758

Biomarkers associated with blockade of prostaglandin E2 signaling by TPST-1495: a novel, dual-antagonist of E-Prostanoid receptors 2 and 4

Nathan Standifer, Yonchu Jenkins, Thomas Dubensky, and Sam Whiting

Tempest Therapeutics, Brisbane, CA

BACKGROUND

Figure 1. Prostaglandin E2 (PGE2) signaling

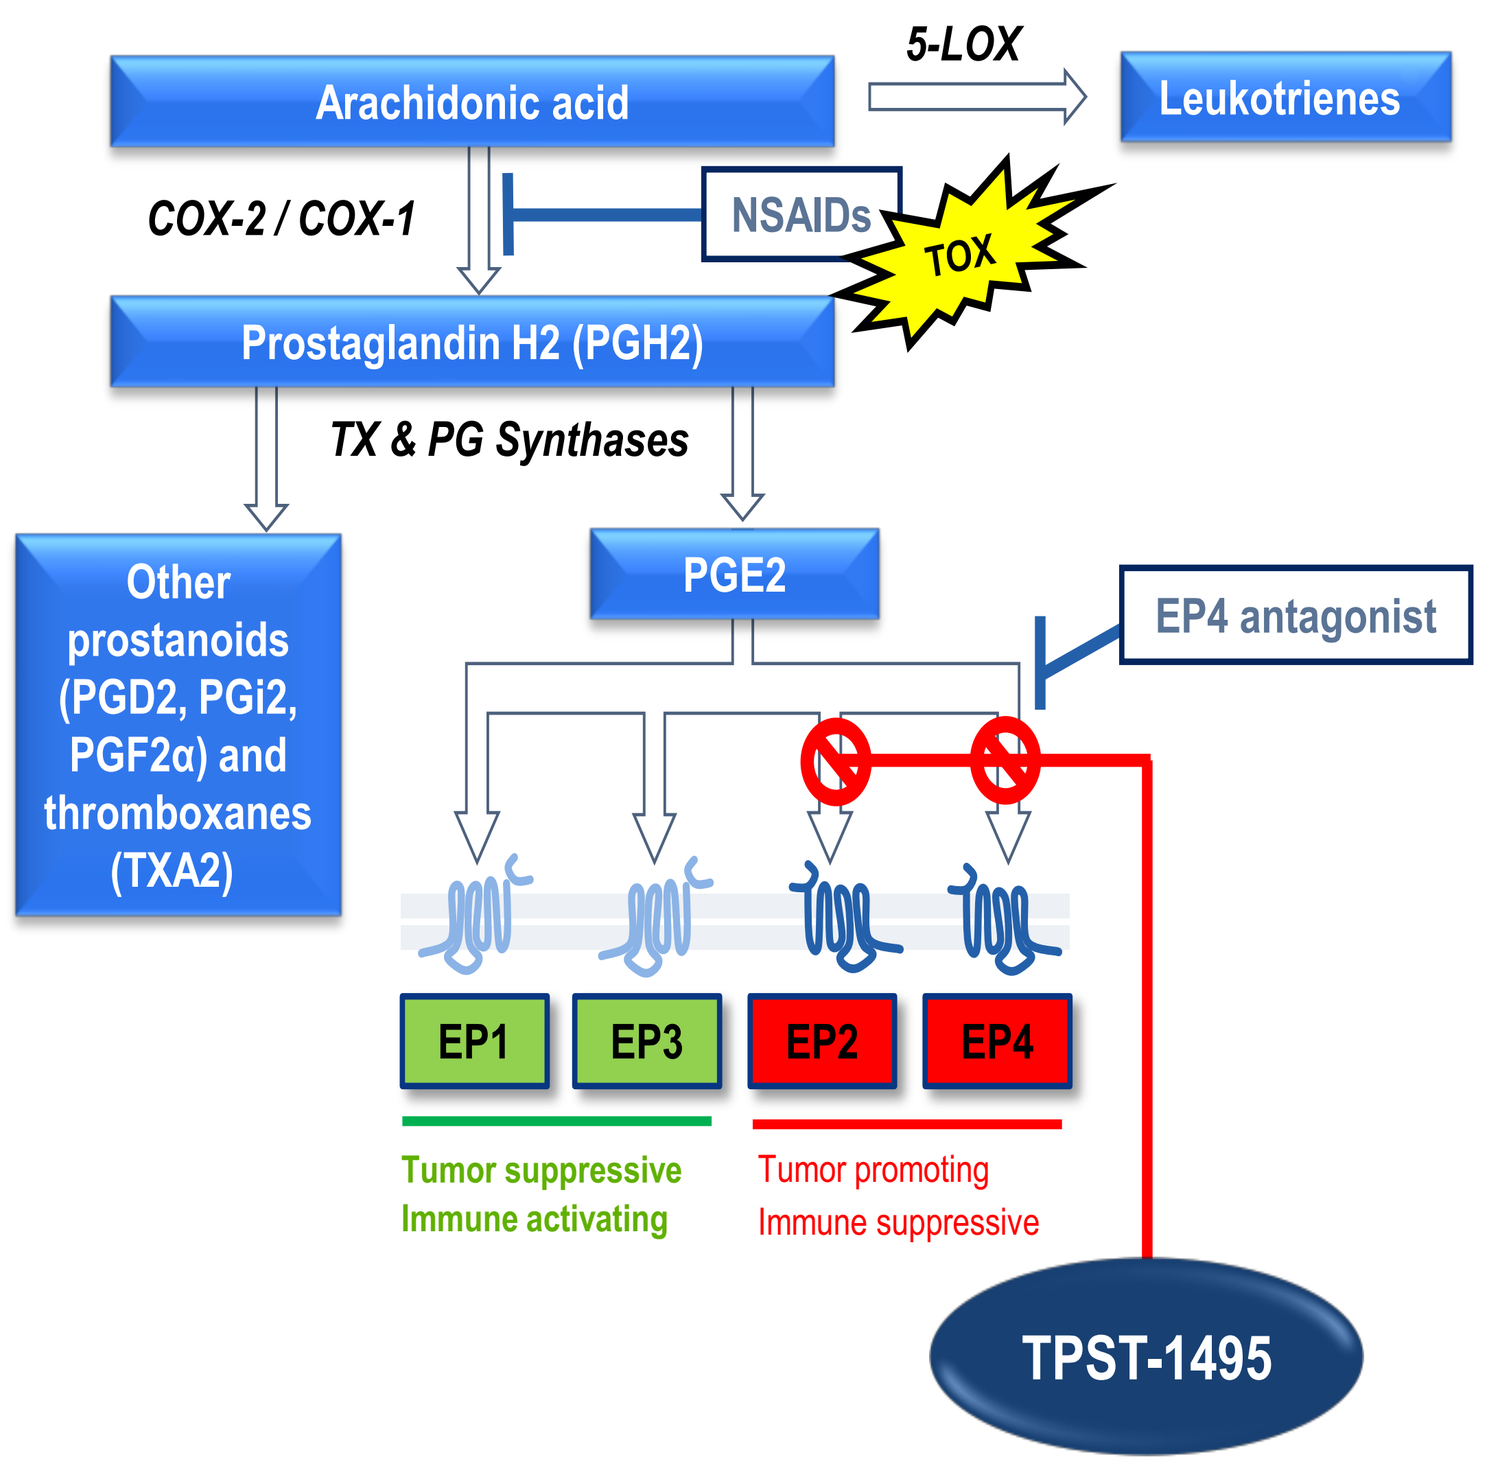
Arachidonic acid
5-LOX
Leukotrienes
COX-2 / COX-1
NSAIDs
TOX
Prostaglandin H2 (PGH2)
TX & PG Synthases
Other prostanoids (PGD2, PGi2, PGF2α) and thromboxanes (TXA2)
PGE2
EP4 antagonist
EP1
EP3
EP2
EP4
Tumor suppressive
Immune activating
Tumor promoting
Immune suppressive
TPST-1495

Prostaglandin E2 (PGE2) Signaling

- COX-2/PGE2 implicated in development and progression of multiple cancer types[1,2]
- Limitations of therapeutic COX inhibition include blocking of beneficial receptors and association with cardiovascular toxicity due to alterations in prostanoids, thromboxane, and leukotriene levels
- PGE2 elevated in tumor microenvironment
 - Mediates tumor proliferation and angiogenesis
 - Facilitates immune evasion via inhibiting myeloid and lymphoid cell effector functions and inducing immune suppressive populations
- PGE2 immune suppressive function associated with binding to E-prostanoid receptors 2 and 4 (**EP2** & **EP4**) and downstream signaling characterized by cAMP production

TPST-1495: A Novel, Specific, Potent, Dual-Antagonist of EP2 and EP4

- Anti-tumor activity, reduced immune suppression, and activated immune effector responses in syngeneic tumor models
- Dual EP2/EP4 inhibition is more active than single EP2 or EP4 antagonists or COX-2 inhibition
- A first-in-human Phase I trial of TPST-1495 as a single agent and in combination with pembrolizumab in 74 patients with treatment-refractory advanced solid tumors showed[3]:
 - Disease control rates of 37.5% for monotherapy and 40.9% for combination, with tumor shrinkage and prolonged stable disease (SD), including a confirmed partial response in a combination therapy patient with microsatellite-stable (MSS) colorectal carcinoma (CRC), an indication not normally responsive to immuno-oncology therapy
 - Manageable safety profile with once daily (QD) dosing; related AEs were predominantly grade 1-2 (no grade 4-5), gastrointestinal in nature, and on-target for the prostaglandin pathway

