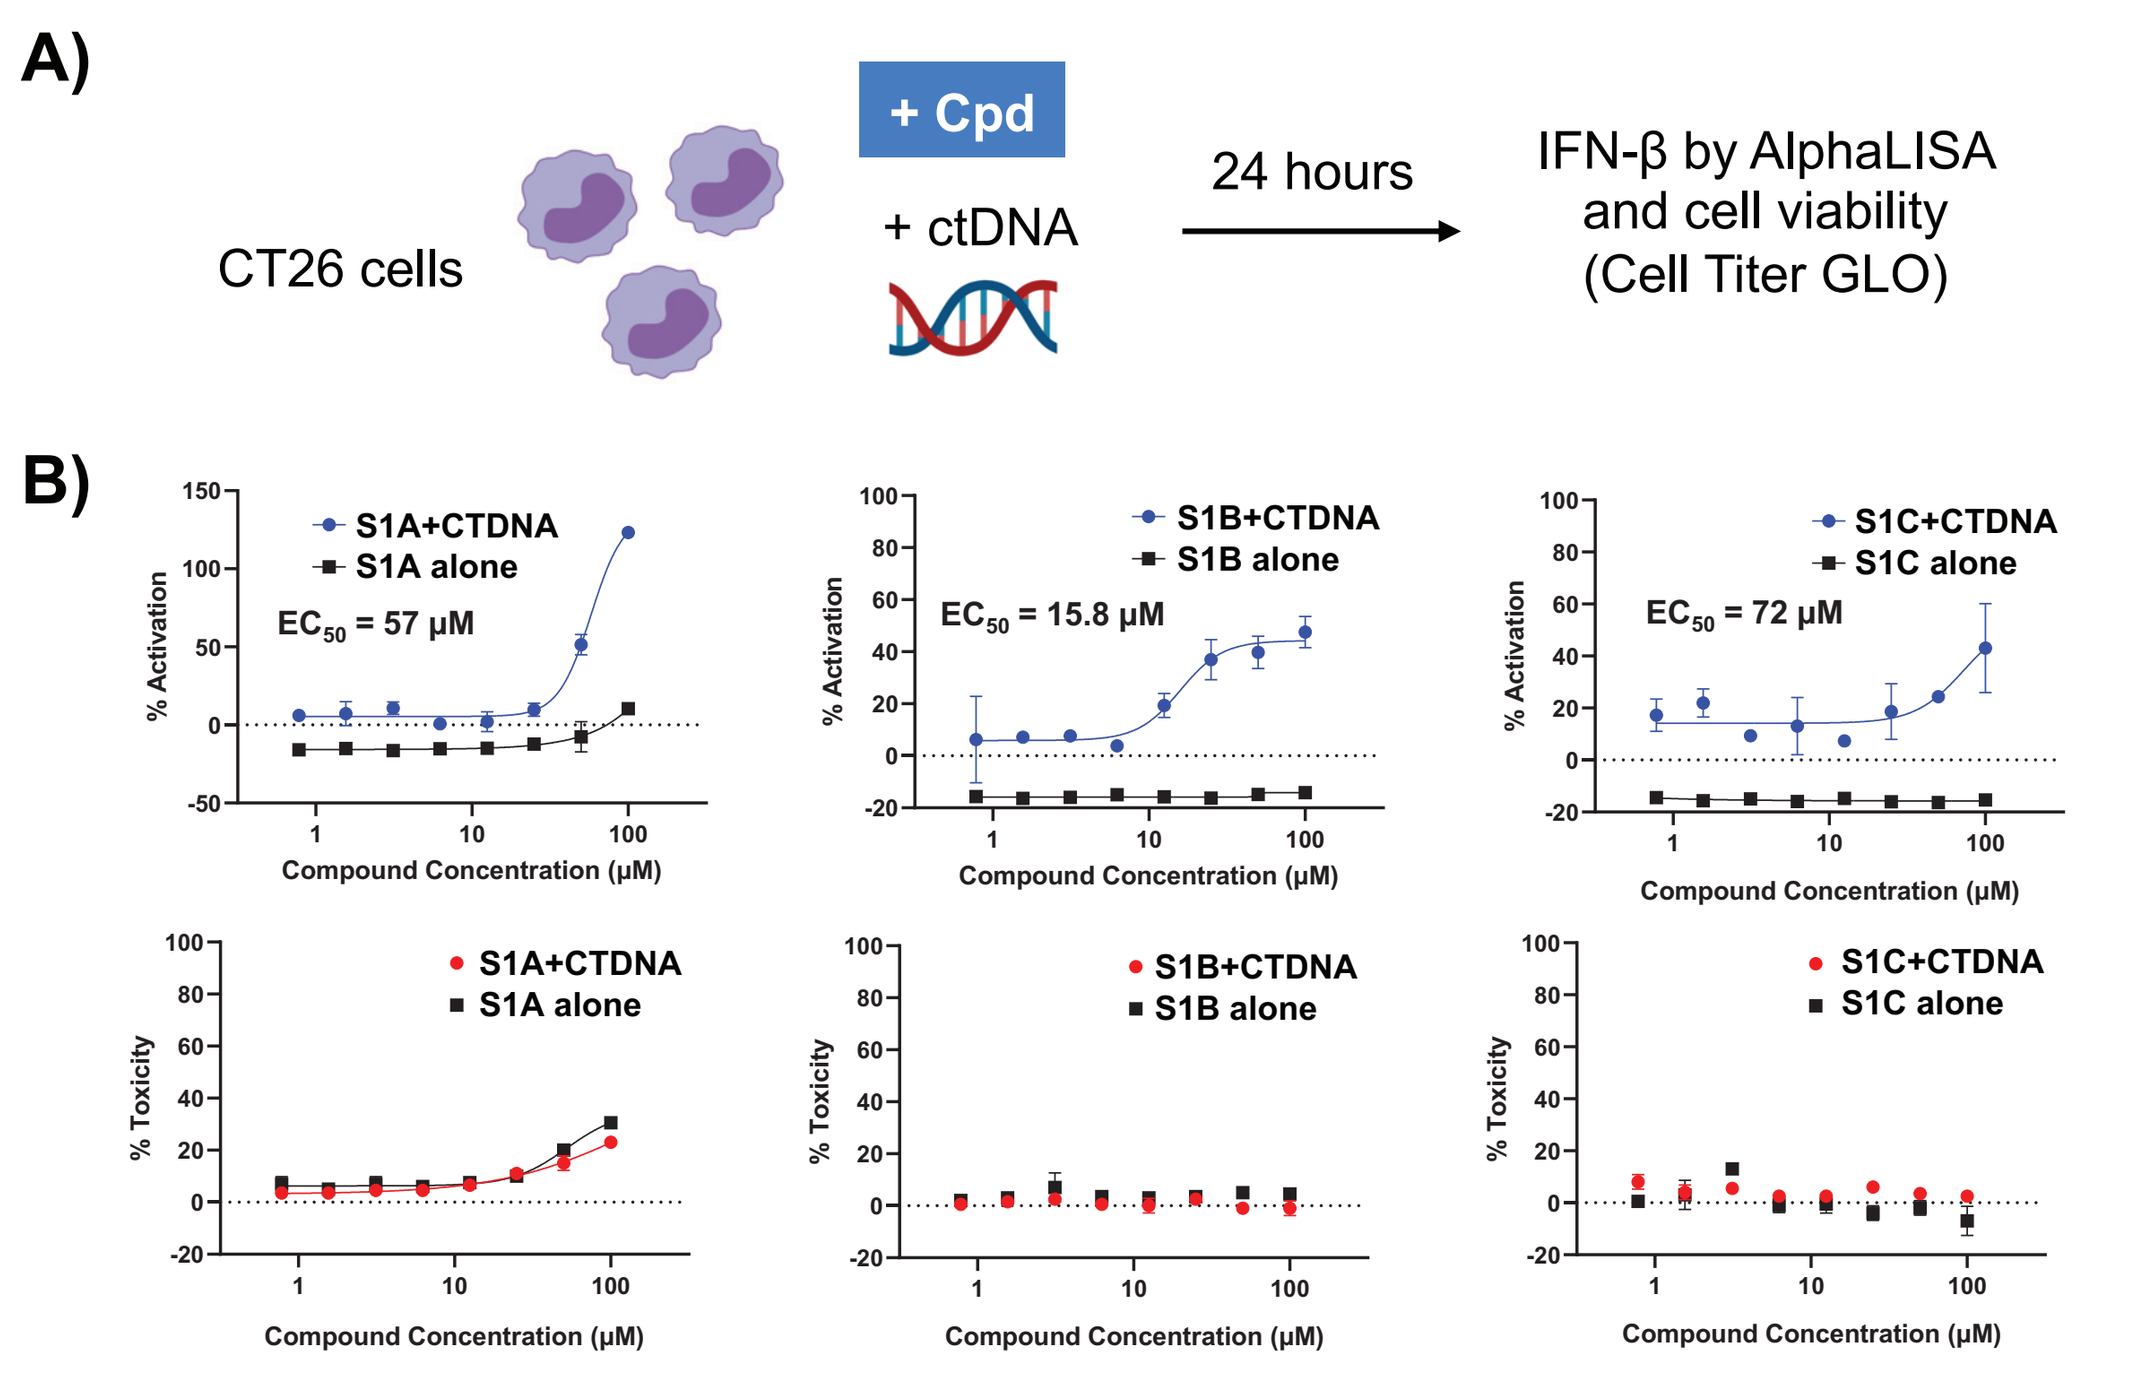
A)
CT26 cells
+ Cpd
+ ctDNA
24 hours
IFN-β by AlphaLISA and cell viability (Cell Titer GLO)
B)
S1A+CTDNA
S1A alone
EC50 = 57 μM
S1B+CTDNA
S1B alone
EC50 = 15.8 μM
S1C+CTDNA
S1C alone
EC50 = 72 μM
% Activation
% Toxicity
Compound Concentration (μM)

