Dual antagonism of prostaglandin receptors EP2 and EP4 by TPST-1495 suppresses tumor growth and stimulates antitumor immunity

Chan C. Whiting[1], Kim Fischer[2], Bryan Laffitte[2], Lisa Rahbaek[2], Davorka Messmer[2], Austin Chen[2], Traci Olafson[2], Natalie Nguyen[2], Alejandro Dearie[2], Yalda Bravo[5], Joe Nagamizo[5], David Powell[5], Joyce Wu[5], Bob Gomez[5], Tao Sheng[5], Jim Ding[5], Amanda Enstrom[1], Derek Metzger[1], Brian Francica[1], Dingzhi Wang[3], Raymond Dubois[3], Dipak Panigrahy[4], Ginna Laport[1], Peppi Prasit[2], Thomas Dubensky[1]

[1]Tempest Therapeutics, Inc., San Francisco, CA; [2]Inception Therapeutics, San Diego, CA; [3]College of Medicine, Medical University of South Carolina, Charleston, SC; [4]Beth Israel Deaconess Medical Center, Harvard Medical School, Boston, MA; [5]Inception Sciences, Vancouver, BC, Canada

ABSTRACT

Background

Progression of diverse malignancies is promoted by elevated levels of Prostaglandin E2 (PGE2). High PGE2 levels result from dysregulation of Cyclooxygenase-2 (COX-2), the enzyme that produces this lipid. PGE2 stimulates tumor cell proliferation, survival, evasion and metastasis along with host angiogenesis. PGE2 suppresses anti-tumor immunity through inhibiting the function of critical anti-tumor immune effectors such as NK and T cells, and macrophages, while promoting the activity of suppressive immune cells including myeloid derived suppressor cells, M2 macrophages, and regulatory T cells. PGE2 signals through a family of four homologous E-prostanoid (EP) G-coupled receptors, known as EP1, EP2, EP3 and EP4; which, are activated via distinct signal transduction pathways. Published literature and experimental results presented here demonstrate that selective antagonism of both EP2 and EP4 receptor signaling, but not EP1 and EP3, effectively overcomes PGE2-mediated immune suppression and results in anti-tumor efficacy. TPST-1495 is a first-in-class orally available small molecule selective dual antagonist of the human PGE2 receptors EP2 and EP4, currently under development by Tempest.

Methods

The effects of TPST-1495 as monotherapy or in combination with anti-PD1 were evaluated in the syngeneic mouse colon models CT26 and ApcMin/+ as well as model of Lewis Lung Carcinoma. The mechanism of anti-tumor immunity of TPST-1495 was evaluated using in vitro primary dendritic cell (DC) differentiation and activation assays. Characterization of in vitro differentiated immune cells or tumor infiltrating lymphocytes were performed using flow cytometry. ELISA was used for measurement of cytokine production.

Results

Treatment with TPST-1495 reversed PGE2 immune suppression in vitro and in vivo compared to antagonism of EP4 alone or all 4 EP receptors. TPST-1495 prevented PGE2 inhibition in vitro of DC differentiation and activation from human donor monocytes; single EP2 or EP4 antagonists were sub-optimal in this assay. Significantly, combination with EP1 and/or EP3 antagonists reversed the effect of dual EP2 and EP4 blockade on PGE2 immune suppression, suggesting that COX-2 inhibition is not optimal for blocking the effects of PGE2. TPST-1495 induced potent anti-tumor immune responses and significant tumor regression as a monotherapy in two different murine tumor treatment models of colon cancer, CT26 and Apcmin/+. CT26 tumors analyzed from mice treated with TPST-1495 revealed a significant increase of infiltrating effector T cells. TPST-1495 combination with anti-PD1 synergistically inhibited CT26 tumor progression.

Conclusions

TPST-1495 is a differentiated highly potent selective dual antagonist of EP2 and EP4 that overcomes prostaglandin-mediated immune suppression and promotes anti-tumor efficacy.

INTRODUCTION

- TPST-1495 is a first-in-class, orally-administered, small molecule, selective dual antagonist of the human prostaglandin E2 (PGE2) receptors, EP2 and EP4
- PGE2 signals through four homologous E-prostanoid, G-protein coupled receptors (GPCRs): EP1, EP2, EP3 and EP4, which activate distinct signal transduction pathways[1-2]
- PGE2 suppresses anti-tumor immunity by inhibiting anti-tumor immune effector cells (NKs, T cells, DCs and M1 macrophages) while enhancing suppressive immune cells (MDSCs, M2 macrophages, and Tregs)[3-6]
- Elevated cyclooxygenase-2 (COX-2) and the components of the PGE2 pathway including its receptors, EP2 and EP4, are correlated with immunosuppression in the tumor microenvironment (TME) and cancer progression in multiple malignancies[7-12]
- Increased levels of PGE2 produced by COX-2 stimulate tumor cell proliferation, survival, evasion and metastasis as well as host angiogenesis[13-15]
- Selective dual antagonist of both EP2 and EP4 is supported by results demonstrating:
 - distinct functional roles for each EP receptor subtype
 - enhanced anti-tumor immunity
 - potentially reduced toxicities compared to NSAIDs

FIGURE 1: Signaling Pathways Associated with PGE2 receptors: Rationale for Inhibiting both EP2 and EP4 Receptors with a Dual Antagonist