OBJECTIVE

To characterize TPST-1495 pharmacodynamic activities by quantifying biomarkers associated with EP2 and EP4 blockade in pre- and post-treatment specimens, testing the hypotheses that TPST-1495 can:

1) Reverse PGE2-mediated immune suppression, measured by lipopolysaccharide (LPS)-stimulated production of TNF-α in whole blood samples of patients
2) Increase systemic levels of PGE2, quantified with urinary tetranor PGE2 metabolite (PGEM) as a surrogate marker
3) Increase intra-tumoral CD8⁺ and granzyme B⁺ T cells

METHODS

Study Design (NCT04344795)

- TPST-1495 was dosed at 15, 25 or 50 mg BID or QD (continuous or days 1-5 every 7 days) as monotherapy or QD continuously in combination with pembrolizumab 200 mg IV q3 weeks (**Figure 2**)
- Key Eligibility Criteria
 - Metastatic or unresectable cancer w/ no remaining standard therapy known to confer clinical benefit
 - ECOG PS 0 or 1
 - Measurable disease (RECIST v1.1)
 - Excluded if intolerant to NSAIDs (including bleeding, ulcer), on anticoagulation therapy or at increased risk for bleeding, or experienced intolerable or unresolved immune-related adverse events (AEs) with prior checkpoint inhibitor therapy
- Study Objectives

 1°: Safety & tolerability, MTD, RP2D & schedule

 2°: Anti-tumor activity, pharmacokinetics (PK)

 Exploratory: Pharmacodynamics

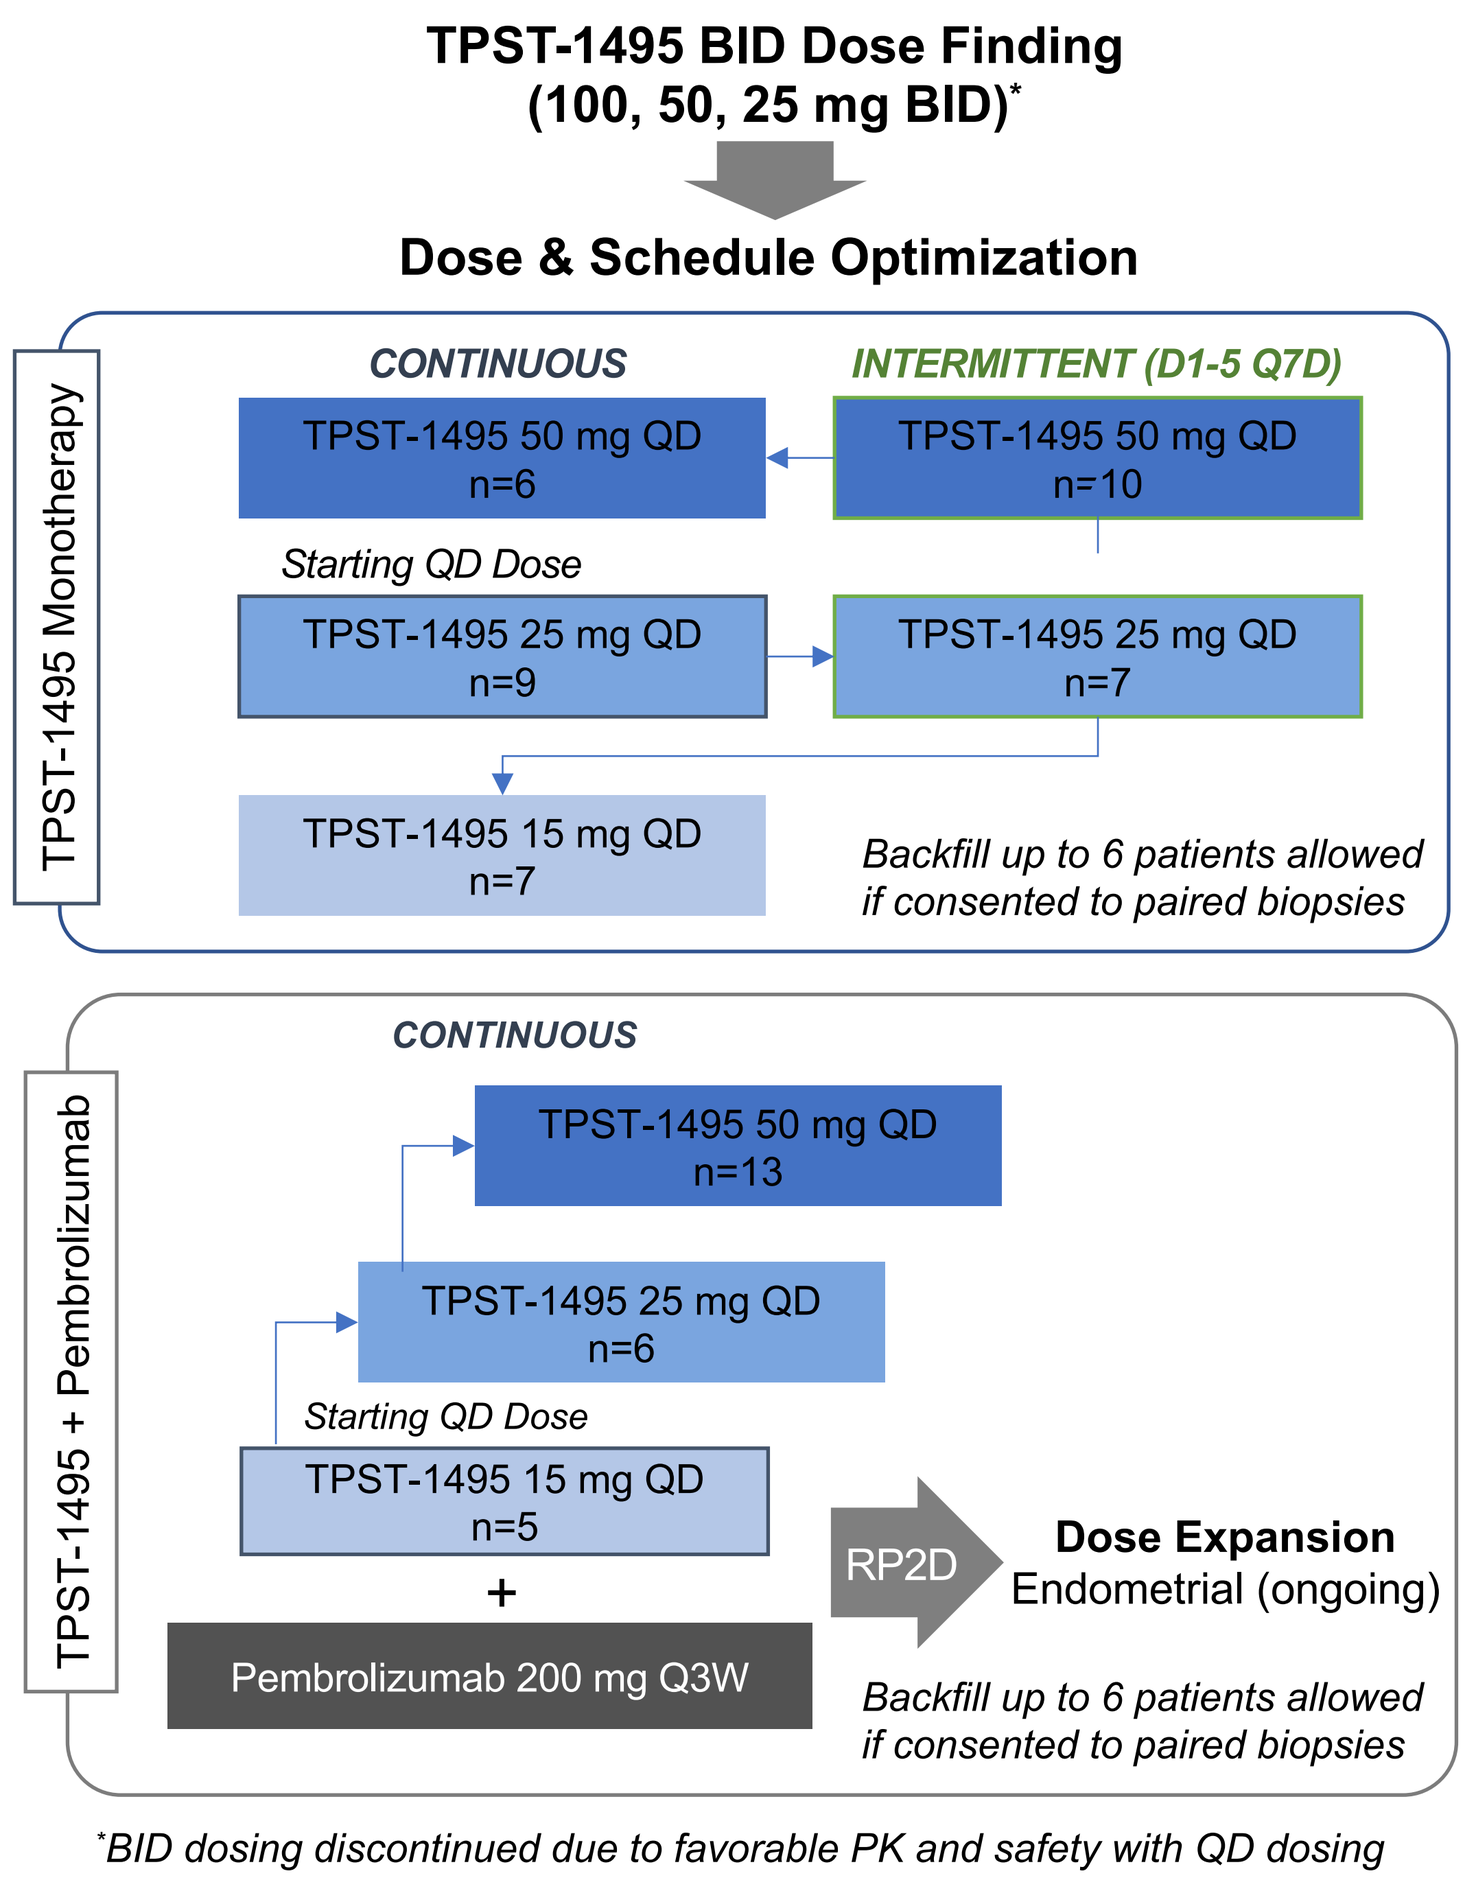
TPST-1495 BID Dose Finding
(100, 50, 25 mg BID)*
Dose & Schedule Optimization
TPST-1495 Monotherapy
CONTINUOUS
INTERMITTENT (D1-5 Q7D)
TPST-1495 50 mg QD n=6
TPST-1495 50 mg QD n=10
Starting QD Dose
TPST-1495 25 mg QD n=9
TPST-1495 25 mg QD n=7
TPST-1495 15 mg QD n=7
Backfill up to 6 patients allowed if consented to paired biopsies
TPST-1495 + Pembrolizumab
CONTINUOUS
TPST-1495 50 mg QD n=13
TPST-1495 25 mg QD n=6
Starting QD Dose
TPST-1495 15 mg QD n=5
+
Pembrolizumab 200 mg Q3W
RP2D
Dose Expansion
Endometrial (ongoing)
Backfill up to 6 patients allowed if consented to paired biopsies
*BID dosing discontinued due to favorable PK and safety with QD dosing