A) TREX1 inhibitors increase IFN-β production in DNA-stimulated CT26 tumor cells. Schematic showing process for assessment of inhibitor activity through measurement of IFN-β production. **B)** Cells were prepared in a 96-well format and stimulated with 50 ng/mL ctDNA in Lyovec and drug in DMSO for 1 h as in Fig 3. IFN-β expression was measured in cell supernatants by AlphaLISA. Cell viability was determined using Cell Titer GLO.

Figure 5. Chemotherapy-mediated induction of TREX1 in tumors

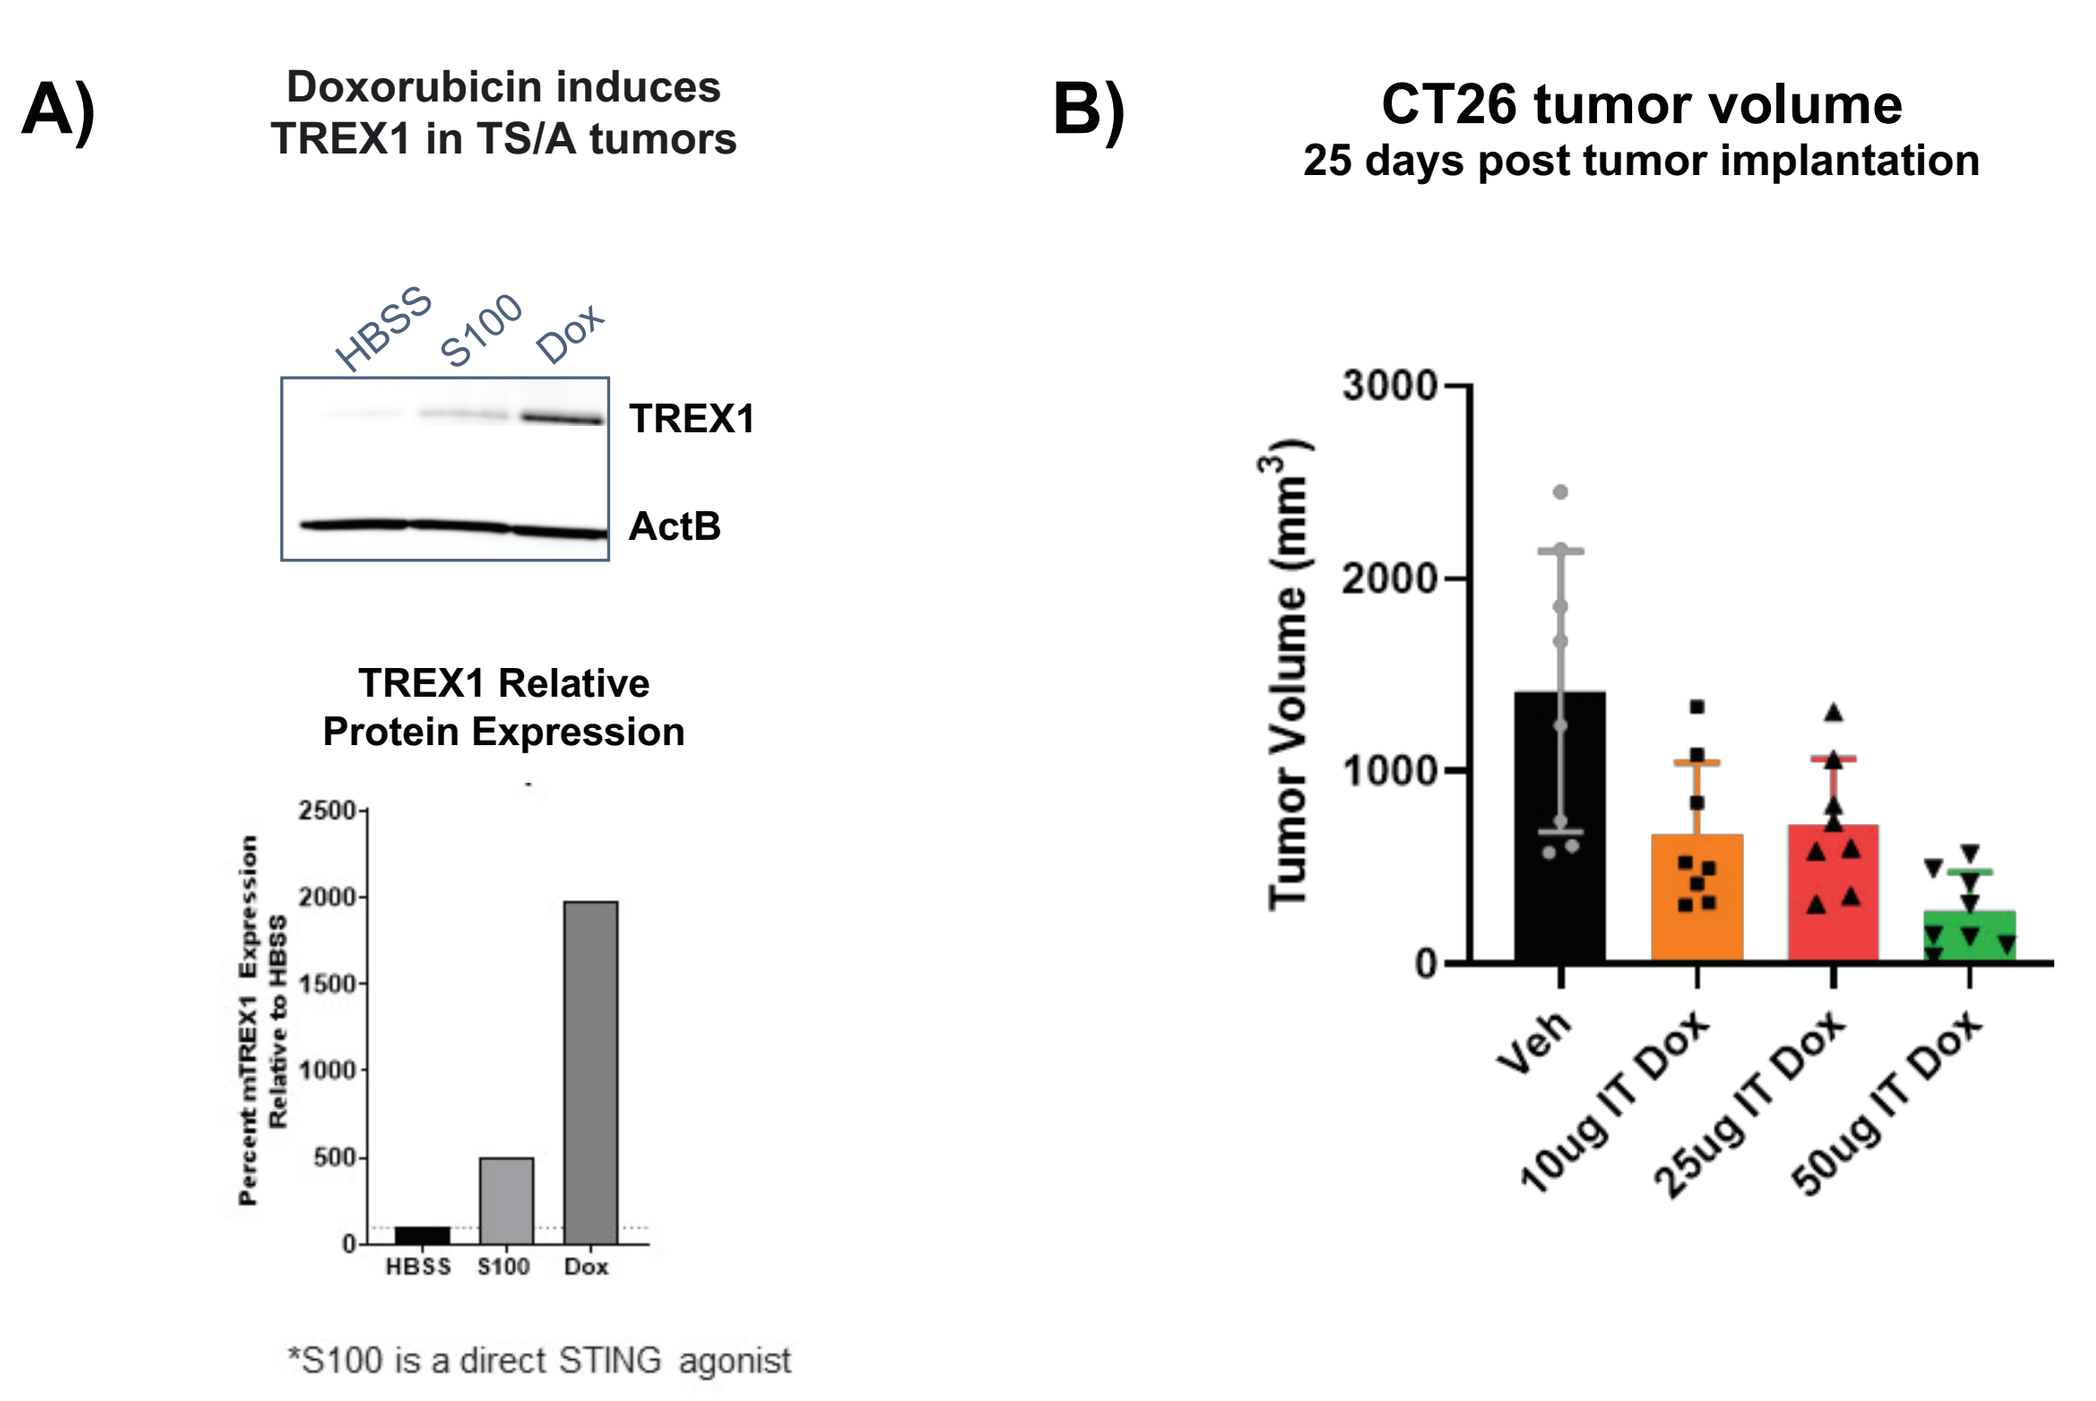
A)
Doxorubicin induces TREX1 in TS/A tumors
HBSS
S100
Dox
TREX1
ActB
TREX1 Relative Protein Expression
Percent mTREX1 Expression Relative to HBSS
*S100 is a direct STING agonist
B)
CT26 tumor volume
25 days post tumor implantation
Tumor Volume (mm³)
Veh
10ug IT Dox
25ug IT Dox
50ug IT Dox