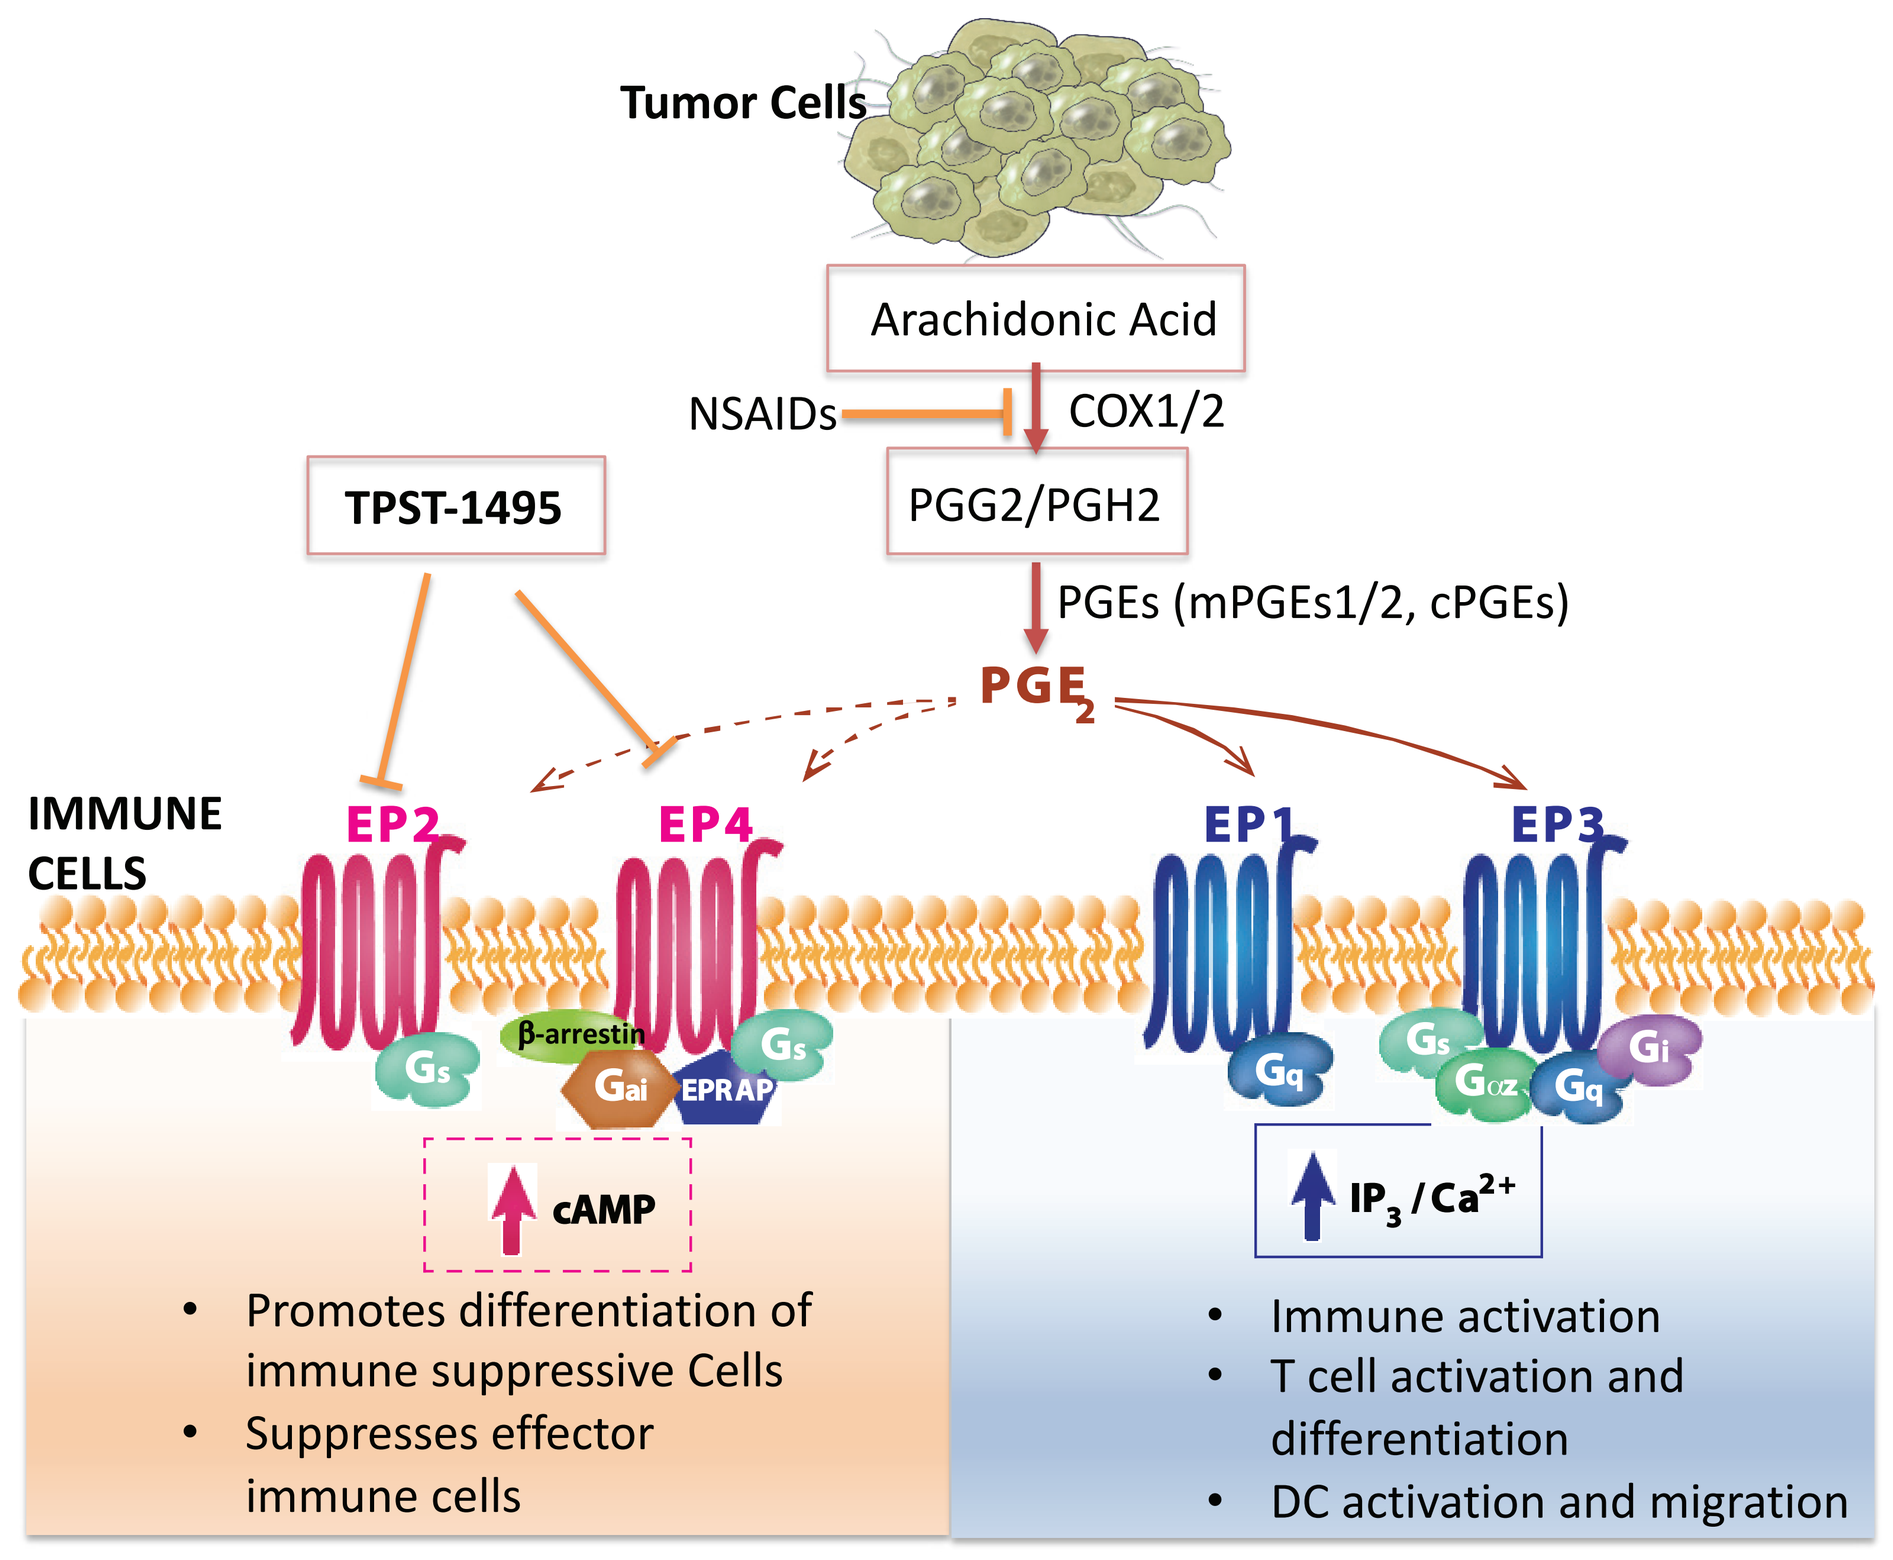
Tumor Cells
Arachidonic Acid
NSAIDs
COX1/2
TPST-1495
PGG2/PGH2
PGEs (mPGEs1/2, cPGEs)
PGE2
IMMUNE CELLS
EP2
EP4
EP1
EP3
β-arrestin
cAMP
IP3 / Ca2+
Promotes differentiation of immune suppressive Cells
Suppresses effector immune cells
Immune activation
T cell activation and differentiation
DC activation and migration

Figure 1: Antagonism of both EP2 and EP4 required for reversal of PGE2-mediated immune suppression.

NSAIDs: nonsteroidal anti-inflammatory drugs; COX: cyclooxygenase; PGG2: prostaglandin G2; PGH2: prostaglandin H2; PGEs: prostaglandin synthase (m: microsomal; c: cytoplasmic); PGE2: prostaglandin E2; EP1-4: E-prostanoid receptors 1 through 4; cAMP: cyclic adenosine monophosphate; IP3: inositol triphosphate

RESULTS

FIGURE 2: TPST-1495 is Significantly More Potent Than EP2- or EP4-Selective Single Antagonists in Reversing PGE2-mediated Immune Suppression