Figure 2. Study schema

Pharmacokinetics

- An HPLC-MS/MS method was developed and validated to determine concentrations of TPST-1495 in human K_2-EDTA plasma (Alturas Analytics, Inc.)
- The peak area of the product ion of TPST-1495 was measured against the peak area of the product ion of an internal standard

Inhibition of PGE2-mediated immune suppression

- Whole blood samples collected in K_2-EDTA tubes prior to and 2- or 4-hours following the first dose of TPST-1495 were cultured with 150 nM LPS in the presence or absence of PGE2 overnight at 37°C
- Cell-free supernatants were collected, and TNF-α was quantified by ELISA (Invitrogen, Inc.)
- Data were expressed as percentage of TNF-α increase in the presence of PGE2, calculated as baseline-subtracted, LPS-induced, post-dose TNF-α concentration divided by baseline LPS-induced, pre-dose TNF-α concentration

PGEM assay

- An HPLC-MS/MS assay was developed and validated to determine concentrations of tetranor PGEM in urine samples stored at -80°C (Alturas Analytics, Inc.)
- PGEM levels were expressed as a function of creatinine levels in the same samples

Immunohistochemistry (IHC)

- Biopsies collected from primary or metastatic lesions were formalin-fixed and paraffin embedded prior to performing IHC for specific markers for T cells, granzyme B (GrB), macrophages, and COX-2-expressing cells, using validated assays (CellCarta, Inc.)
- For available paired biopsies, on-treatment specimens were collected between study days 43 and 64

Data analysis

- Wilcoxon's pair-wise analysis was used to compare TNF-α or PGEM levels among patients at different dose levels (α=0.05)

RESULTS

Pharmacokinetics

- Linear PK (slope = 0.99 [95% CI: 0.57, 1.09]), with dose-dependent increase in exposure
- No changes in PK parameters with addition of pembrolizumab