Subtherapeutic doses of Doxorubicin induces dsDNA breaks and activates TREX1 expression. A) TS/A tumors were injected with Vehicle, 25 μg Doxorubicin (dox) or with S100 (direct STING agonist) and 48 h later were prepared for Western blot and qPCR analyses by dissociating tumor in collagenase and benzonase at 37 °C. Processed tumors were filtered and aliquoted. Lysates were prepared for Western blot in RIPA buffer with protease/phosphatase inhibitor cocktail (PIC). After filtering and aliquoting tumors, lysates were prepared for western blot by resuspending the tumor aliquot for western blot in 500uL RIPA buffer with PIC. 50 μg of protein was electrophoresed on 4-12% bis-Tris gels. TREX1 and Actin-B were detected with specific mAbs (Cell Signaling Technology), diluted 1:1000 and 1:2000, respectively. Gel images were obtained with the Azure system. RNAeasy (Qiagen) was used to prepare RNA for qPCR. **B)** Groups of 8 Balb/c mice bearing flank CT26 tumors were given two intratumoral (IT) injections of dox separated by 1 week at the doses shown in the Figure, once tumors were ca.100 mm³ at 11 days post cell implantation.

Figure 6. Anti-tumor activity of TREX1 inhibitors in CT26 tumor model

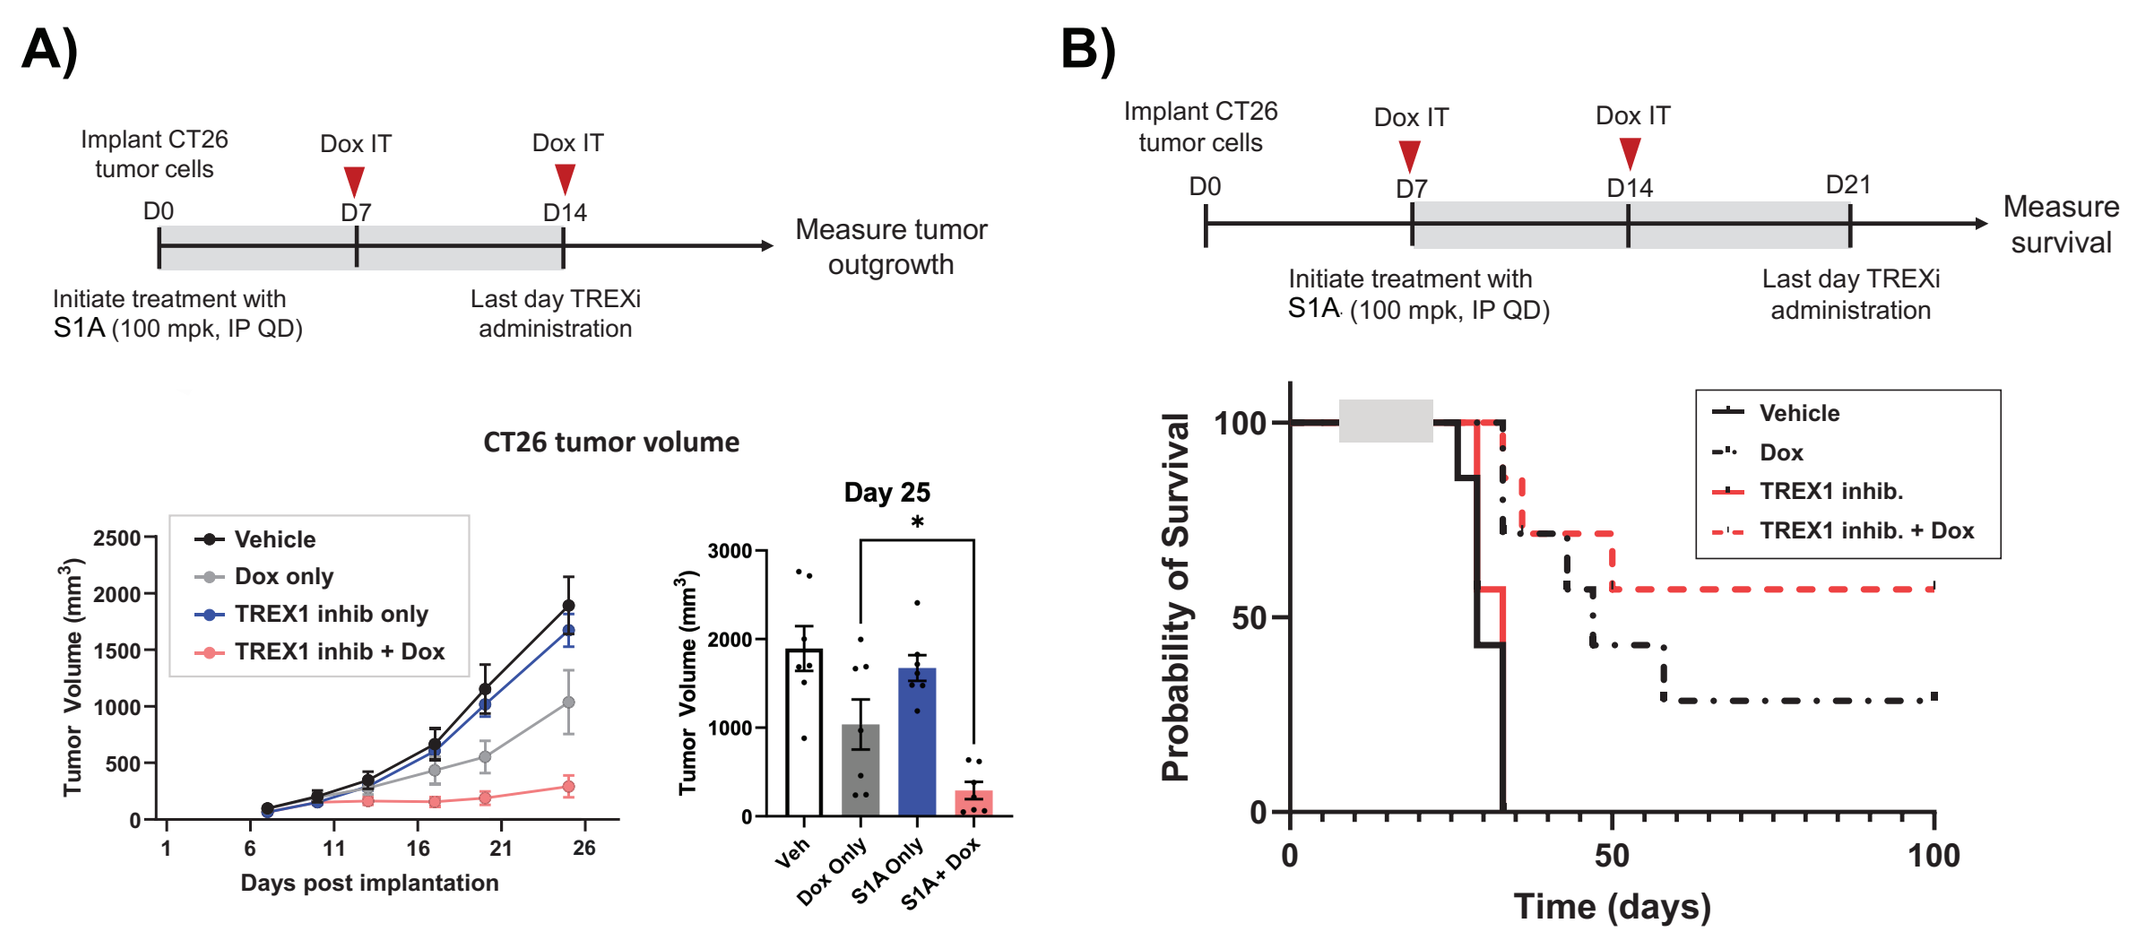
A)
Implant CT26 tumor cells
D0
Dox IT
D7
Dox IT
D14
Measure tumor outgrowth
Initiate treatment with S1A (100 mpk, IP QD)
Last day TREXi administration
CT26 tumor volume
Vehicle
Dox only
TREX1 inhib only
TREX1 inhib + Dox
Days post implantation
Day 25
B)
D21
Measure survival
Probability of Survival
Time (days)
Vehicle
Dox
TREX1 inhib.
TREX1 inhib. + Dox

TREX1 Lead Series molecule therapy results in significant anti-tumor efficacy in combination with subtherapeutic doses of doxorubicin. A) Groups of 7 Balb/c mice were implanted with CT26 tumor cells and on D0 were given 100 MPK of S1A TREX1 inhibitor by IP injection, QD for 14 days. Tumors on D7 and D14 were given 10 μg of dox intratumorally. Tumor growth was measured every 3 days until D25. **B)** Groups of 7 CT26 tumor-bearing Balb/c mice were treated with vehicle, Dox on days 7 and 14, or 14 days of S1A at 100 MPK administered IP QD beginning on D7 when tumors were ca. 100 mm³ and monitored for survival. Significance (combination versus dox alone) was determined by Student's 2-sided t test.