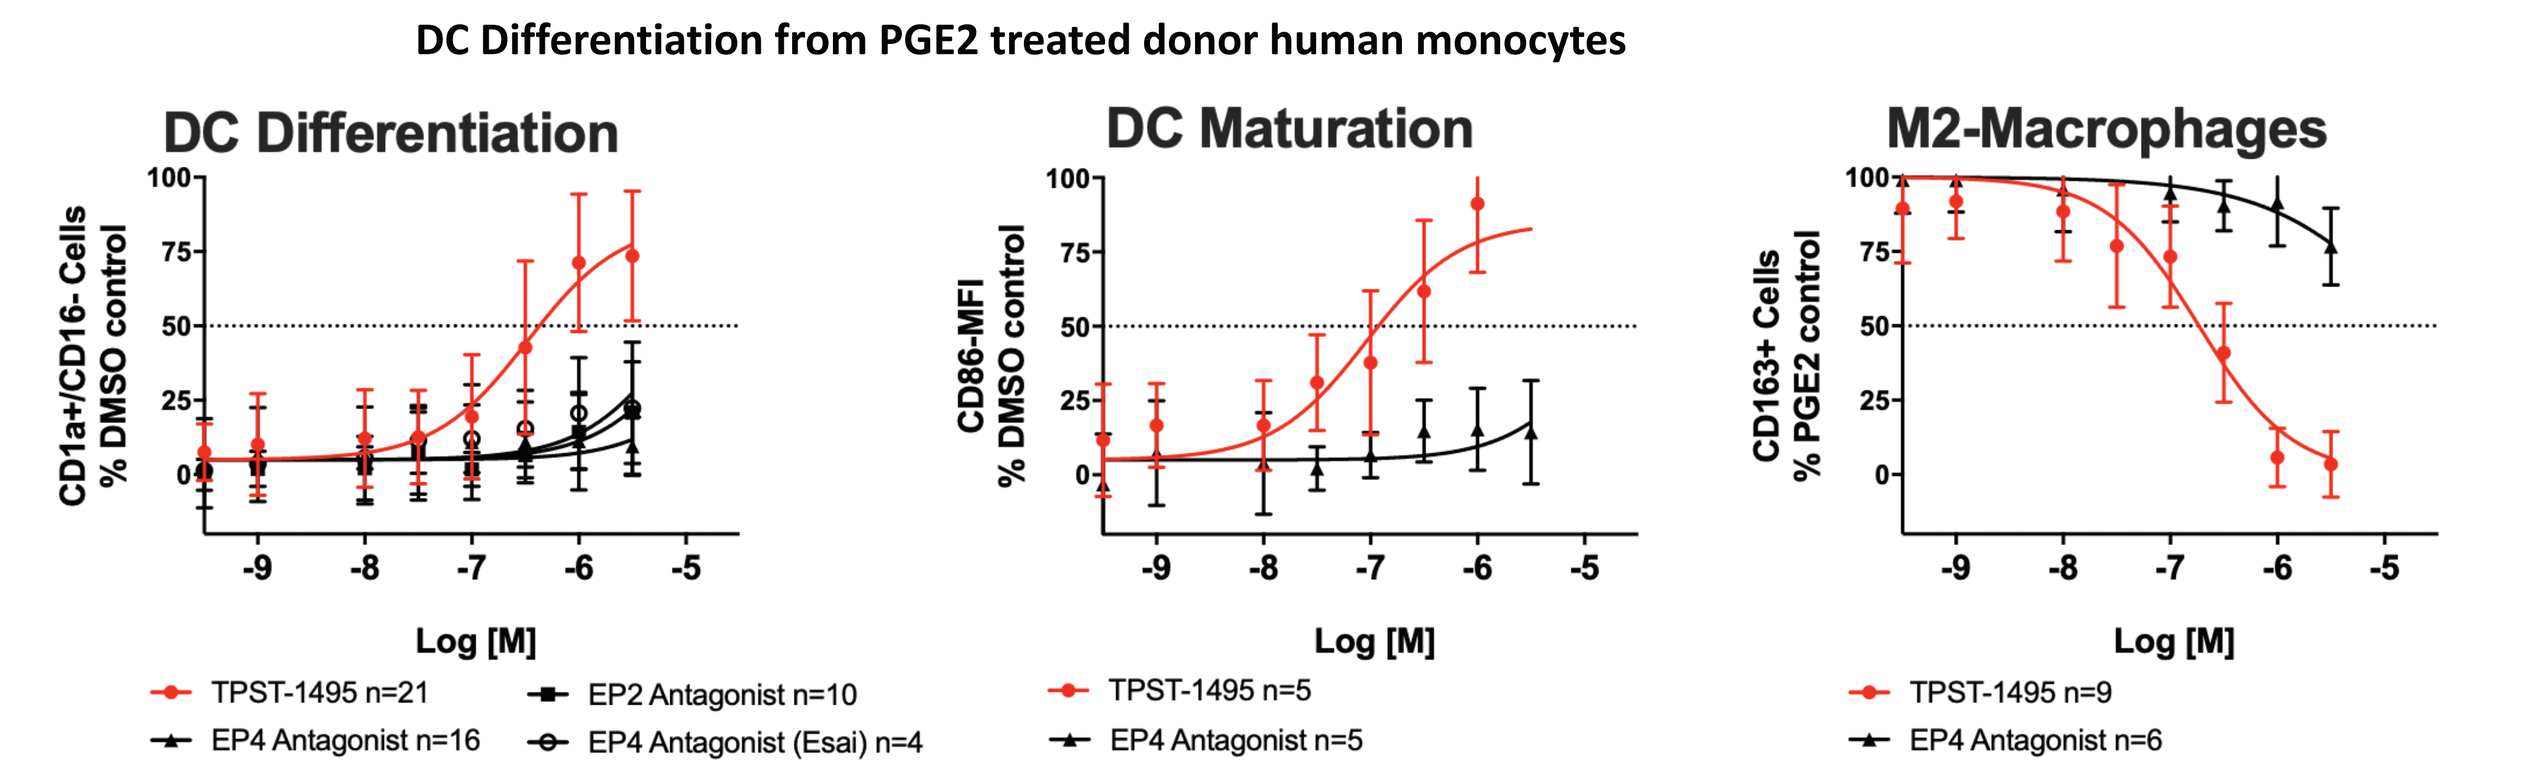
DC Differentiation from PGE2 treated donor human monocytes
DC Differentiation
DC Maturation
M2-Macrophages
CD1a+/CD16- Cells % DMSO control
CD86-MFI % DMSO control
CD163+ Cells % PGE2 control
Log [M]
TPST-1495 n=21
EP4 Antagonist n=16
EP2 Antagonist n=10
EP4 Antagonist (Esai) n=4
TPST-1495 n=5
EP4 Antagonist n=5
TPST-1495 n=9
EP4 Antagonist n=6

Figure 2: TPST-1495 reversed PGE2-mediated inhibition of human donor monocyte to dendritic cell (DC) differentiation (left panel) and activation (middle panel). TPST-1495 inhibited PGE2-induced monocyte to M2 macrophage differentiation (right panel). Single EP2 or EP4 antagonists were sub-optimal in this in vitro DC/M2 macrophage differentiation assay. Results shown are averaged from n=4-21 human healthy donors as indicated next to each drug name. Source: Tempest data (on file).

FIGURE 3: TPST-1495 has Significant Monotherapy Activity in Lewis Lung Carcinoma Model

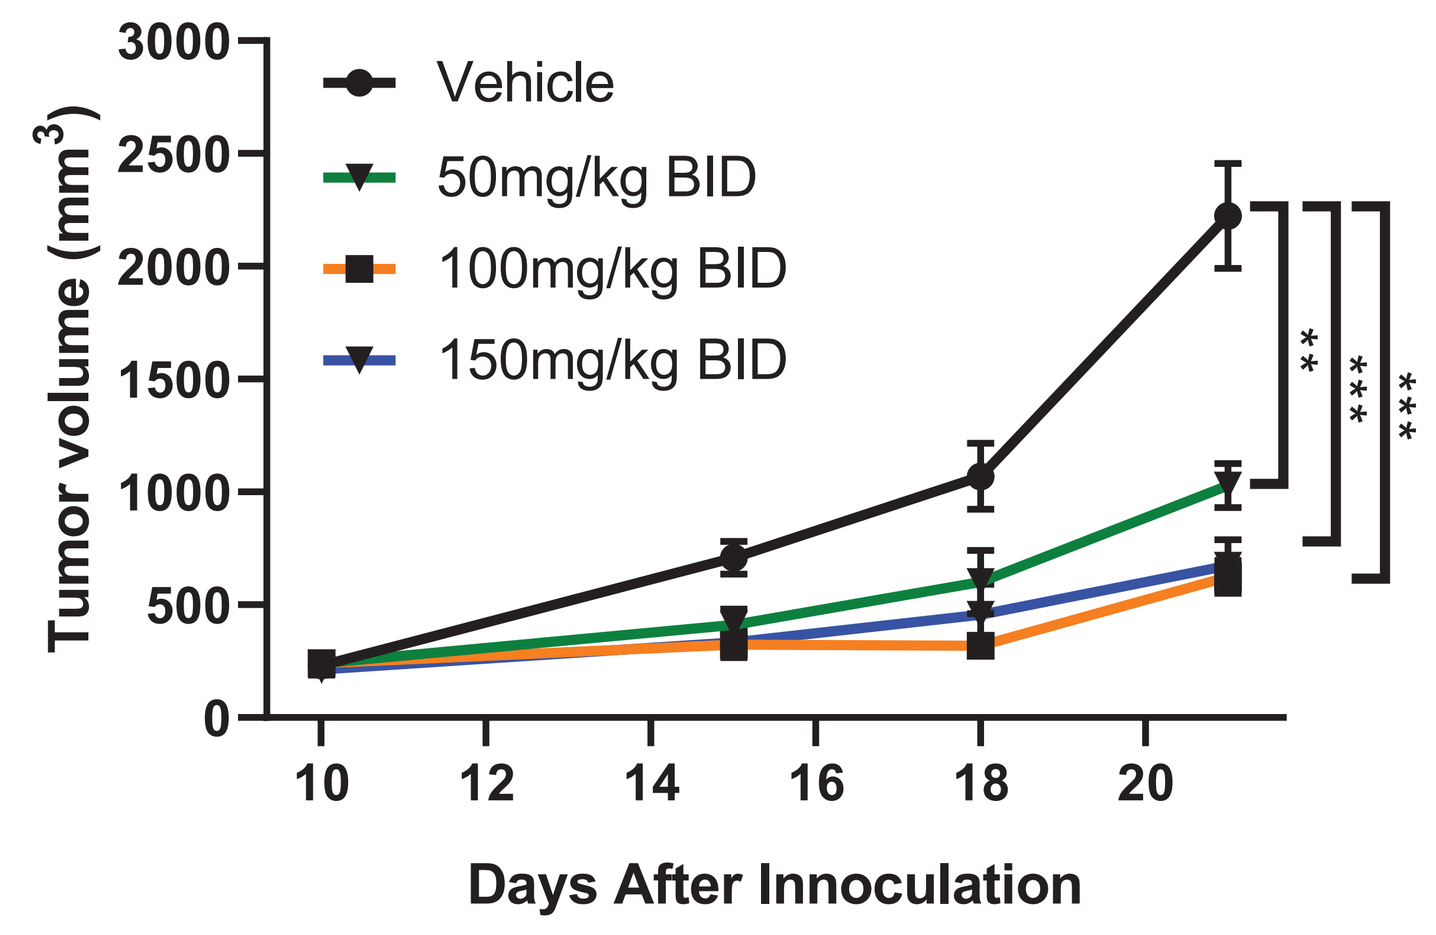
Vehicle
50mg/kg BID
100mg/kg BID
150mg/kg BID
Tumor volume (mm3)
Days After Innoculation