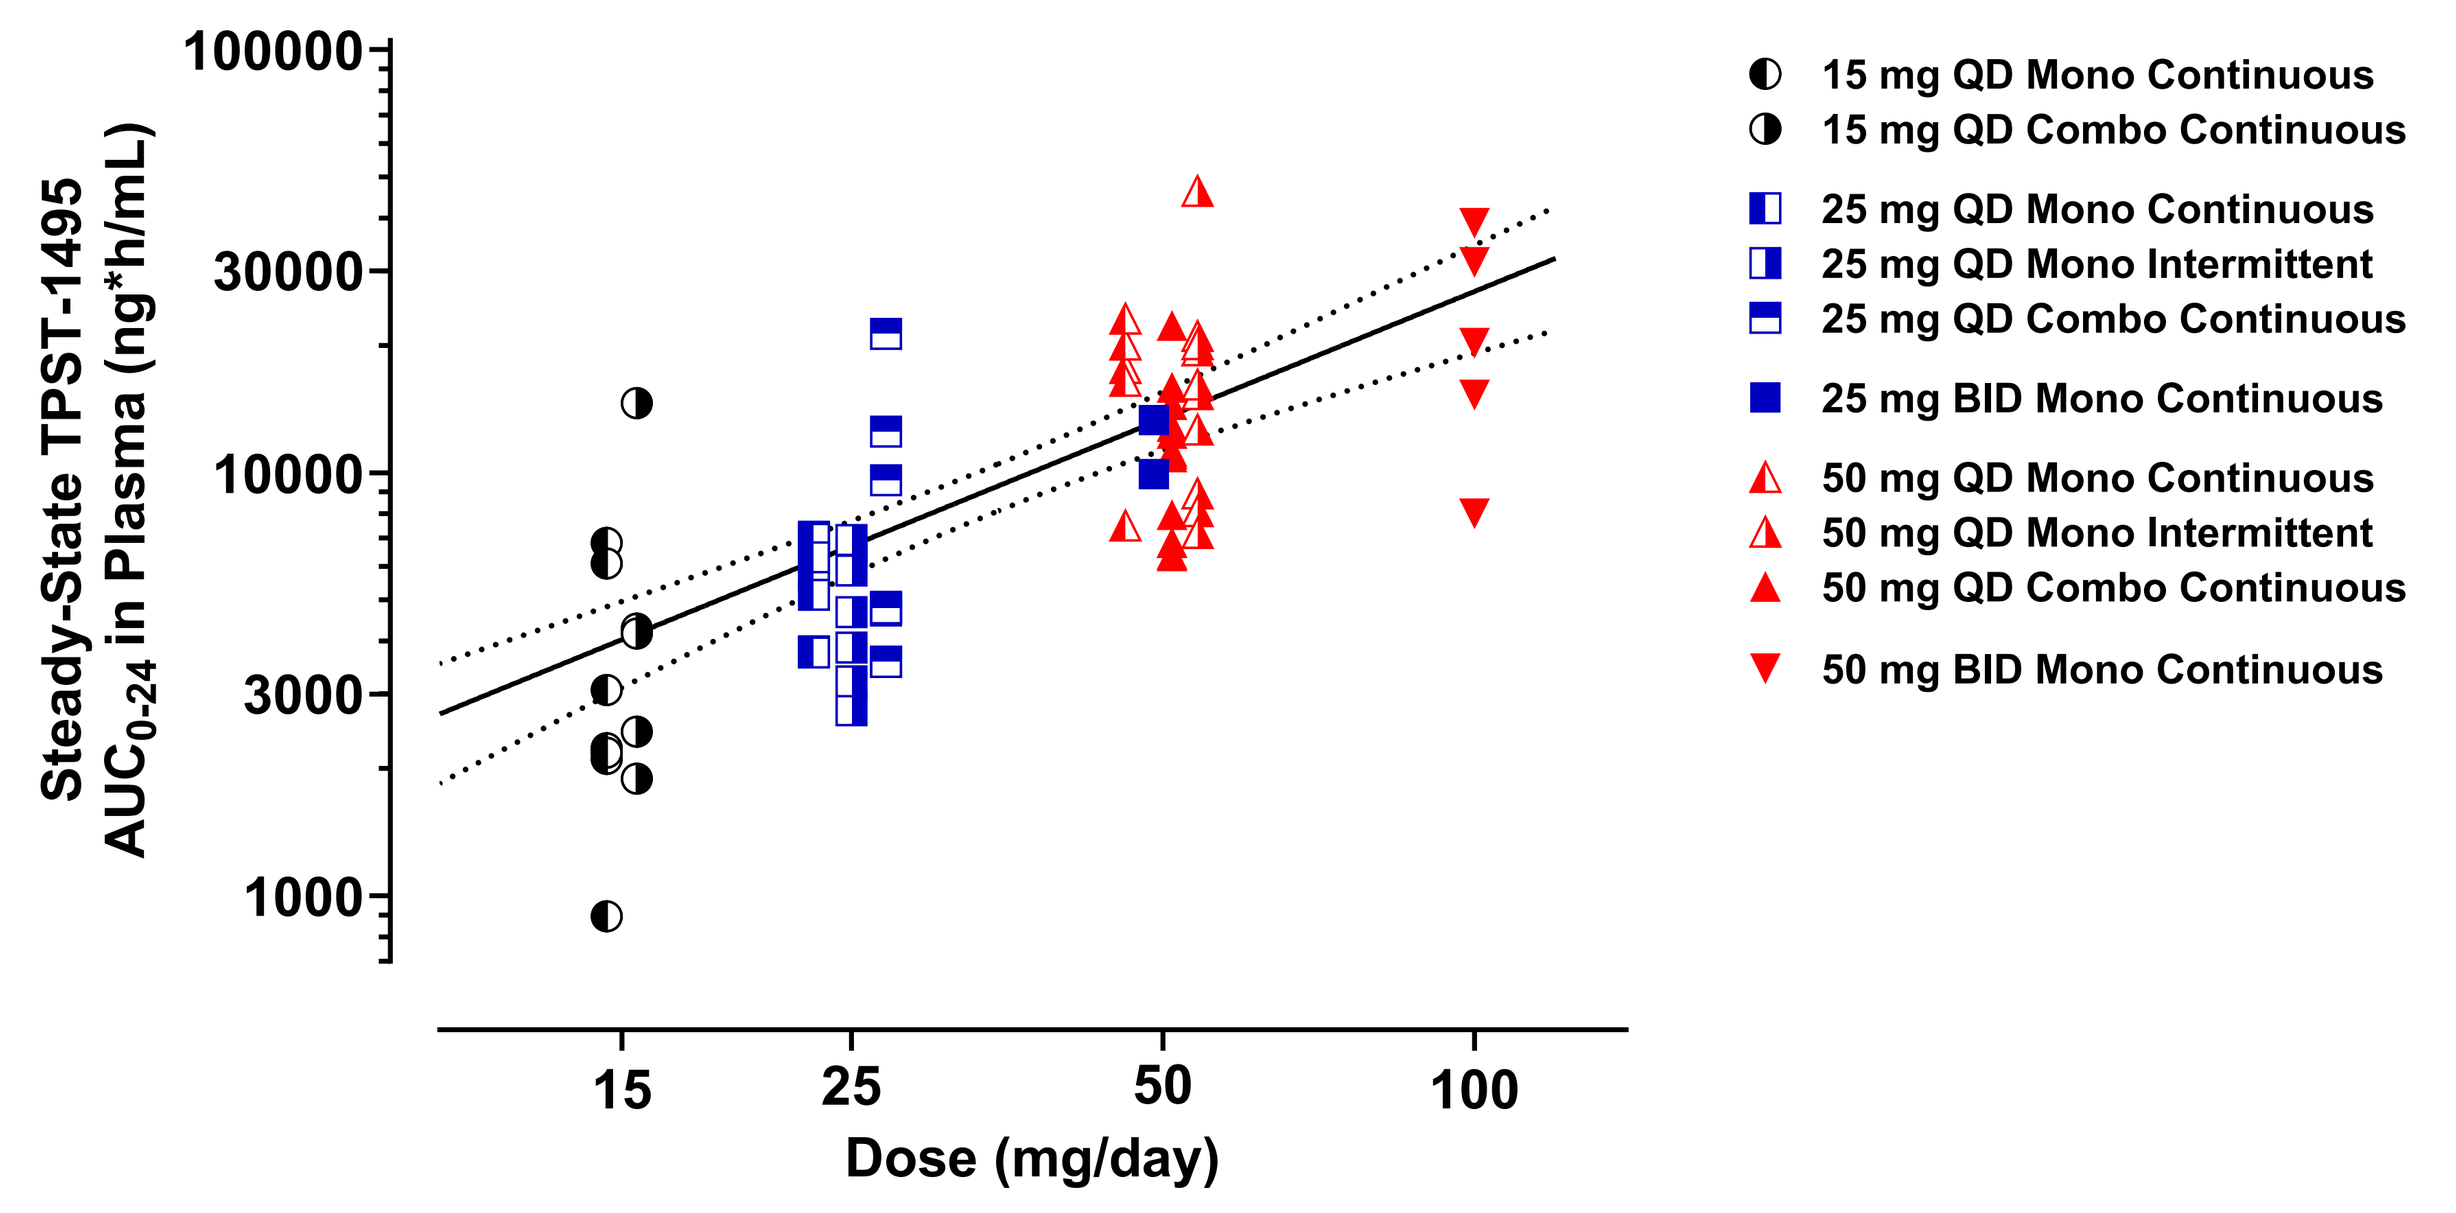
Steady-State TPST-1495 AUC_{0-24} in Plasma (ng*h/mL)
100000
30000
10000
3000
1000
15
25
50
100
Dose (mg/day)
15 mg QD Mono Continuous
15 mg QD Combo Continuous
25 mg QD Mono Continuous
25 mg QD Mono Intermittent
25 mg QD Combo Continuous
25 mg BID Mono Continuous
50 mg QD Mono Continuous
50 mg QD Mono Intermittent
50 mg QD Combo Continuous
50 mg BID Mono Continuous

Figure 3. Relationship between dose and AUC at steady state.

Pharmacodynamics

Reversal of PGE2-Induced Immune Suppression with TPST-1495

- Whole blood samples collected prior to and 2- or 4-hours following TPST-1495 were incubated with LPS + PGE2 overnight before measuring TNF-α secretion
- Patients receiving TPST-1495 at 25 or 50 mg exhibited statistically higher median TNF-α recovery from PGE2-mediated suppression than those receiving 15 mg (*P* < 0.05 by Wilcoxon's pair-wise analysis) (**Figure 4**)