Figure 7. Dose response of TREX1 inhibitors and activity in MC38 tumor model

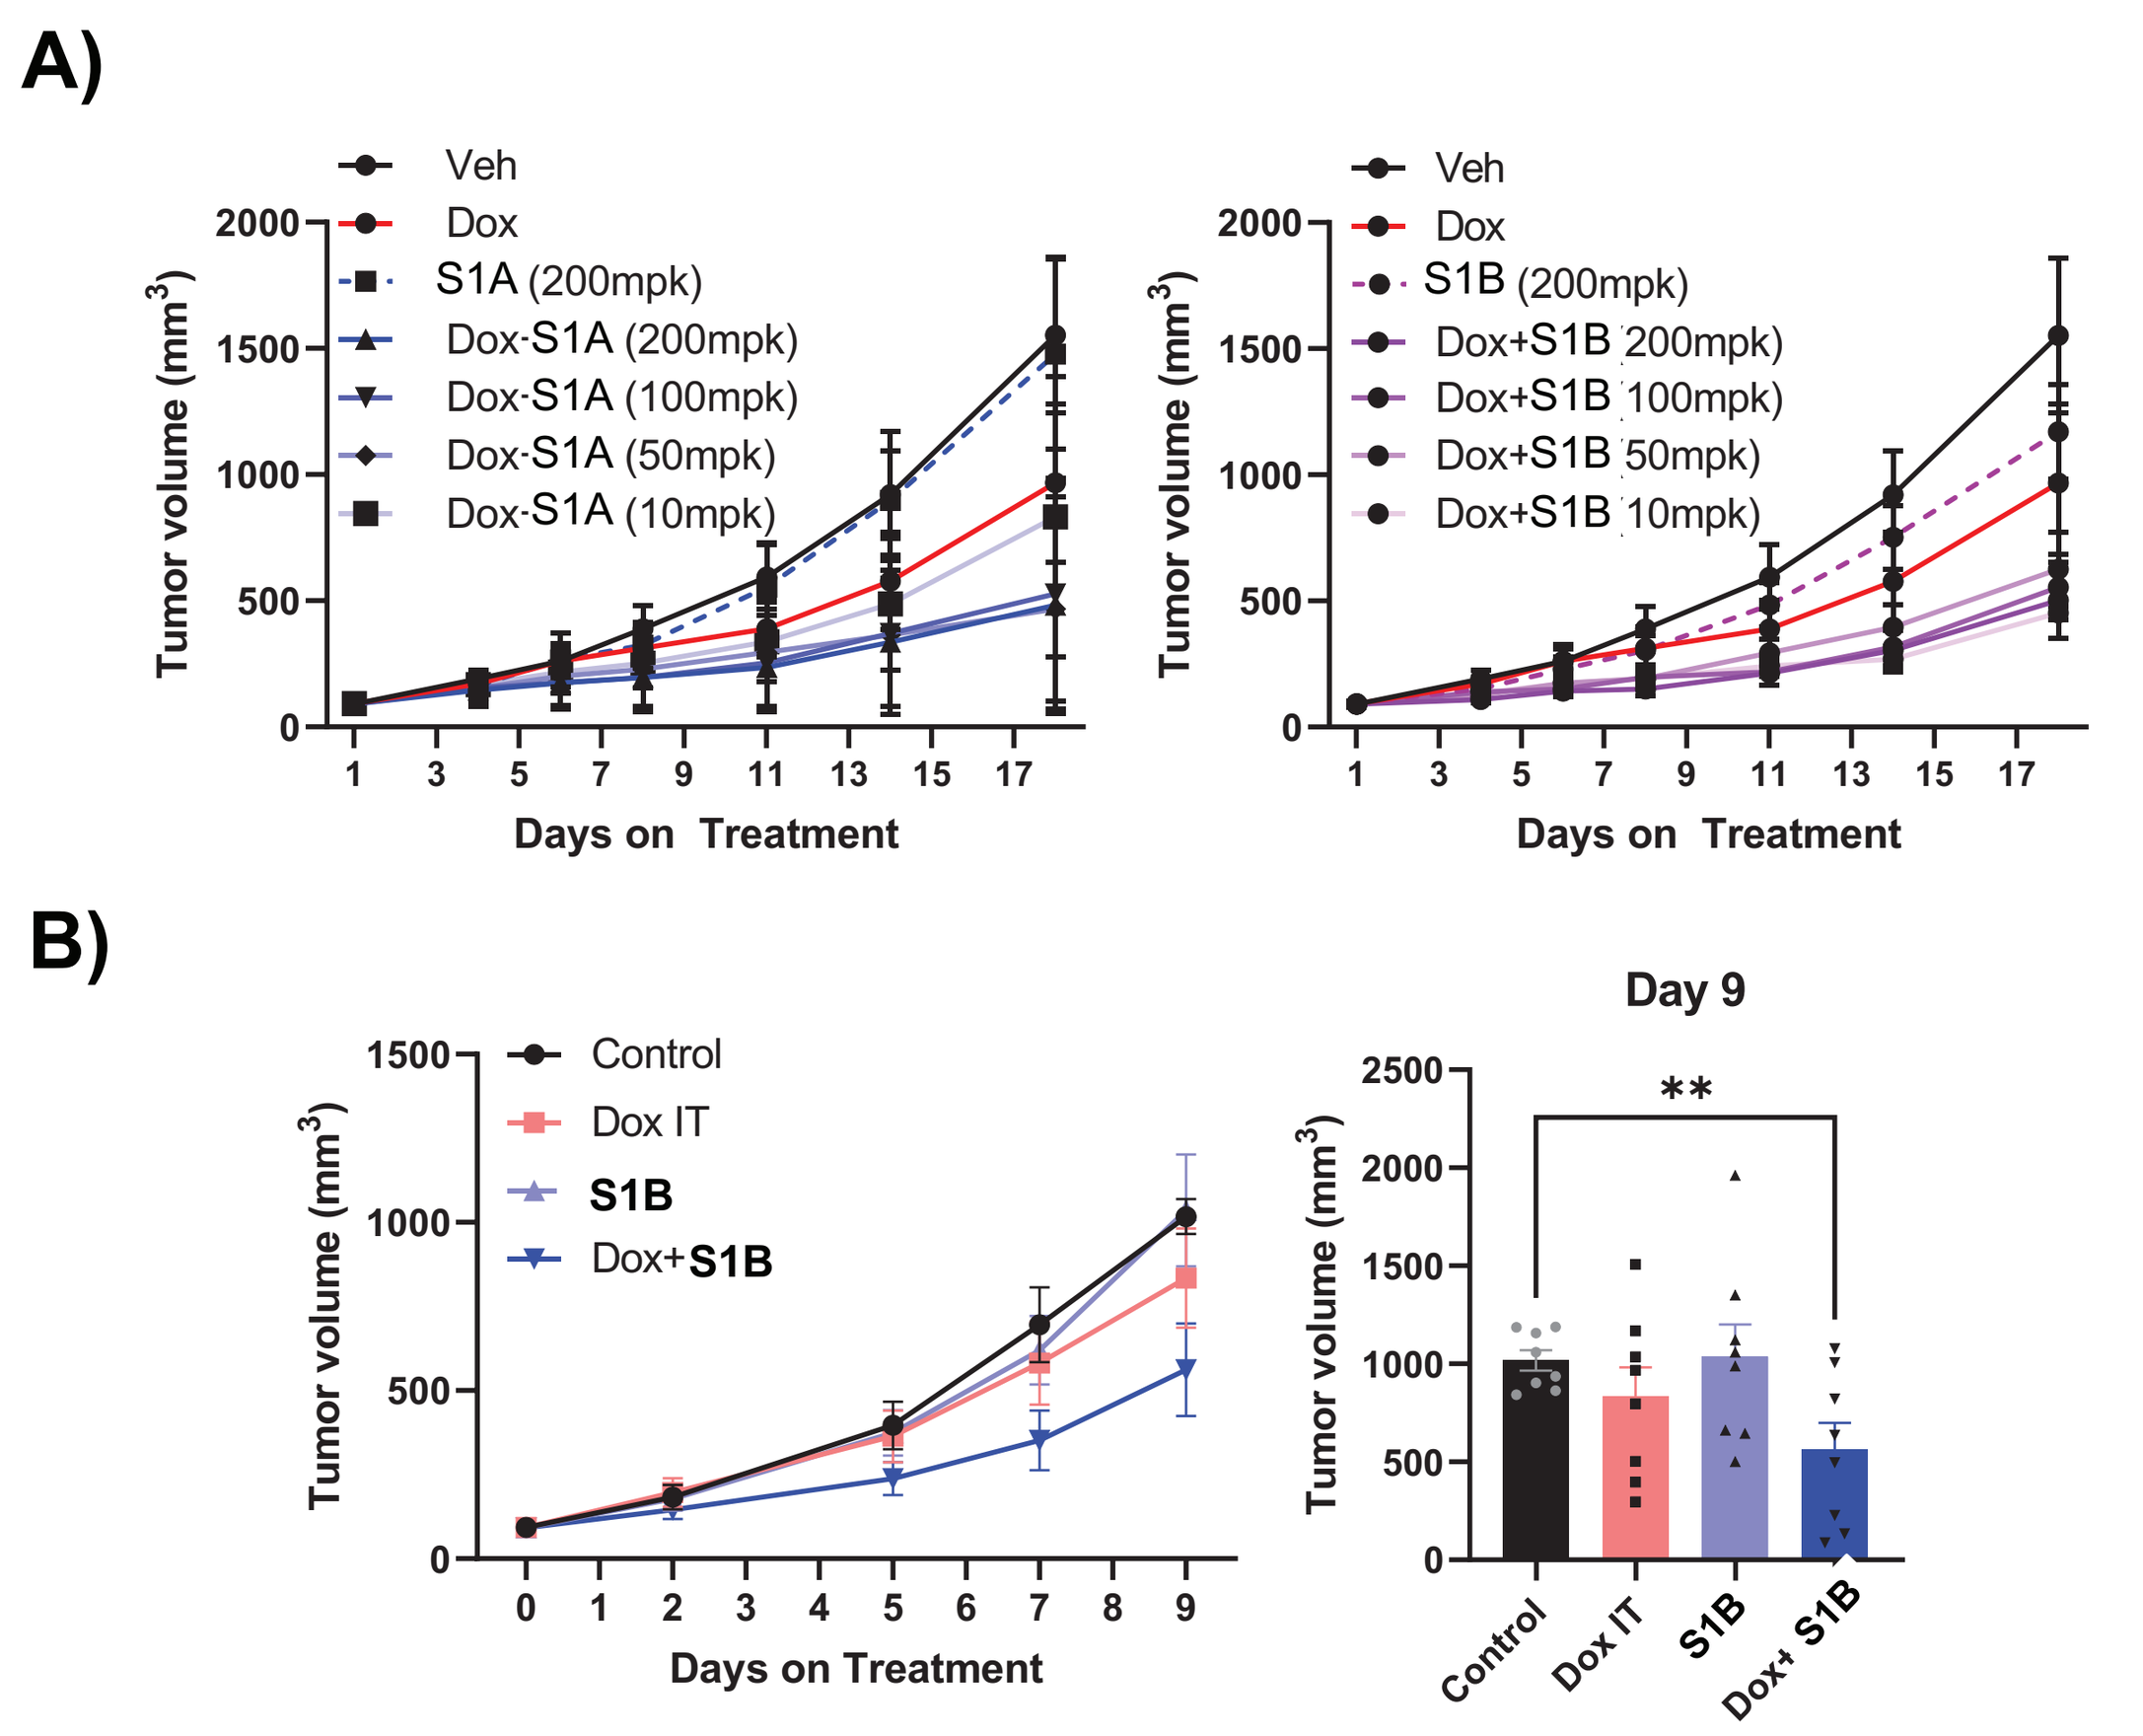
A)
Veh
Dox
S1A (200mpk)
Dox-S1A (200mpk)
Dox-S1A (100mpk)
Dox-S1A (50mpk)
Dox-S1A (10mpk)
S1B (200mpk)
Dox+S1B 200mpk)
Dox+S1B 100mpk)
Dox+S1B 50mpk)
Dox+S1B 10mpk)
Tumor volume (mm³)
Days on Treatment
B)
Control
Dox IT
S1B
Dox+S1B
Day 9
**