Figure 3: TPST-1495 treatment 50mg/kg, BID (**p=0.0014), 100mg/kg, BID (*** p=0.0002), 150mg/kg, BID (***p=0.0003) significantly inhibited tumor growth in Lewis Lung Carcinoma model, compared to vehicle control. Source: Dipak Panigrahy collaboration (data on file).

FIGURE 4: TPST-1495 has Potent Single Agent Anti-tumor Activity Associated with Increased T Cell Infiltration in the TME

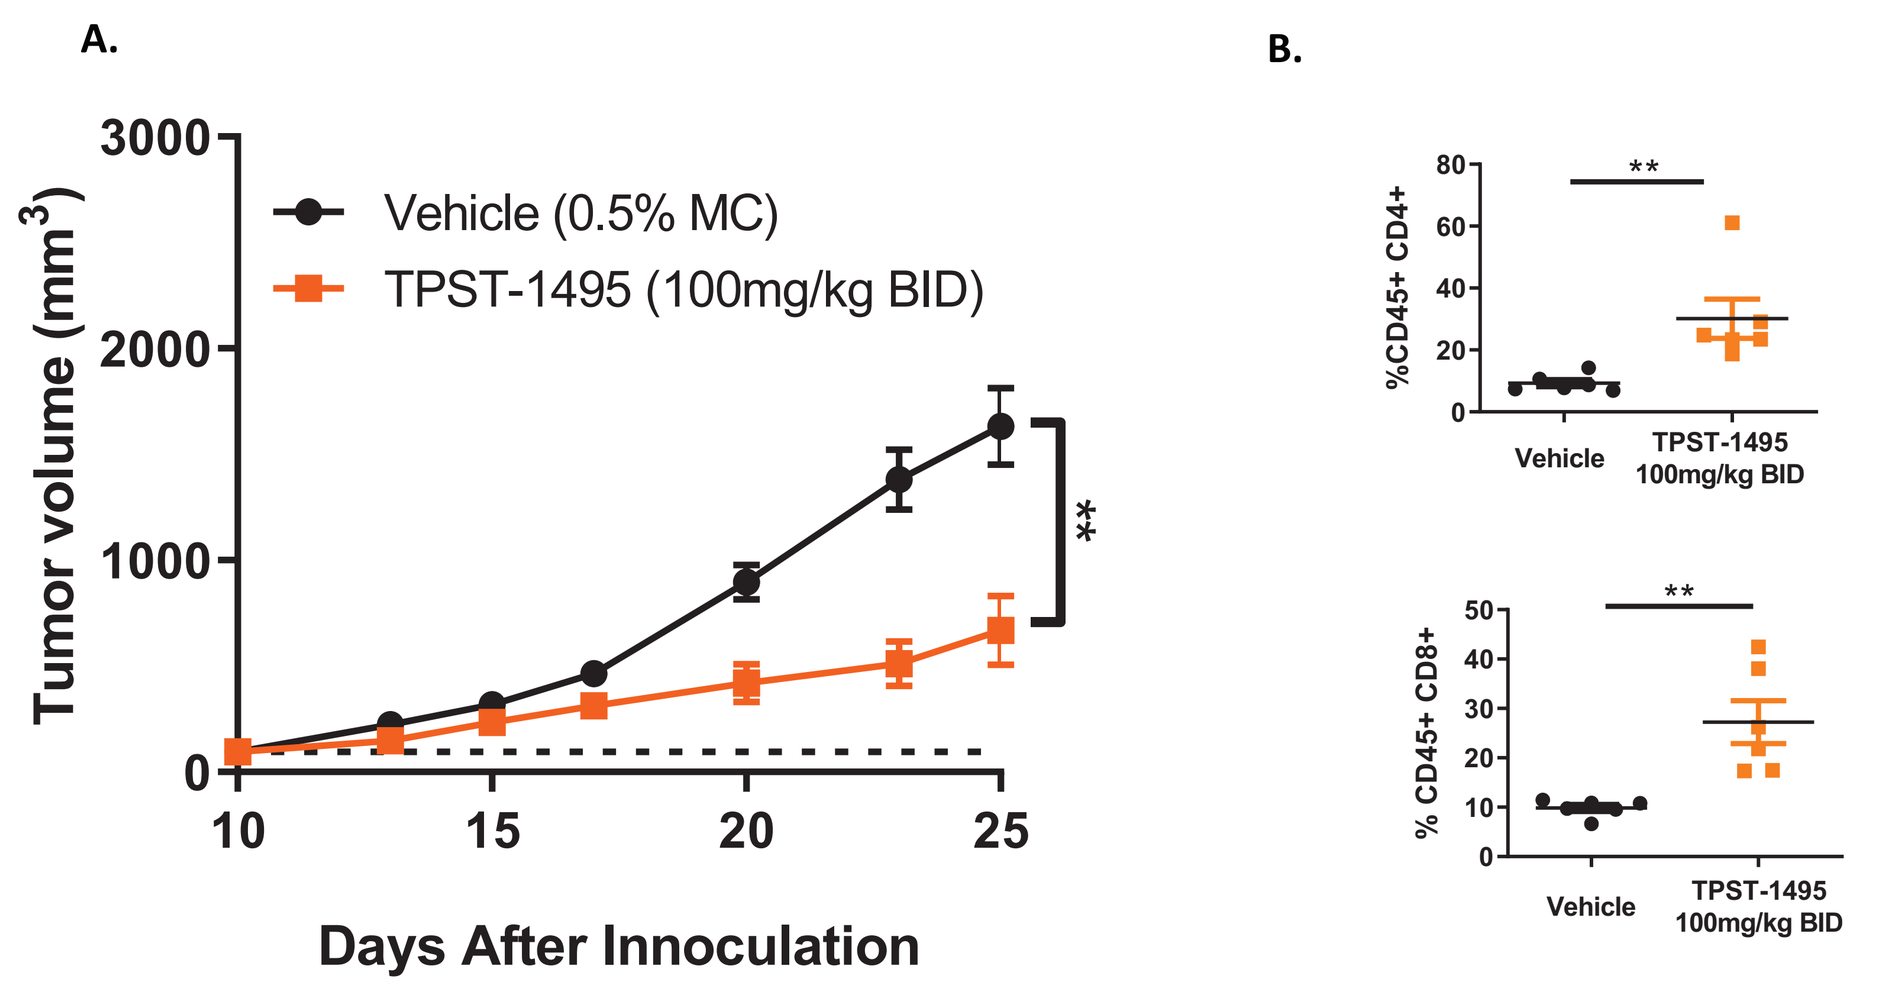
A.
B.
Vehicle (0.5% MC)
TPST-1495 (100mg/kg BID)
Tumor volume (mm3)
Days After Innoculation
%CD45+ CD4+
% CD45+ CD8+
Vehicle
TPST-1495 100mg/kg BID

Figure 4: A. TPST-1495 (100mg/kg, BID) significantly inhibited CT26 colon tumor growth compared to vehicle alone (** p=0.0012). B. CT26 tumors analyzed from mice treated with TPST-1495 (100mg/kg BID) revealed a significant increase of infiltrating T cells CD4+ (** p=0.009) and CD8+ (** p=0.0028) compared to vehicle control. Source: Tempest data (on file).