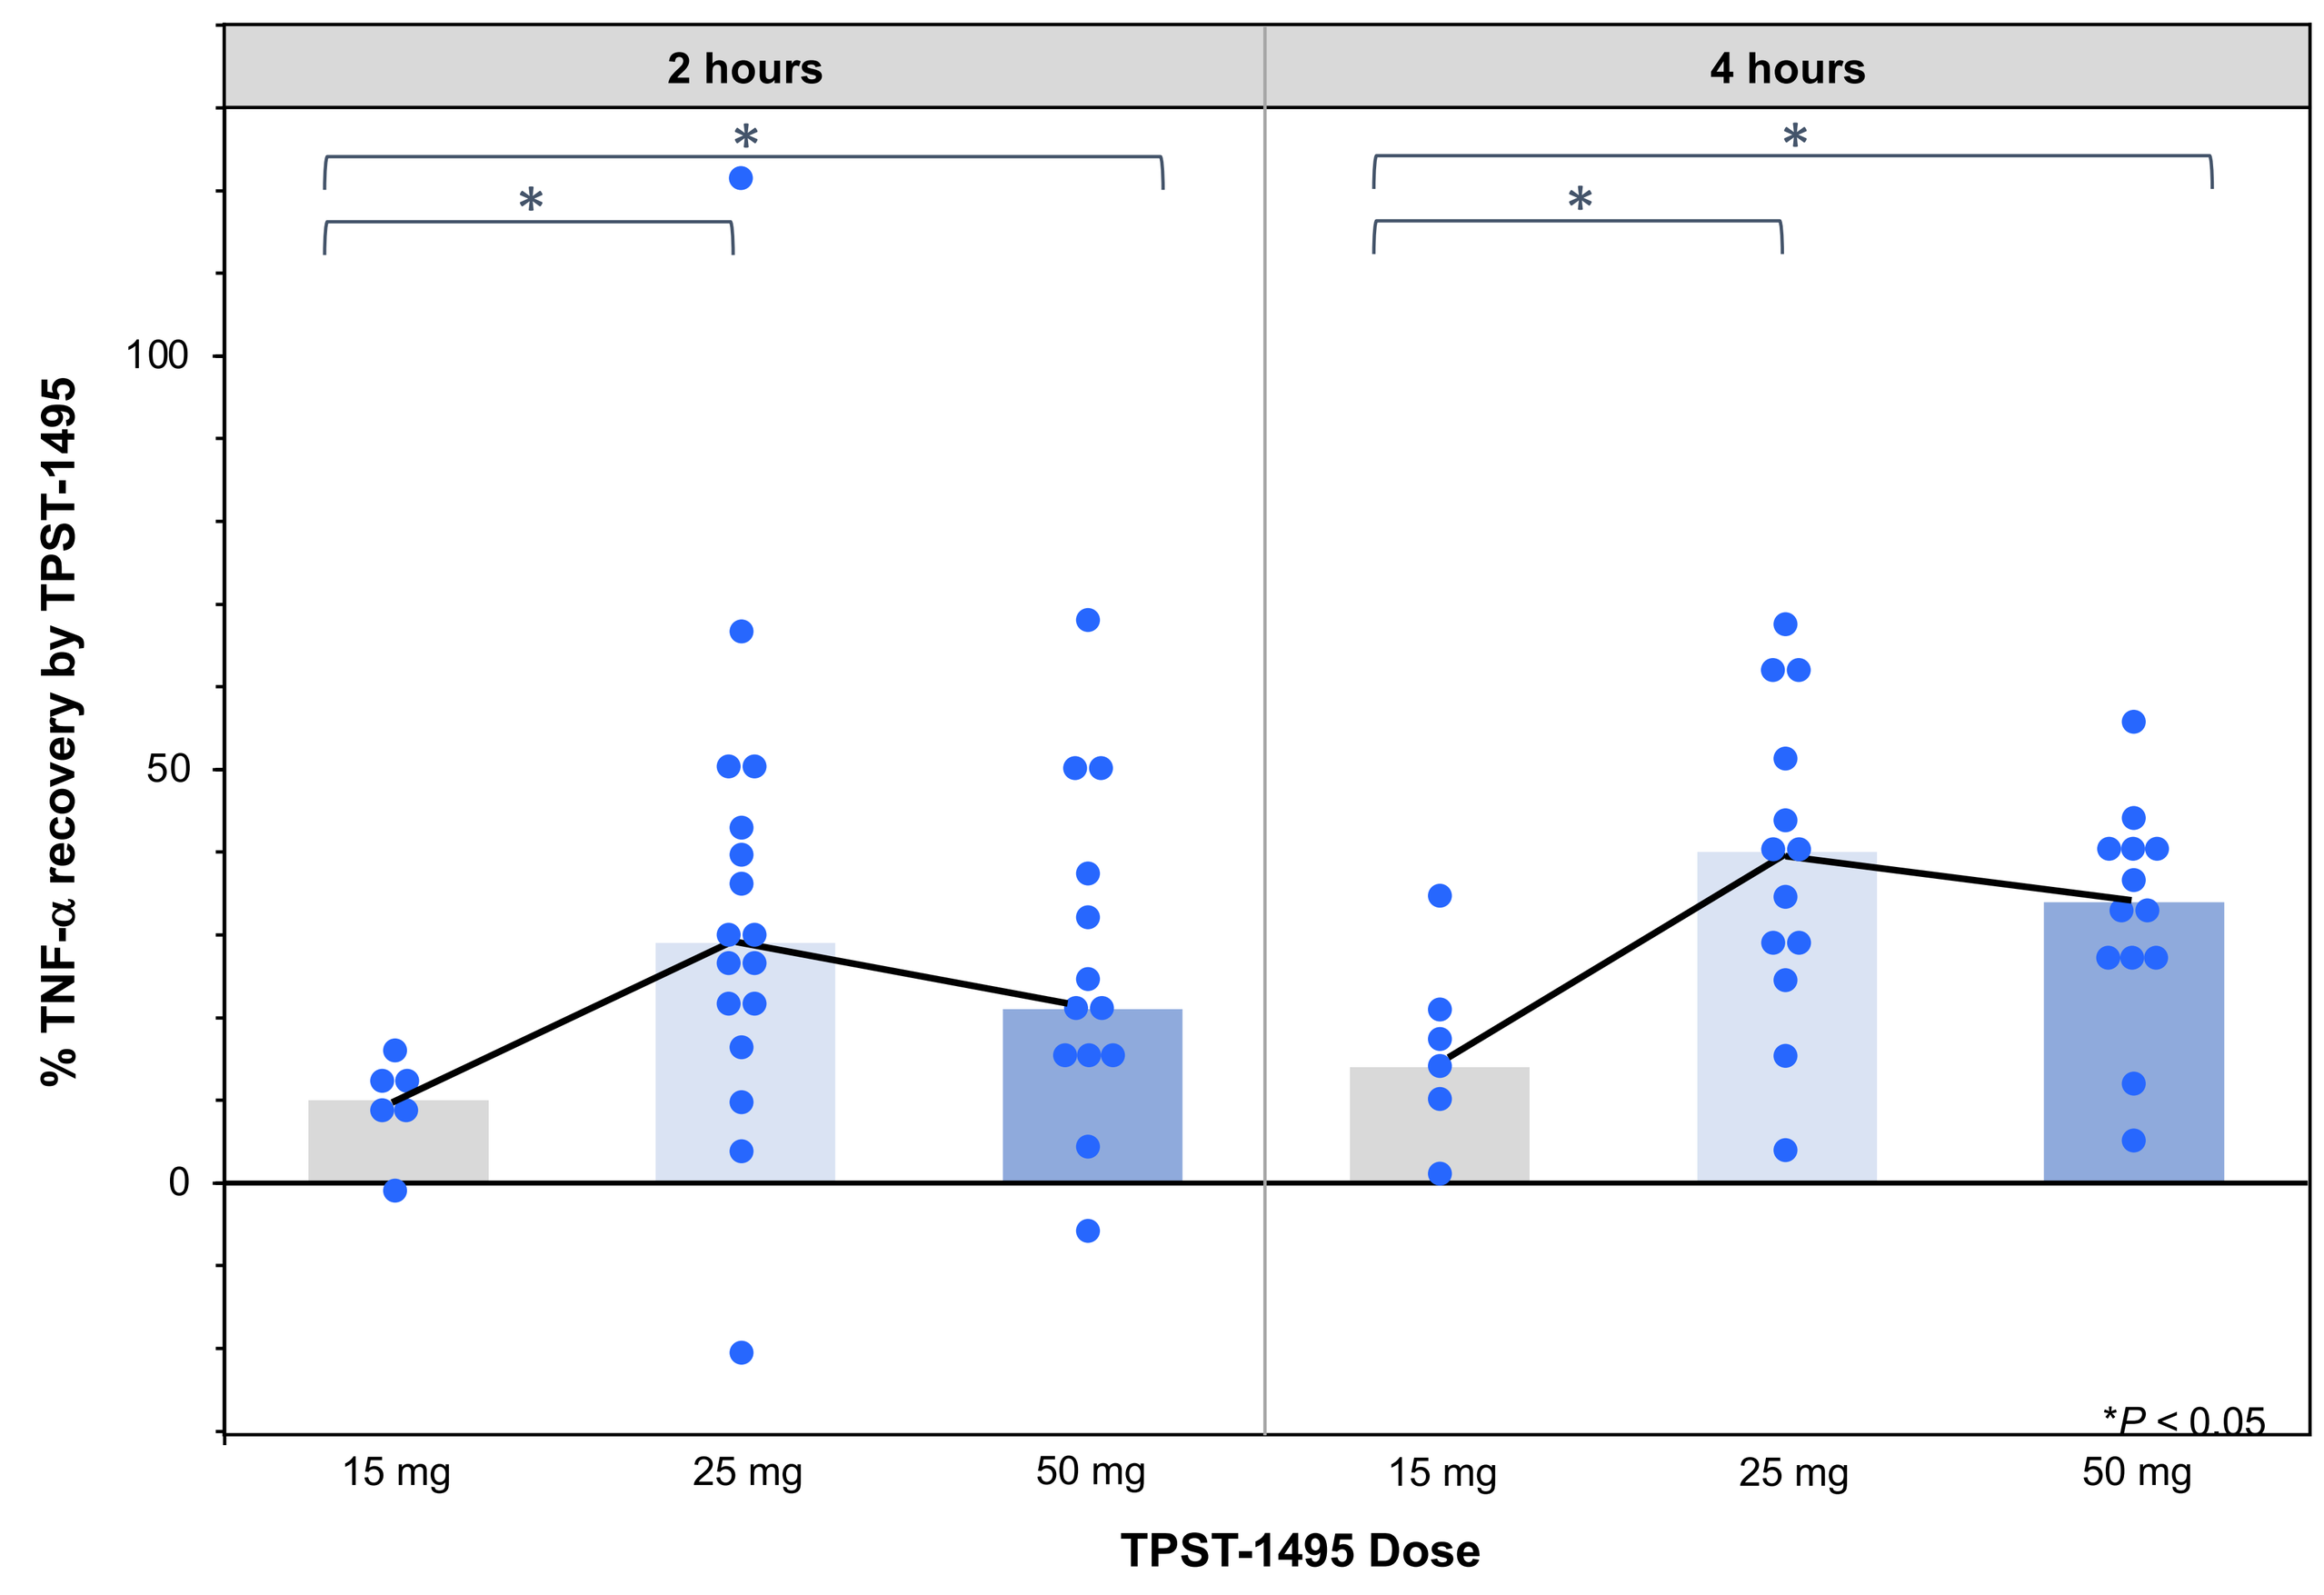
2 hours
4 hours
*
% TNF-α recovery by TPST-1495
100
50
0
15 mg
25 mg
50 mg
TPST-1495 Dose
*P < 0.05

Figure 4. Reversal of PGE2-induced immune suppression with TPST-1495
Bars represent median % TNF-α recovery. Data from patients treated with TPST-1495 as monotherapy

Elevated PGEM Following TPST-1495 Dosing

- Patients treated with TPST-1495 at the 25 or 50 mg doses demonstrated significant elevations in baseline-normalized urinary PGEM (ng/mg creatinine) above those receiving 15 mg on day 2 (*P* < 0.05 by Wilcoxon's paired test), with trends of dose-dependent increases on days 8 and 22 (**Figure 5**)