A) Anti-tumor efficacy correlates with TREX1 inhibitor IC_{50} value. Groups of 8 CT26 tumor-bearing Balb/c mice were treated with Vehicle, Dox on Days 7 and 14, or a subtherapeutic dose of Dox in combination with TREX1 inhibitors S1A or S1B given IP over a dose range between 10 and 200 MPK for 18 days. Tumor growth was measured every 3 days during TREX1 inhibitor therapy. Relative in vivo efficacy correlates with in vitro activity (Figure 2). **B) TREX1 inhibitors have comparable anti-tumor activity in the MC38 tumor model.** Groups of 8 C57Bl/6 mice bearing ca. 100 mm³ tumors were treated with vehicle, once with Dox, or once with Dox and 200 MPK of S1B given IP for 9 days and tumor growth was monitored. Significance was determined by Student's 2-sided t test. (**: p<0.01)

SUMMARY & NEXT STEPS

- We have developed several series of potent TREX-1 specific inhibitors
 - Docking model based on published mTREX1/DNA X-ray crystal structures strongly aligns with SAR
 - pM inhibition of human TREX1 in biochemical assays
 - Enhanced activation of STING pathway in DNA-stimulated human and mouse cells
 - Significant therapeutic anti-tumor efficacy and survival when combined with the DNA damaging agent doxorubicin
- Two features of TREX1 biology can be targeted clinically
 - Activation of cGAS/STING pathway selectively in the TME
 - Synthetic lethality partner in DNA repair mutant tumors (preliminary results not shown)
- Unique approach to selectively activate the STING pathway in the TME with a systemically administered TREX1 inhibitor
- Ongoing SAR activities with lead series compounds together with immunologic and tumor lethality MOA studies towards selection of a Development Candidate for IND-enabling activities are ongoing

TREX1 is a DNA repair enzyme

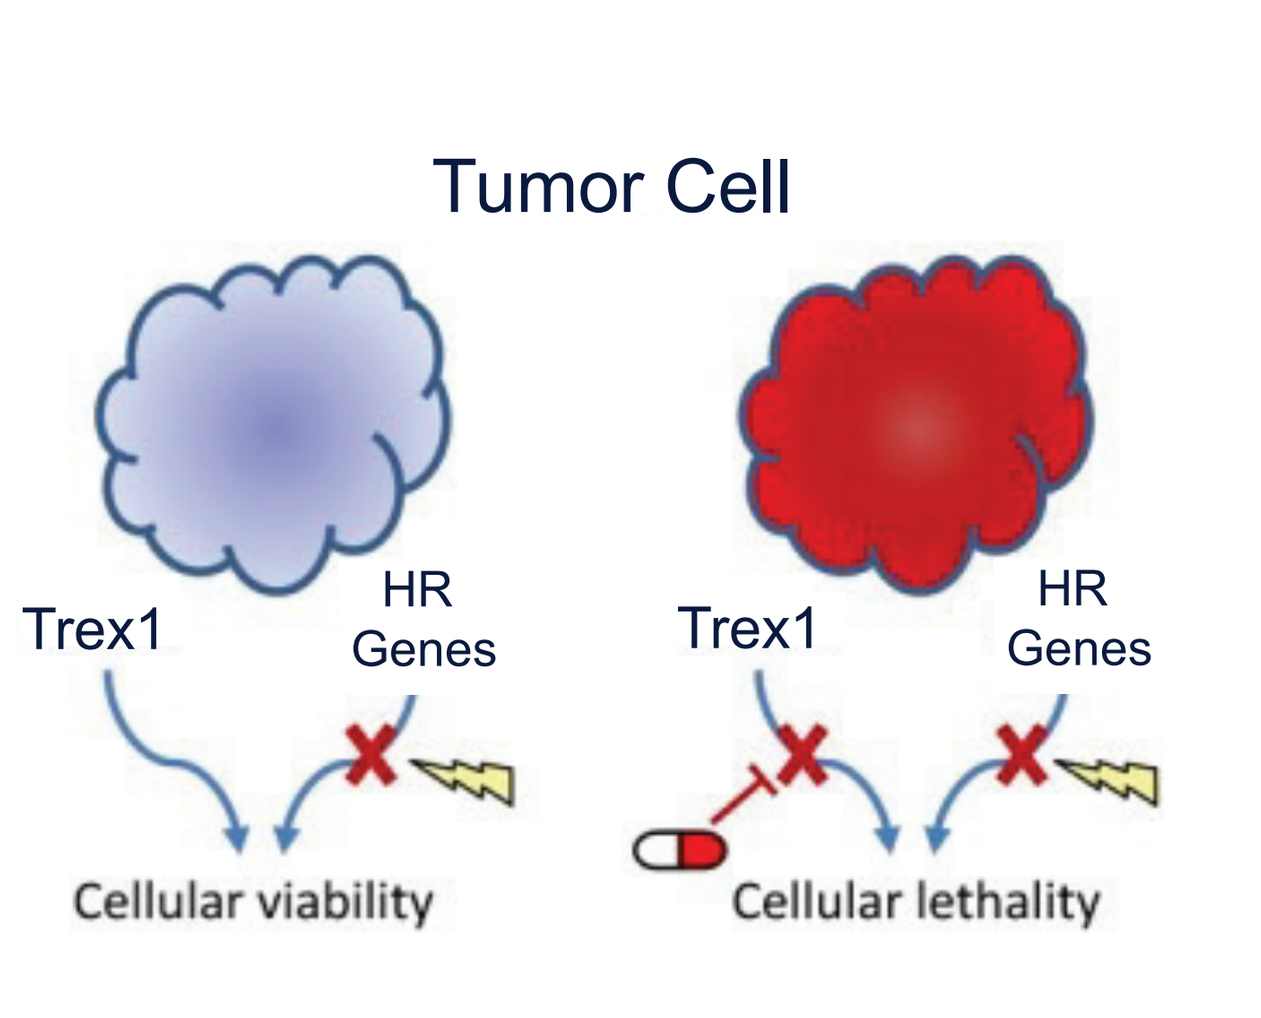
Tumor Cell
Trex1
HR Genes
Cellular viability
Trex1
HR Genes
Cellular lethality

Homologous recombination (HR) deficient tumors cells may be susceptible to TREX1 inhibition

Adopted from Thompson et. Al. Pig Cell Mel Res, 2017