FIGURE 5: TPST-1495 EP2/EP4 Dual Antagonist is Significantly More Potent than EP4-Selective Single Antagonist in Clinical Development

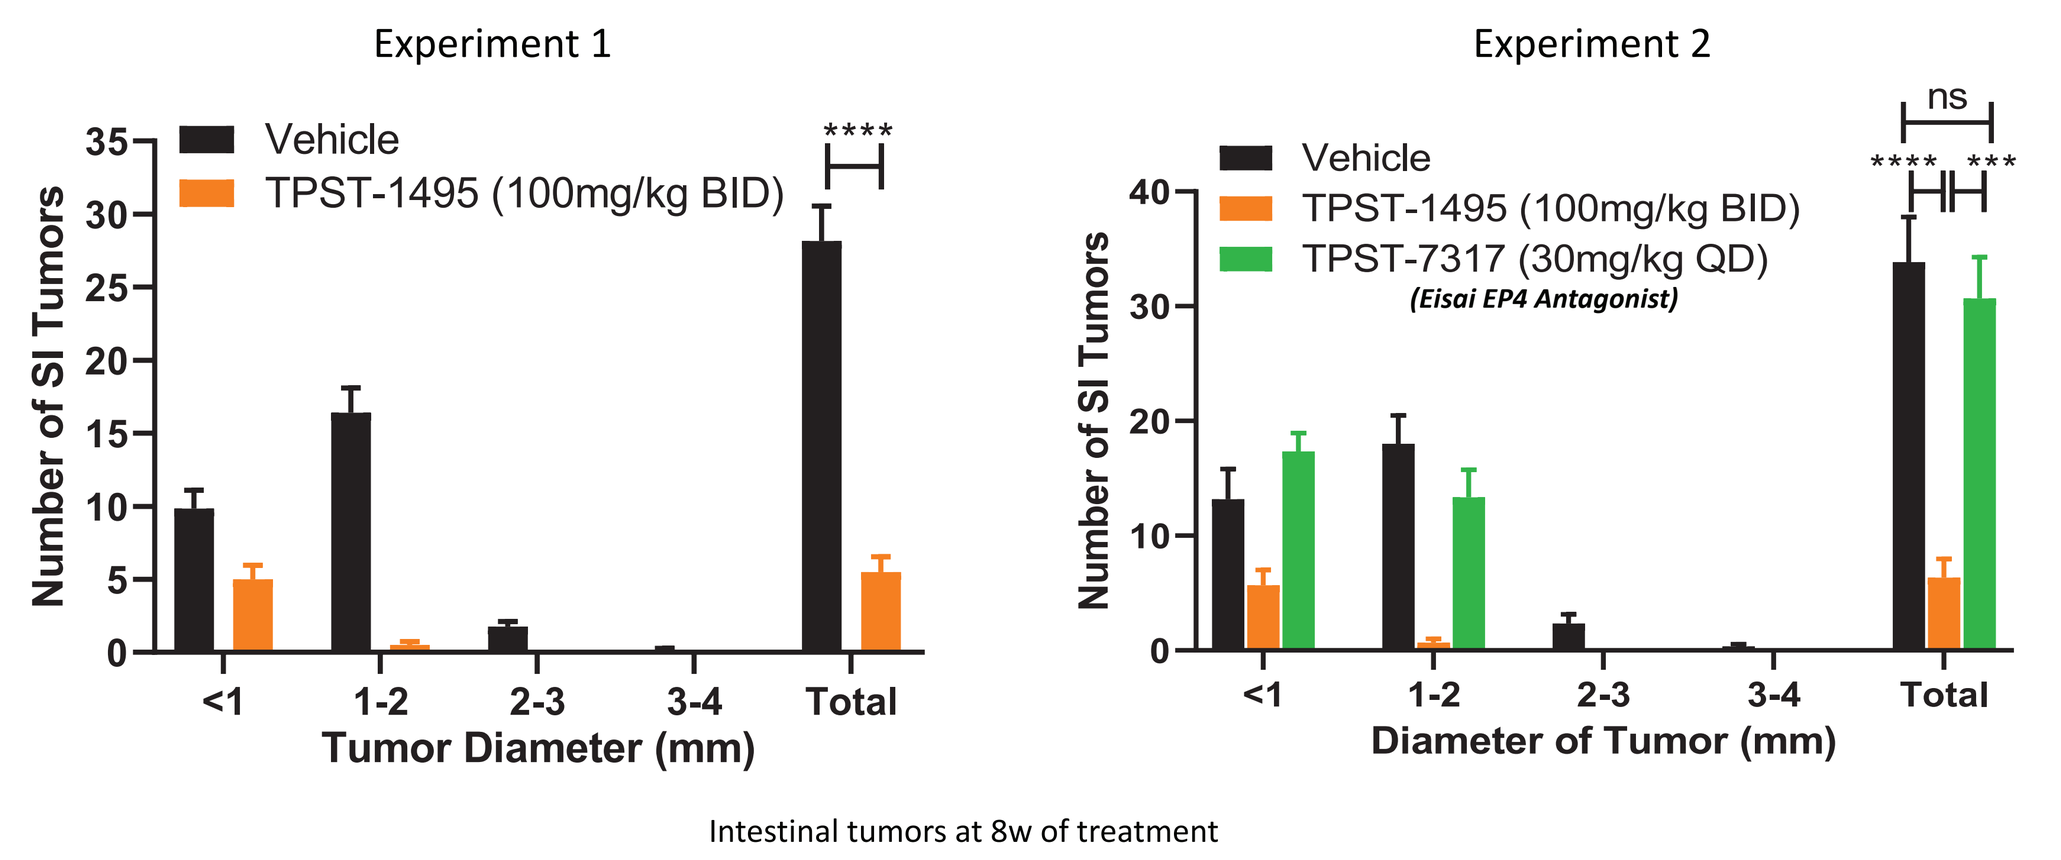
Experiment 1
Experiment 2
Vehicle
TPST-1495 (100mg/kg BID)
TPST-7317 (30mg/kg QD)
(Eisai EP4 Antagonist)
Number of SI Tumors
Tumor Diameter (mm)
Diameter of Tumor (mm)
Intestinal tumors at 8w of treatment

Fiigure 5: TPST-1495-induced significant tumor regression as a monotherapy in spontaneous colon tumor model: Adenomatous polyposis coli gene ApcMin/+ mice. Experiment 1: TPST-1495 (100mg/kg, BID) vs Control (**** p<0.0001); Experiment 2: TPST-1495 vs Control (**** p<0.0001), vs TPST-7317 (100mg/kg, BID, Eisai EP4-specific antagonist) (*** p=0.0001). No effect on animal weights observed. Source: Raymond Dubois collaboration (data on file).