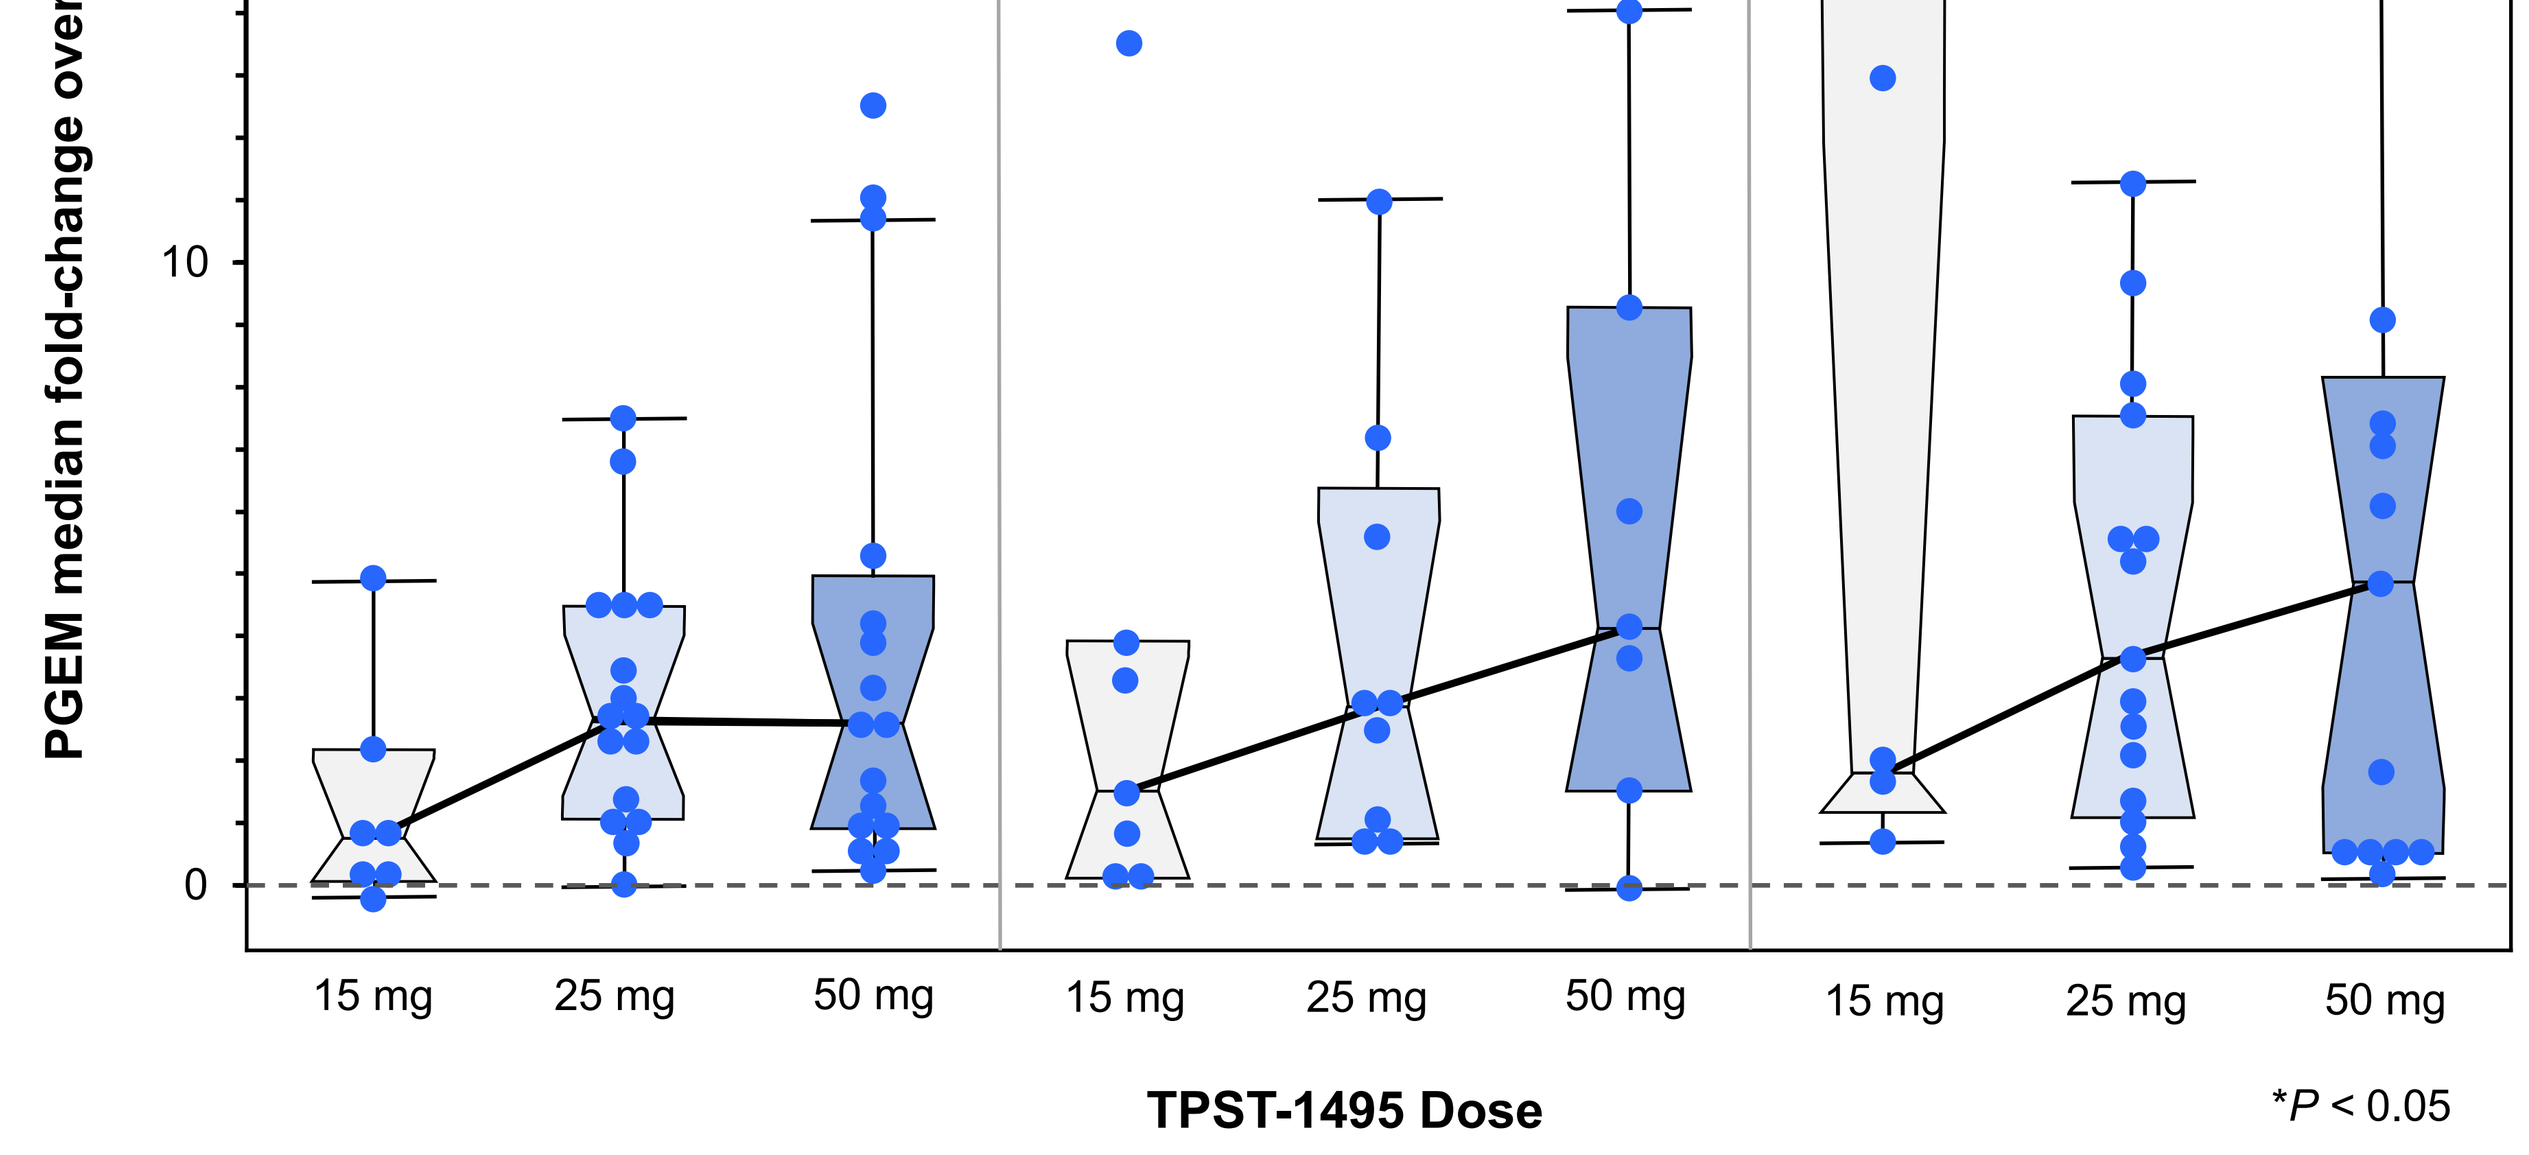
Day 2
Day 8
Day 22
*
PGEM median fold-change over baseline
20
10
0
15 mg
25 mg
50 mg
TPST-1495 Dose
*P < 0.05

Figure 5. Elevation in urinary PGEM over time
Data from patients treated with TPST-1495 as monotherapy