FIGURE 6: TPST-1495 Combination with Chemotherapy Enhances Anti-tumor Response in Lewis Lung Carcinoma

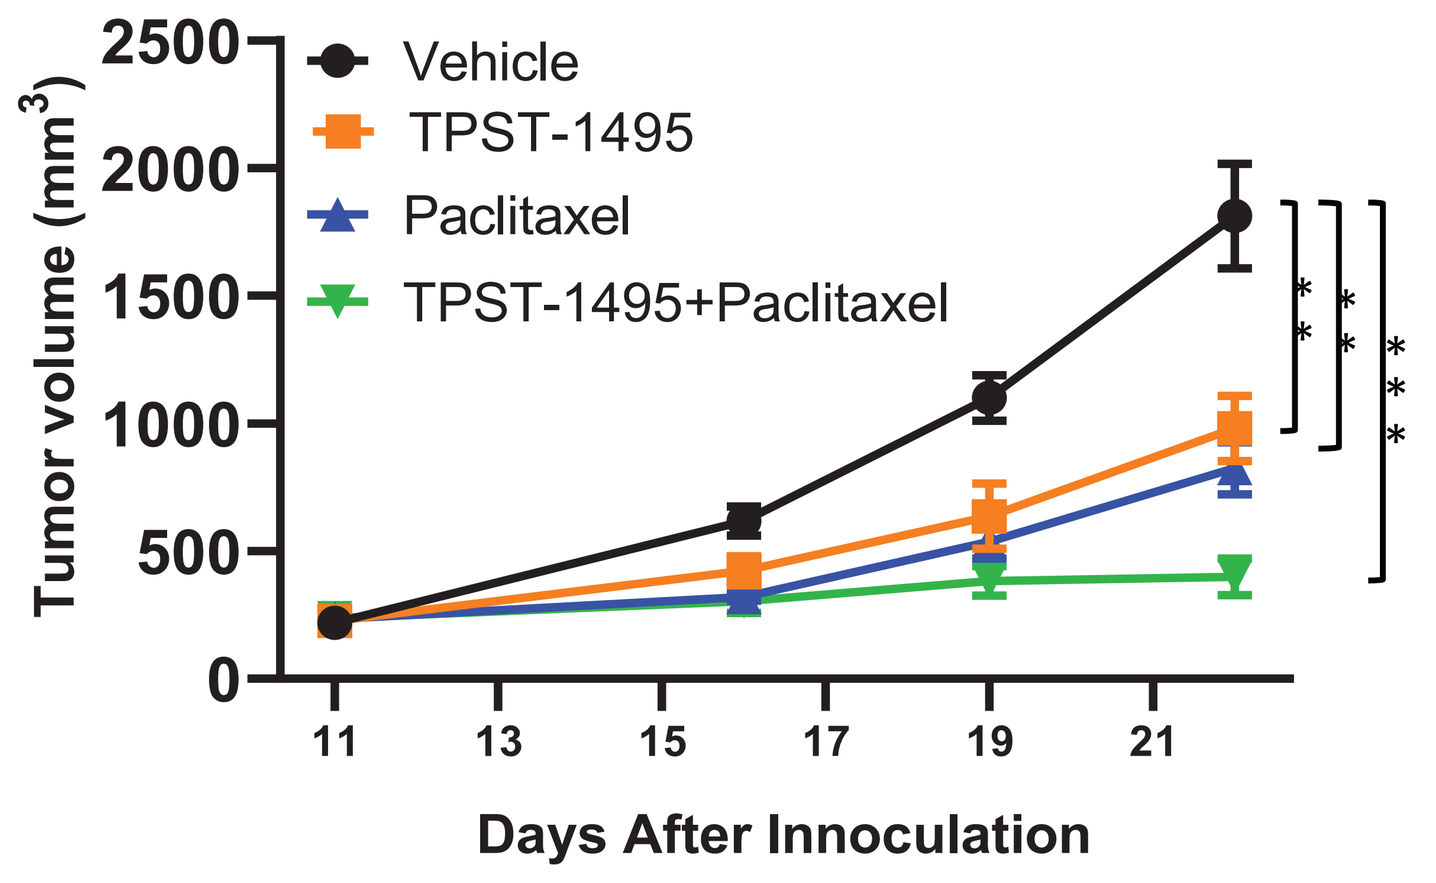
Vehicle
TPST-1495
Paclitaxel
TPST-1495+Paclitaxel
Tumor volume (mm3)
Days After Innoculation

Figure 6: TPST-1495 (100mg/kg BID), chemotherapy (paclitaxel, 10mg/kg, Q3D) alone or in combination significantly inhibited tumor growth in Lewis Lung Carcinoma compared to vehicle; TPST-1495 vs.Vehicle (**p=0.0088), Paclitaxel vs. Vehicle ((**p=0.0026). TPST-1495 + Paclitaxel vs. Vehicle (*** p=0.0002). Paclitaxel vs. Combination (p=.0091), TPST-1495 vs. Combination (p=.0041). Statistics on tumor volumes were taken at day 22 post treatment. Source: Dipak Panigrahy collaboration (data on file).

FIGURE 7: TPST- 1495 Combination with Anti-PD-1 Enhances Anti-tumor Response in the Mouse CT26 Colon Cancer Model

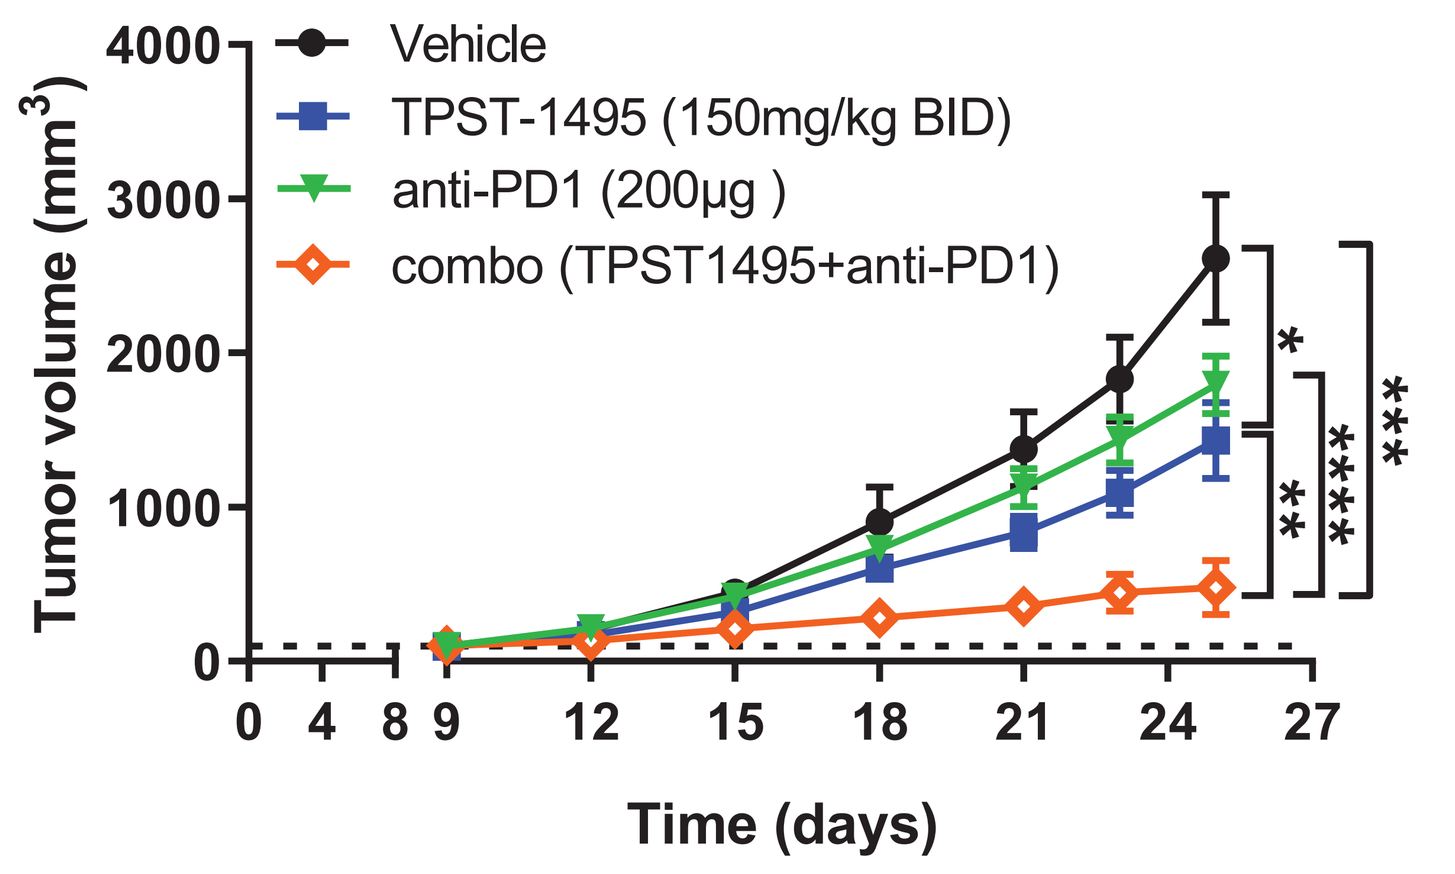
Vehicle
TPST-1495 (150mg/kg BID)
anti-PD1 (200µg)
combo (TPST1495+anti-PD1)
Tumor volume (mm3)
Time (days)

Figure 7: TPST-1495 (150mg/kg BID), anti-programmed cell death protein-1 (anti-PD-1) antibody (200ug, 3QD) inhibits CT26 tumor growth compared to vehicle or monotherapy alone (* p=0.029, ** p=0.0065, *** p=0.0003, **** p<0.00001). Source: Tempest data (on file).