Biomarkers

Baseline COX-2 Expression

- COX-2 is a key driver of PGE2 biosynthesis and associated with poor cancer prognosis
- IHC analysis revealed COX-2 expression levels >10% in 3 of 11 patients with baseline biopsies (**Table 1**), including two patients with best objectives responses of stable disease

Table 1. Percent COX-2 positive expression at baseline by patient

Patient	Cancer Type	% COX-2-positive
26[a]	**Oropharyngeal SCC**	**100**
02	**Endometrial**	**40**
63[a]	**Endometrial**	**15**
18	Pancreatic	5
03	Anal SCC	3
44	MSS CRC	3
19	CRC	0
53	CRC	0
40	CRC	0
11	Prostate	0
12	Rectal	0

CRC, colorectal carcinoma; SCC, squamous cell carcinoma
[a]Best objective response = stable disease

Case Study: Endometrial Cancer Patient 63

- One of the high COX-2-expressing patients was a 78-year-old patient with Stage IV metastatic MSS endometrial cancer and lung metastasis, with 5 prior therapies in the metastatic setting[3]
- Treatment with TPST-1495 + pembrolizumab:
 - Tumor shrinkage of -22% (**Figure 6**)
 - 2-fold increases in tumor-infiltrating CD8+ T cells in an on-treatment biopsy (**Figure 7**)
 - Increase in granzyme B⁺ CD8⁺ T cells from 1% to 4% after treatment (data not shown)

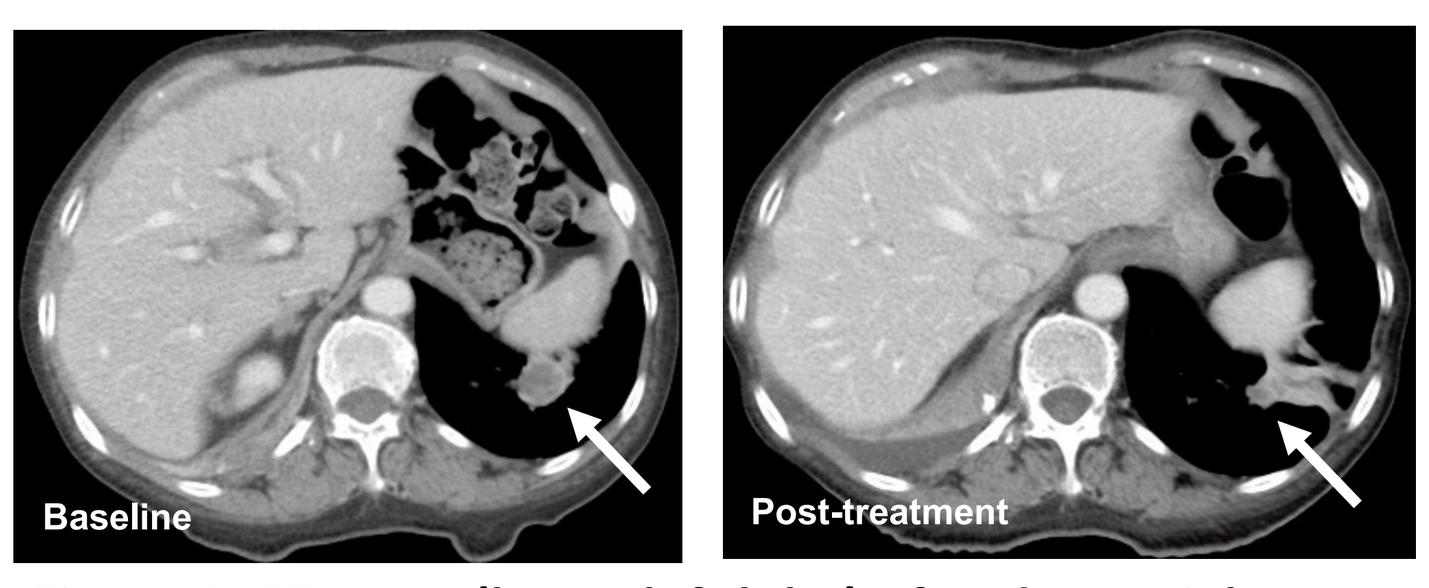
Baseline
Post-treatment

Figure 6. CT scans (lower left lobe) of endometrial patient 63 at baseline and after 4 cycles

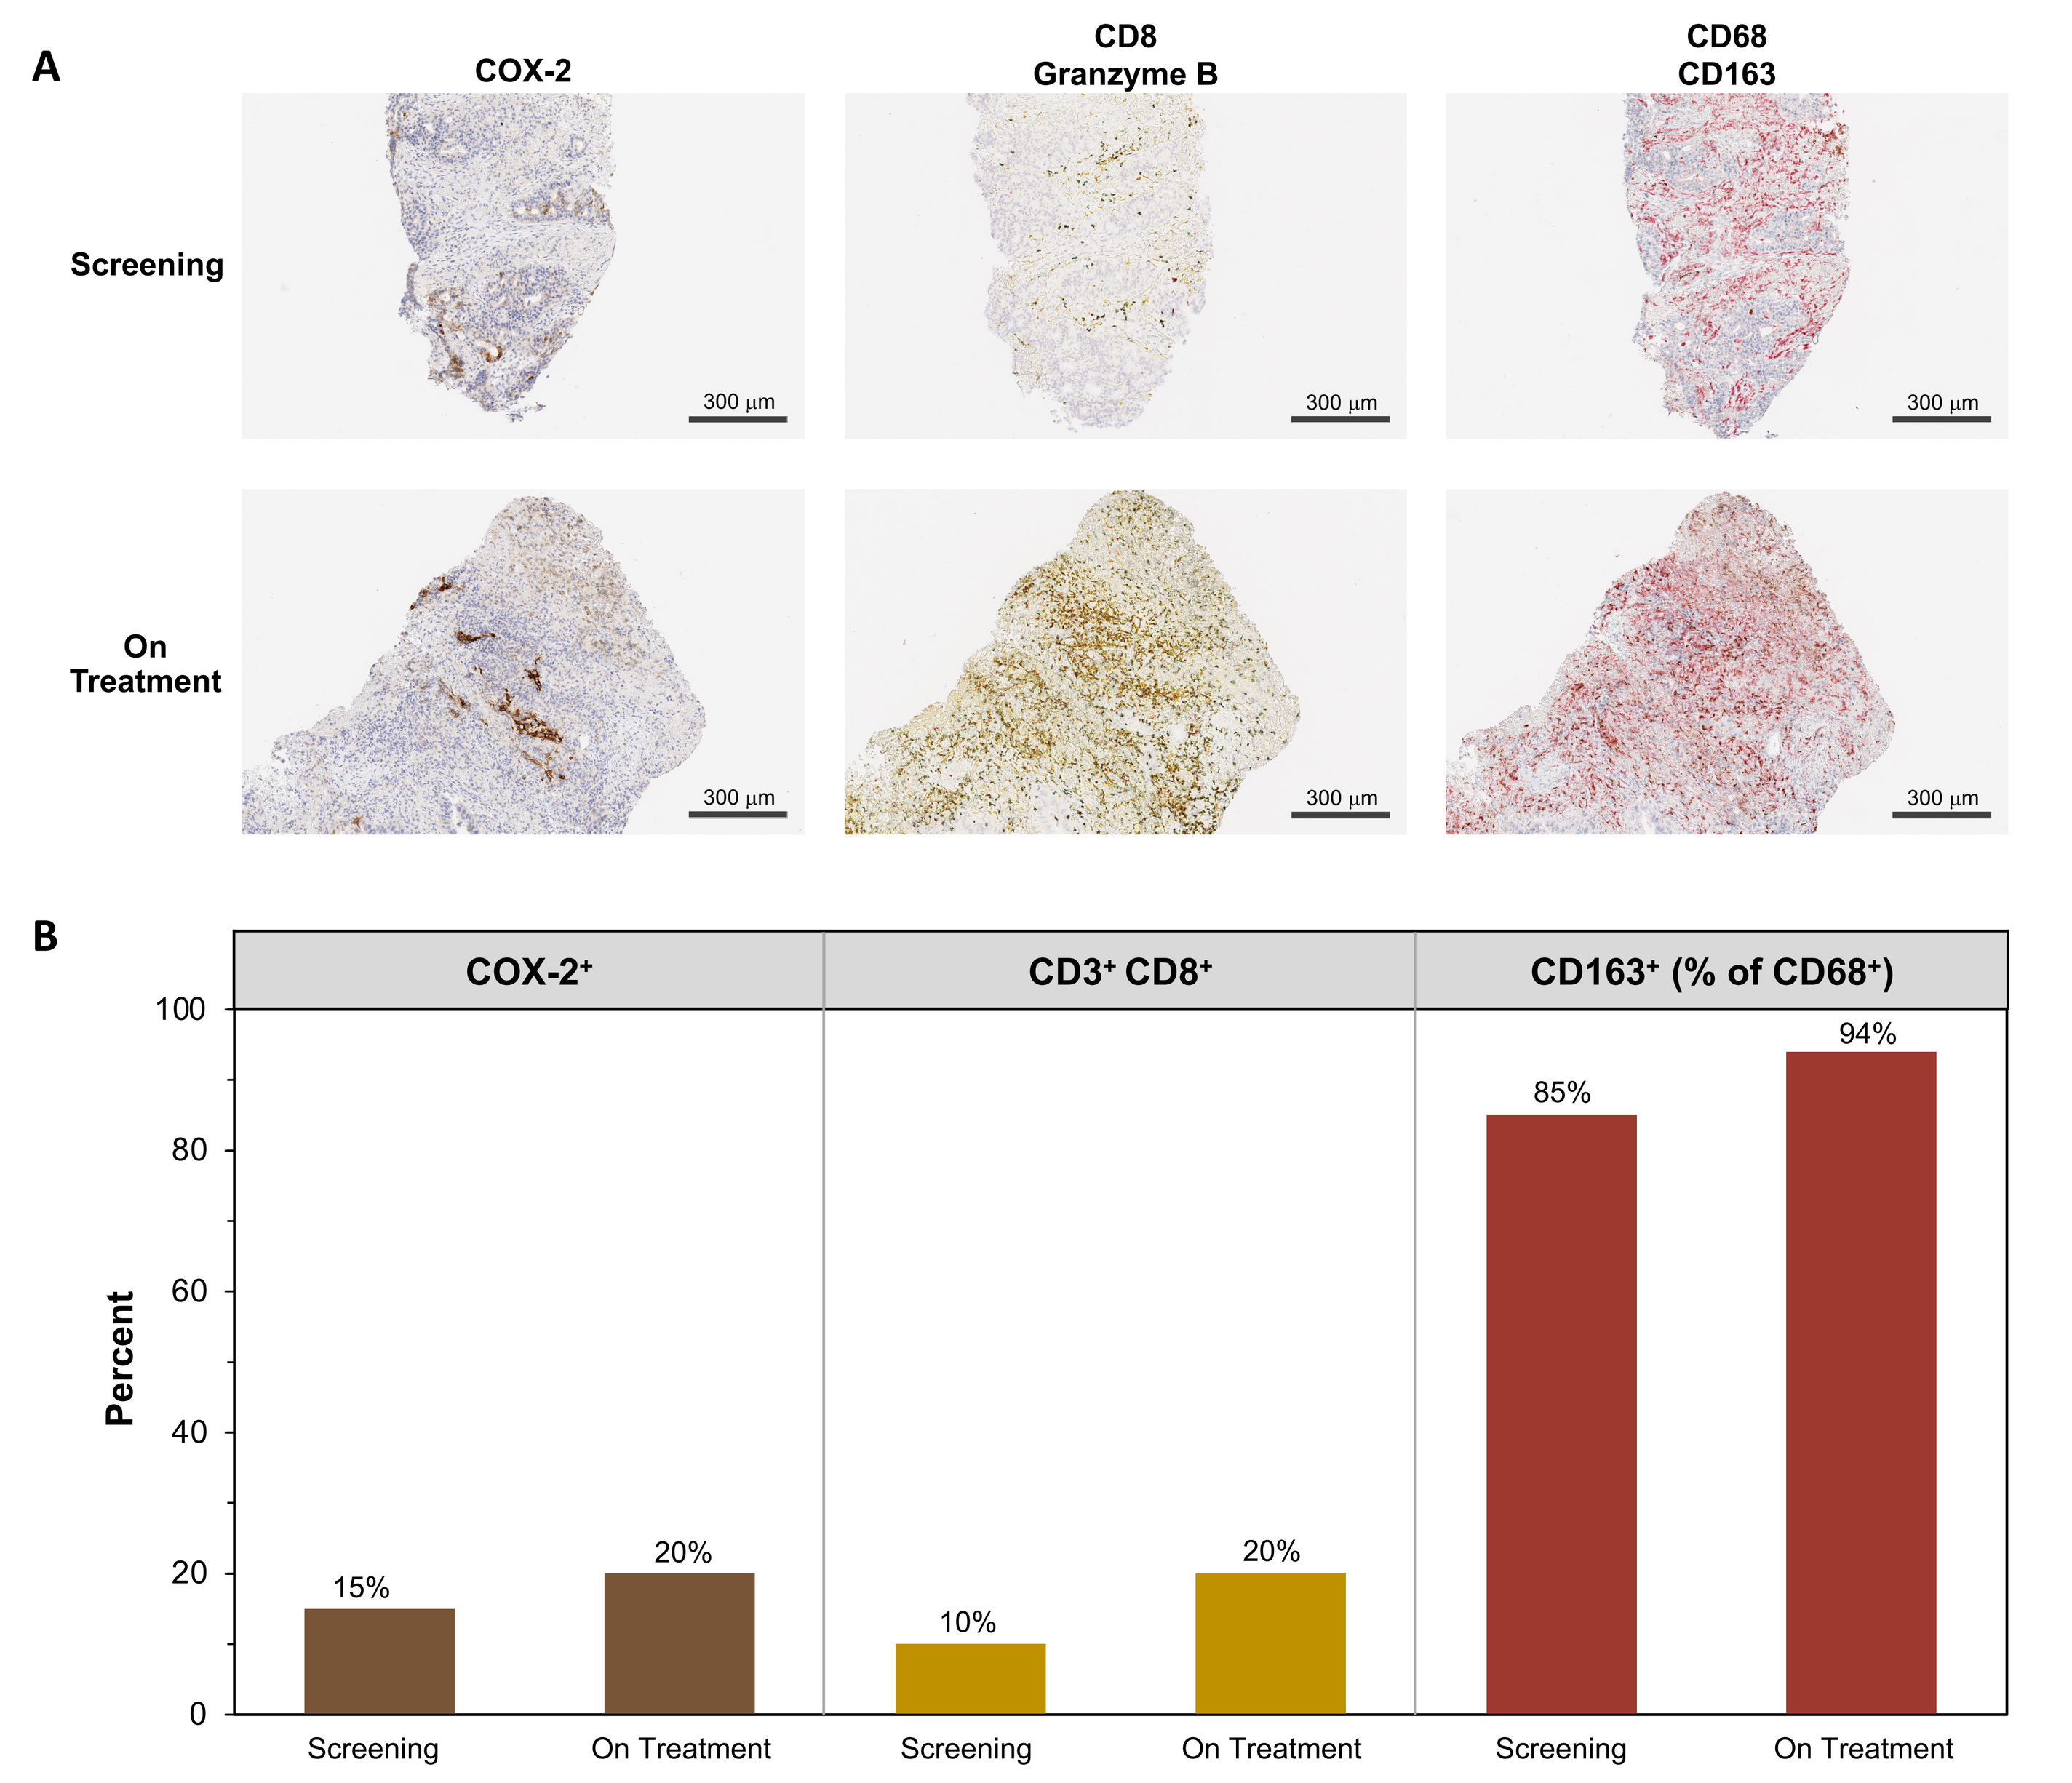
A
COX-2
CD8
Granzyme B
CD68
CD163
Screening
On Treatment
300 μm
B
COX-2⁺
CD3⁺ CD8⁺
CD163⁺ (% of CD68⁺)
Percent
100
80
60
40
20
0
15%
20%
10%
20%
85%
94%
Screening
On Treatment

Figure 7. IHC staining of COX-2⁺, CD8⁺/granzyme B⁺, and CD68⁺/CD163⁺ cells in paired biopsies from an endometrial cancer patient at screening and on treatment. (A) Images from paired biopsies stained for COX2 (DAB: brown)/hematoxylin (blue); CD3 (HIGHDEF® yellow chromogen)/CD8 (HIGHDEF® blue chromogen)/granzyme B (Vulcan Fast red chromogen); CD163 (red)/CD68 (DAB: brown)/ hematoxylin (blue). (B) Quantitative image analysis

CONCLUSIONS & FUTURE DIRECTIONS

- TPST-1495 is a novel inhibitor of PGE2 signaling that specifically antagonizes the tumor-promoting and immune-suppressing EP2 and EP4 receptors
- In this first-in-human Phase 1 study conducted in patients with treatment-refractory solid tumors, TPST-1495 demonstrated linear PK and both immune-specific and PGE2-specific pharmacodynamic activity consistent with blockade of PGE2 signaling through EP2 and EP4 receptors
- COX-2 expression was >10% in baseline biopsies of 2 patients who experienced best responses of SD, suggesting that baseline COX-2 levels may be a predictive biomarker of response in patients
- Currently, the combination of TPST-1495 and pembrolizumab is being assessed in endometrial cancer patients, a malignancy reported to express comparatively high levels of EP2, EP4, and COX-2
- Opportunities are being explored in familial adenomatous polyposis, a high-risk pre-cancer condition
- Future analyses will include paired biopsies collected from a subset of patients providing consent for biomarker evaluation

REFERENCES: 1. Pelly et al. *Cancer Discov*. 2021; 11(10):2602-2619; 2. Tury et al. *Oncotarget* 2016;7(51):85124-85141; 3. Ulahannan et al. 2023 ASCO Annual Meeting. June 2-6, 2023. Chicago, IL. Abstract #3107

ACKNOWLEDGMENTS: We thank the patients and their families, investigators, and coordinators for participation in the study. Ingrid Koo, PhD, provided editorial support for the poster.

SITC 38th Annual Meeting, November 1-5, 2023, San Diego, CA [This presentation is the intellectual property of the author/presenter. Contact nstandifer@tempesttx.com for permission to reprint and/or distribute]