FIGURE 8: Multiple Tumor Types Express Elevated Levels of PGE2 Pathway Genes

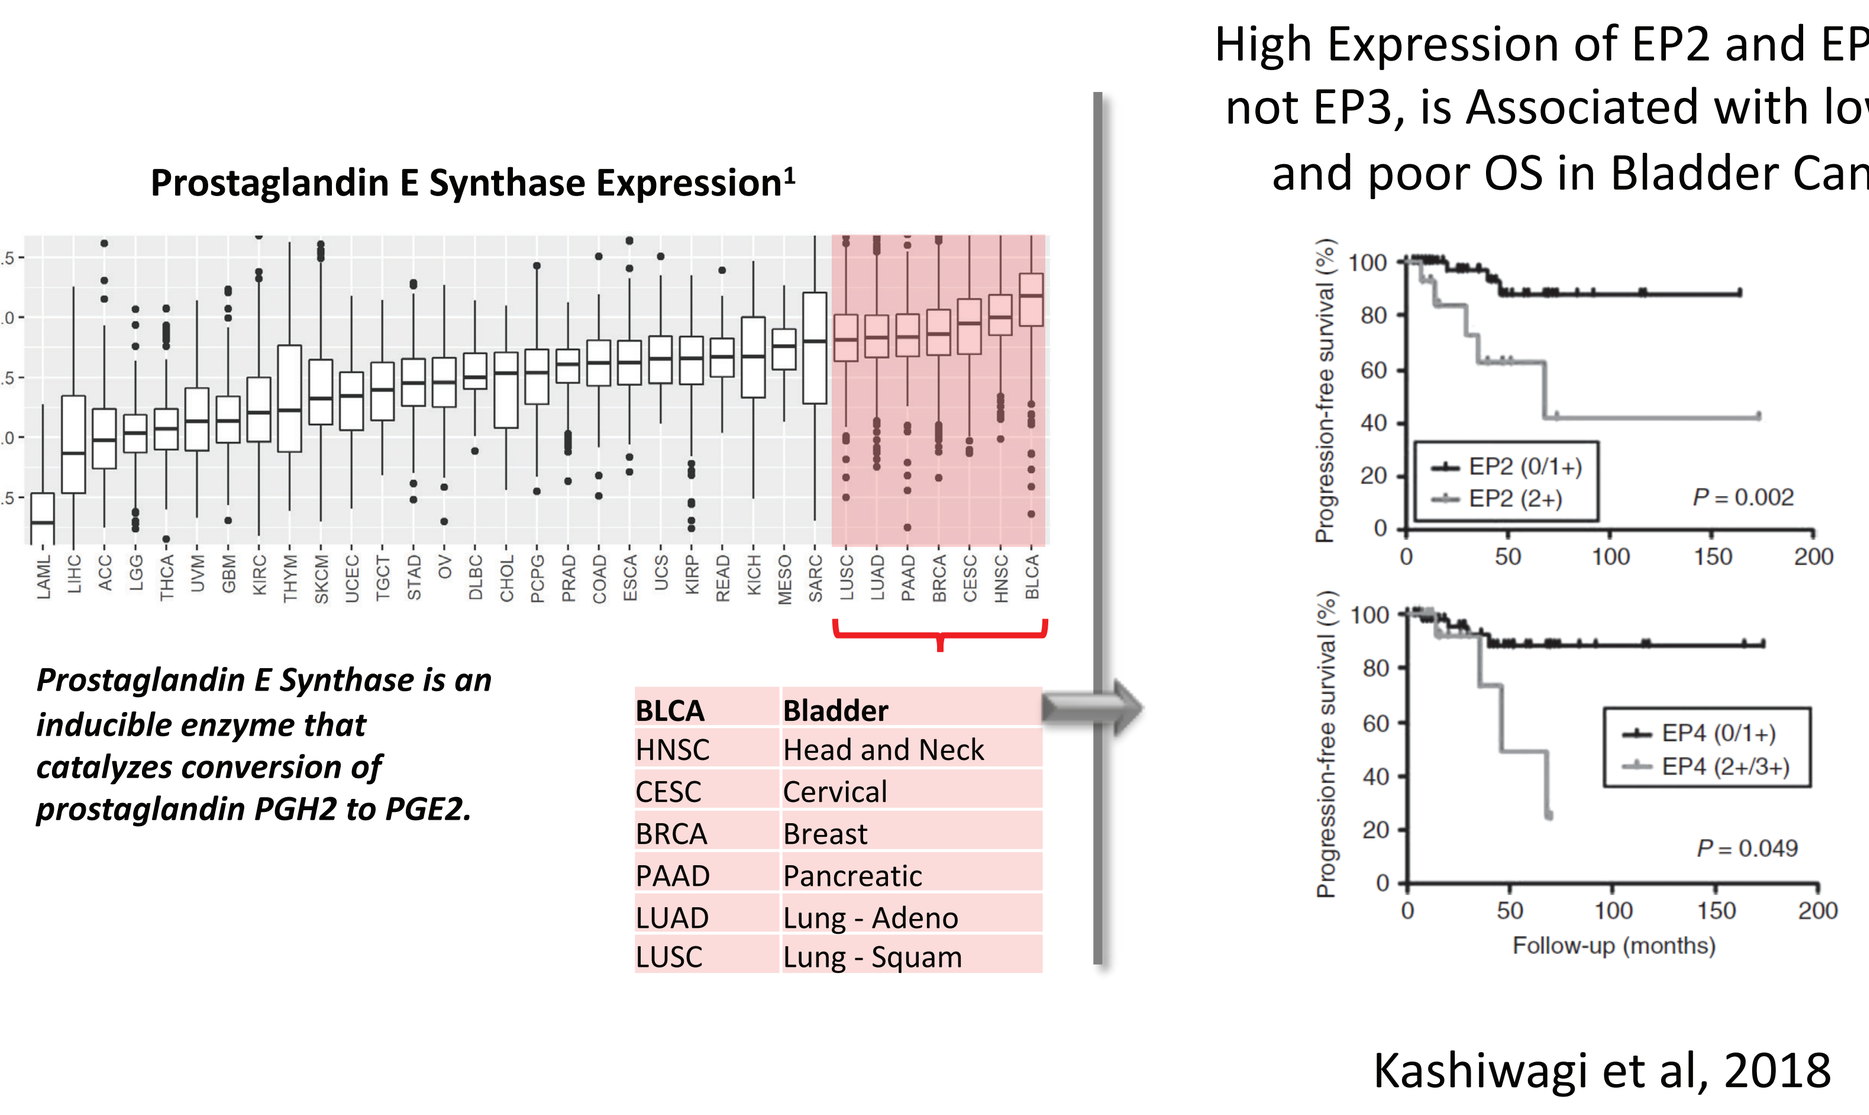
Prostaglandin E Synthase Expression1
PTGES
Prostaglandin E Synthase is an inducible enzyme that catalyzes conversion of prostaglandin PGH2 to PGE2.
BLCA Bladder
HNSC Head and Neck
CESC Cervical
BRCA Breast
PAAD Pancreatic
LUAD Lung - Adeno
LUSC Lung - Squam
High Expression of EP2 and EP4, but not EP3, is Associated with low PFS and poor OS in Bladder Cancer
Progression-free survival (%)
EP2 (0/1+)
EP2 (2+)
P = 0.002
EP4 (0/1+)
EP4 (2+/3+)
P = 0.049
Follow-up (months)
Kashiwagi et al, 2018

Figure 8: Analysis of 10,000 RNAsequencing datasets from The Cancer Genome Atlast (TCGA) revealed ranked expression of Prostaglandin E Synthase, inducible enzyme that catalyzes conversion of prostaglandin PGH2 to PGE2, in different tumor types (left panel) suggesting production of PGE2 by tumors as a mechanism of immune suppression. High expression of EP2 and EP4 receptors is associated with poor prognosis in bladder cancer (right panel, Kashiwagi et al, 2018).

FIGURE 9: Multiple Tumor Types Express Elevated Levels of EP2 and EP4 PGE2 Receptors

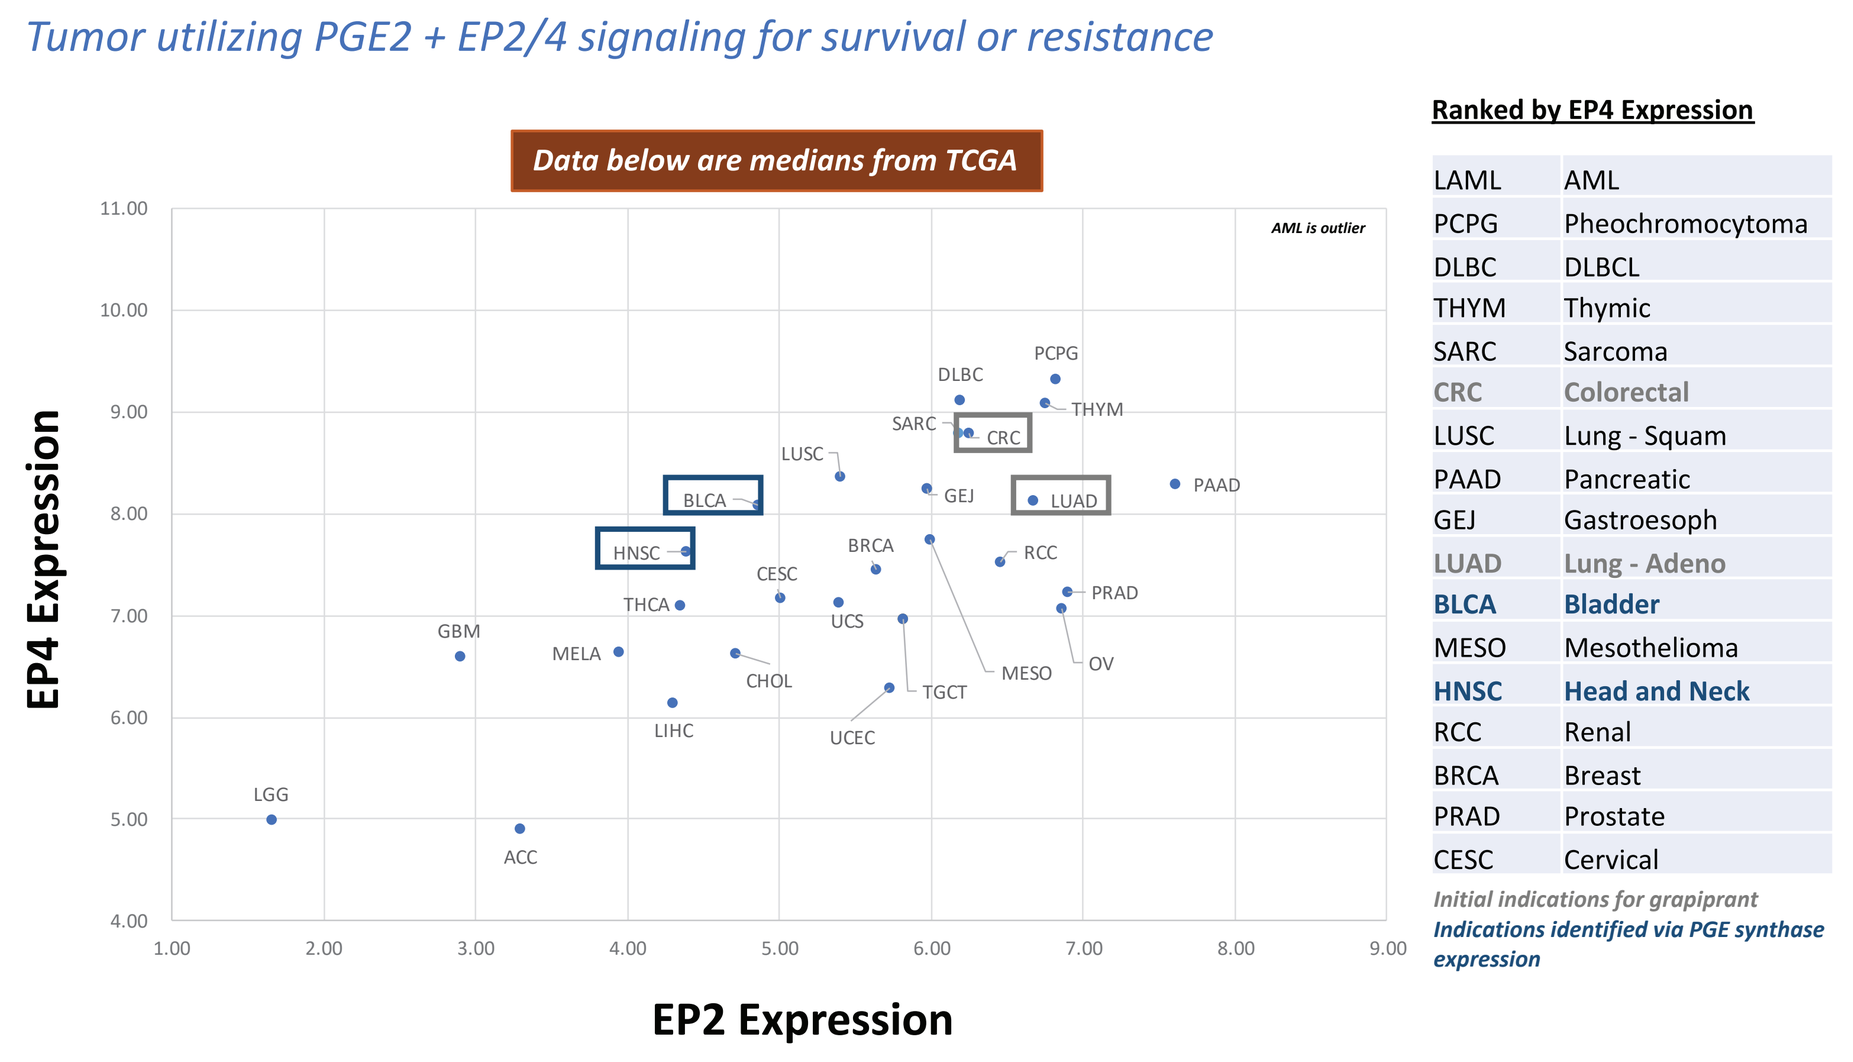
Tumor utilizing PGE2 + EP2/4 signaling for survival or resistance
Data below are medians from TCGA
EP4 Expression
EP2 Expression
Ranked by EP4 Expression
LAML AML
PCPG Pheochromocytoma
DLBC DLBCL
THYM Thymic
SARC Sarcoma
CRC Colorectal
LUSC Lung - Squam
PAAD Pancreatic
GEJ Gastroesoph
LUAD Lung - Adeno
BLCA Bladder
MESO Mesothelioma
HNSC Head and Neck
RCC Renal
BRCA Breast
PRAD Prostate
CESC Cervical
Initial indications for grapiprant
Indications identified via PGE synthase expression

Figure 9: Analysis of 10,000 RNAsequencing datasets in TCGA revealed high expression of EP2 and EP4 in multiple tumor types.

TPST-1495 CONCLUSIONS

- First-in-class dual antagonist of ***both*** EP2 and EP4 receptors
- Significantly more potent than EP4-selective single antagonists in clinical development
- Antagonism of both EP2 and EP4 receptors required for optimal reversal of PGE2-mediated immune suppression
- Potent single agent activity in multiple preclinical tumor models and enhances anti-tumor efficacy with chemotherapy and anti-PD1 Abs
- Increases infiltration of effector T lymphocytes into the TME
- TCGA analysis reveal multiple tumor types that appear reliant on EP2/4 pathway, informing clinical development
- A Phase 1/1b open-label, dose-escalation and dose-expansion study of TPST-1495 monotherapy or in combination with anti-PD-1 mAb is planned to initiate in early 2020

REFERENCES

1. Hirata et al (2012), Cancer Res 58(17): 3761-3764
2. O'Callaghan et al (2015), Br J Pharmacol 172(22): 5239-5250.
3. Kalinski et al (2012), J Immunol 188(1): 21-28.
4. Nakanishi et al (2013), Semin Immunopathol 35(2): 123-137.
5. Zelenay et al (2015), Cell 162(6): 1257-1270.
6. Wang e al (2016), Trends Mol Med 22(1): 1-3.
7. Subbaramaiah et al (1996), Cancer Res 56(19): 4424-4429
8. Buckman et al (1998), Carcinogenesis 19(5): 723-729.
9. Hida et al (1998), Cancer Res 58(17): 3761-3764.
10. Hwang et al (1998), J Natl Cancer Inst 90(6): 455-460.
11. Soslow et al (2000, Cancer 89(12): 2637-2645.
12. Maier et al (2004), Biochem Pharmacol 67(8): 1469-1478
13. Tsujii et al (1995), Cell 83(3): 493-501.
14. Tsujii et al (1998), Cell 93(5): 705-716.
15. Xu et al (2014), Sci Transl Med 6(242): 242ra284.
16. Kashiwagi et al (2018), Br J Cancer 118(2): 213-223.